Expanded and engineered NK cells upregulate expression of activation and survival genes associated with increased cytotoxicity and persistence

Mira Tohmé, Ph.D.; Sasha Lazetic; Kate Jamboretz; Luxuan Buren, Ph.D.; Chao Guo, Ph.D.; James B. Trager, Ph.D.

Nkarta Therapeutics, South San Francisco, California, USA

nkarta THERAPEUTICS

Poster # 2304

ABSTRACT

Background: Natural Killer (NK) cells expanded using irradiated K562 cells engineered to express membrane-bound IL-15 and 4-1BBL (K562-mb15-4-1BBL) have been employed in the clinic **[1].** Expansion upregulated activating NK cell receptors and increased NK cytotoxicity against tumor cells. Expanded NK cells can be engineered to express chimeric receptors to further improve their cytotoxicity. Chimeric receptors commonly incorporate an antigen binding domain, a costimulatory domain (i.e. 41BB, CD28) and a CD3ζ signaling domain. Receptor structure can bias engineered cells to chronic activation and exhaustion **[2,3]**, and can regulate metabolic pathways that influence cell phenotype and function. Characterization of NK cells expressing activating Chimeric Receptors (aCRs) may enable assessment of their clinical potency. We therefore evaluated the phenotypic characteristics of expanded and engineered NK cells and investigated signatures of activation or exhaustion upon antigen stimulation.

Material and Methods: NK cells from peripheral blood mononuclear cells were expanded on irradiated K562-mbIL15-41BBL cells and transduced with NKG2D based chimeras (NKG2D.aCR) and mbIL15. The expression of relevant receptors on expanded and transduced NK cells was assessed by flow cytometry. To evaluate the impact of tonic aCR chimeric receptor signaling versus antigen-driven stimulation on NK activation, we analyzed gene expression after stimulation with either isotype or anti-NKG2D coated beads using a Nanostring immune function panel.

Results: *Ex vivo* expansion generates potent, activated NK cells with high expression of NKG2D, Nkp30, Nkp44, Nkp46, CD69, CD25, and DNAM-1 activation markers and decreased expression of KLRG1 and CD158b inhibitory receptors. Expanded NK cells also upregulated TIGIT and TIM-3 expression, markers associated with functional exhaustion. However, expanded NK cells retained enhanced tumor cell cytotoxicity and cytokine production.
NKG2D.aCR construct expression induced tonic signaling in engineered NK cells characterized by the upregulation of genes involved in cell survival (Bcl-2), cell activation (CD25) and cell mediated cytotoxicity (TRAIL).
NKG2D engagement further enhanced expression of anti-apoptotic genes, and genes encoding cytokines and chemokines required to control cell cytotoxicity (INF-γ, TNF-α) and recruitment to tumor sites (XCL1, CCL3, CCL4). Lastly, NKG2D stimulation of engineered cells led to a significant increase in the expression of TNF family members (41BB, Ox40L, Ox40, TRAIL). Increased 41BB, Ox40L and TRAIL expression was confirmed by flow cytometry and correlated with enhanced cytotoxicity. Our data suggest that, upon stimulation, NKG2D.aCR cells are highly activated with greater longevity. Moreover, our data allow the identification of biomarker candidates that reflect NK.aCR potency.

Figure 1: Nkarta Natural Killer (NK) cell platform showing ex vivo (NK) cell expansion, activation and genetic modification procedures

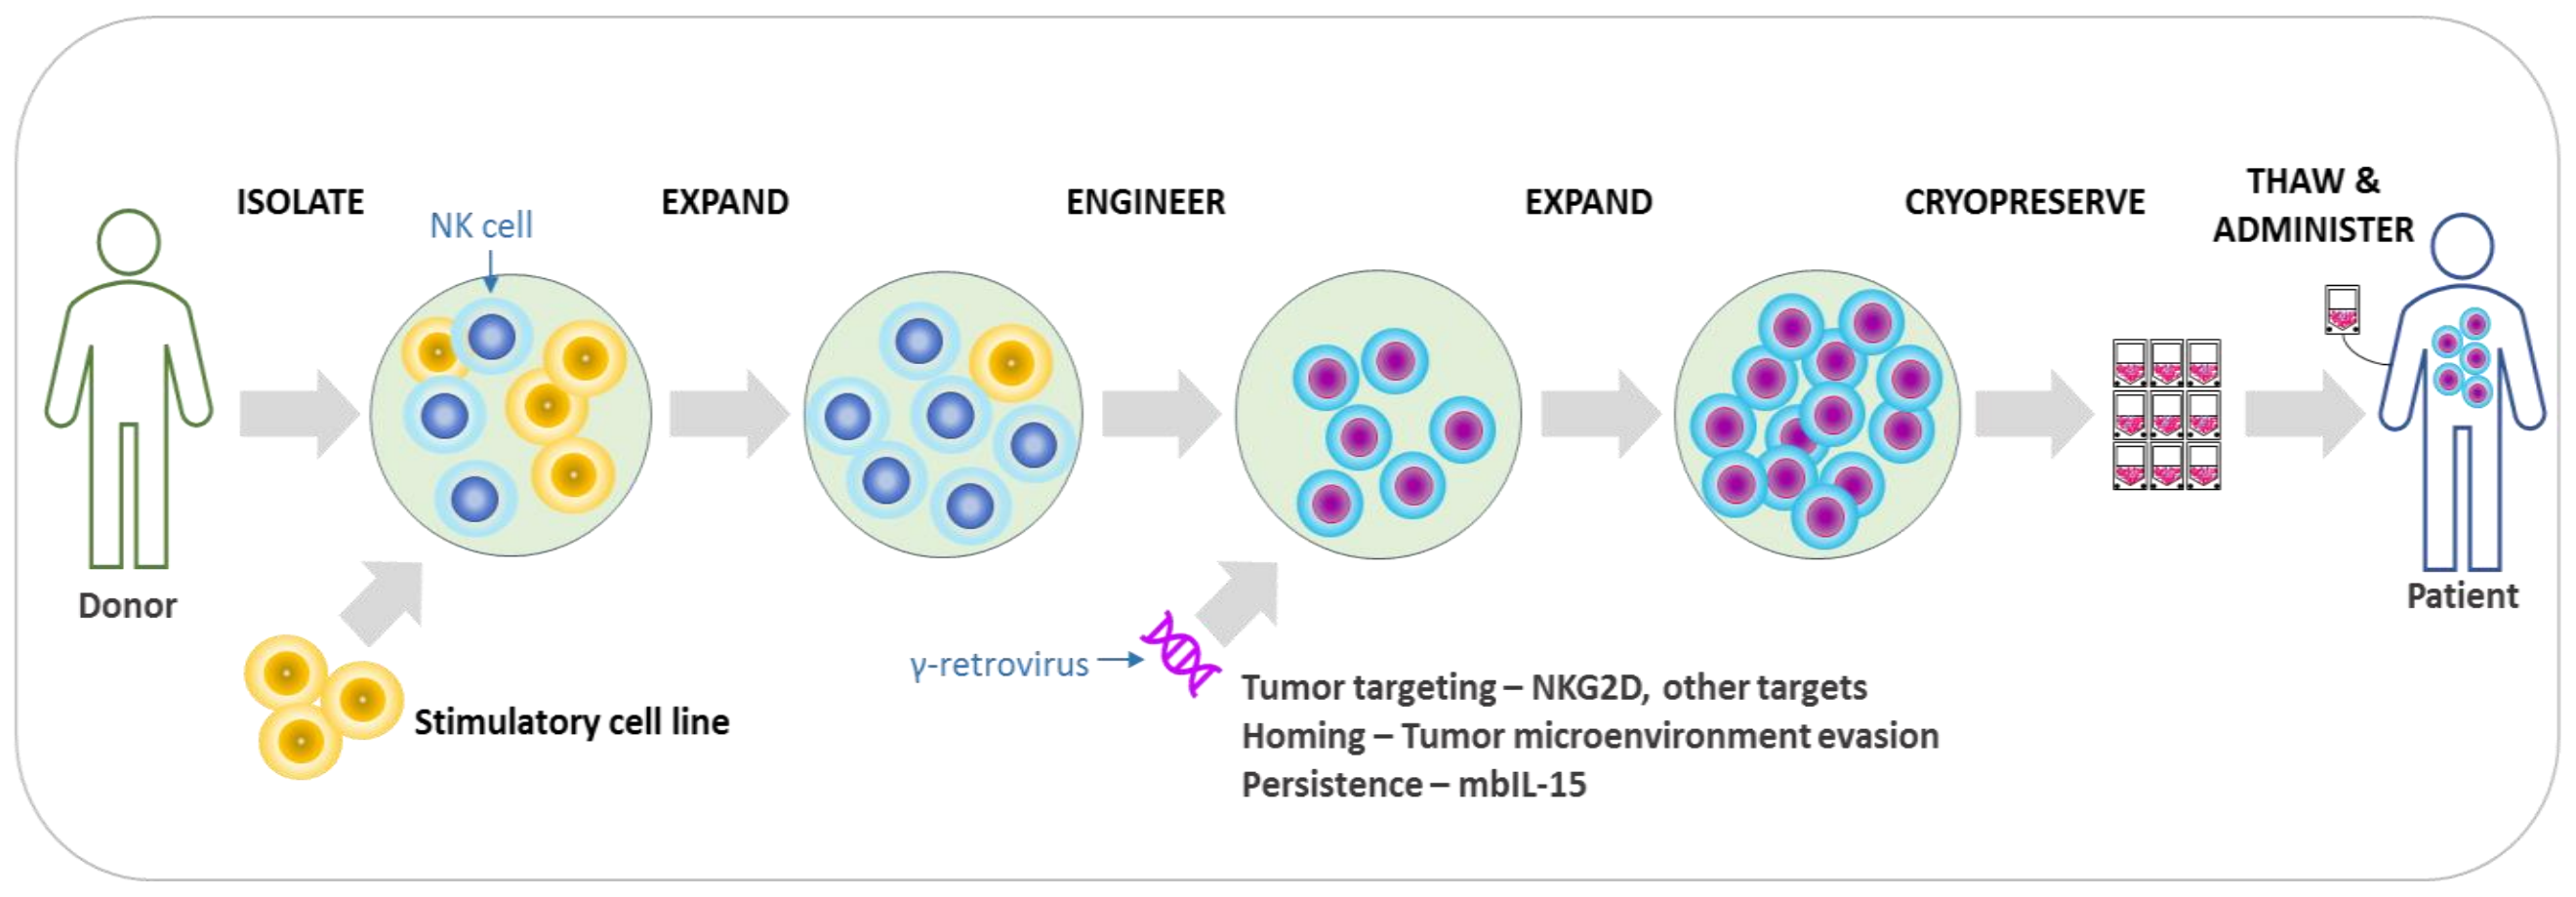
ISOLATE
NK cell
EXPAND
ENGINEER
EXPAND
CRYOPRESERVE
THAW & ADMINISTER
Donor
Patient
Stimulatory cell line
γ-retrovirus
Tumor targeting – NKG2D, other targets
Homing – Tumor microenvironment evasion
Persistence – mbIL-15

Figure 2: A balance of signals delivered by activating and inhibitory receptors regulates the recognition of healthy and tumor cells by NK cells.

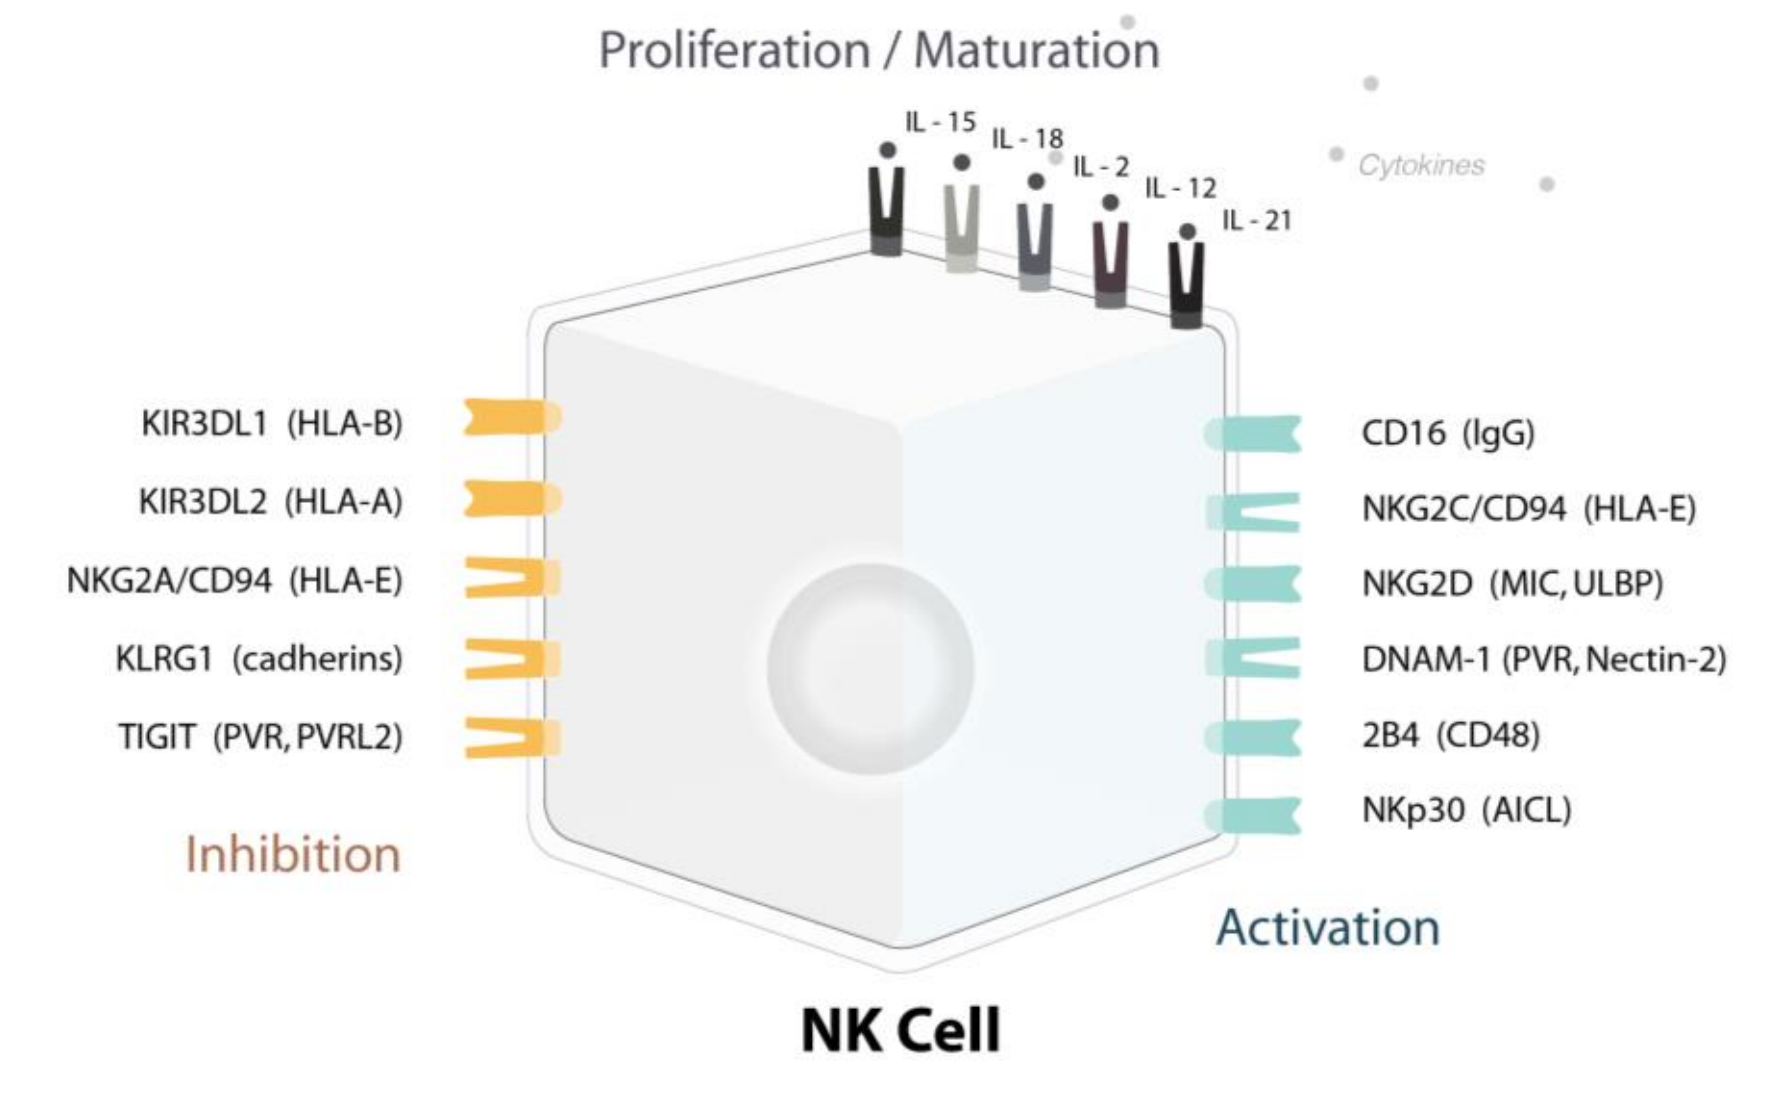
Proliferation / Maturation
IL - 15
IL - 18
IL - 2
IL - 12
IL - 21
Cytokines
KIR3DL1 (HLA-B)
KIR3DL2 (HLA-A)
NKG2A/CD94 (HLA-E)
KLRG1 (cadherins)
TIGIT (PVR, PVRL2)
Inhibition
CD16 (IgG)
NKG2C/CD94 (HLA-E)
NKG2D (MIC, ULBP)
DNAM-1 (PVR, Nectin-2)
2B4 (CD48)
NKp30 (AICL)
Activation
NK Cell

Figure 3: Importance of the NKG2D pathway in immune surveillance of cancer.

A. Human NKG2D complex

B. Schematic diagram showing all human NKG2D ligands identified to date

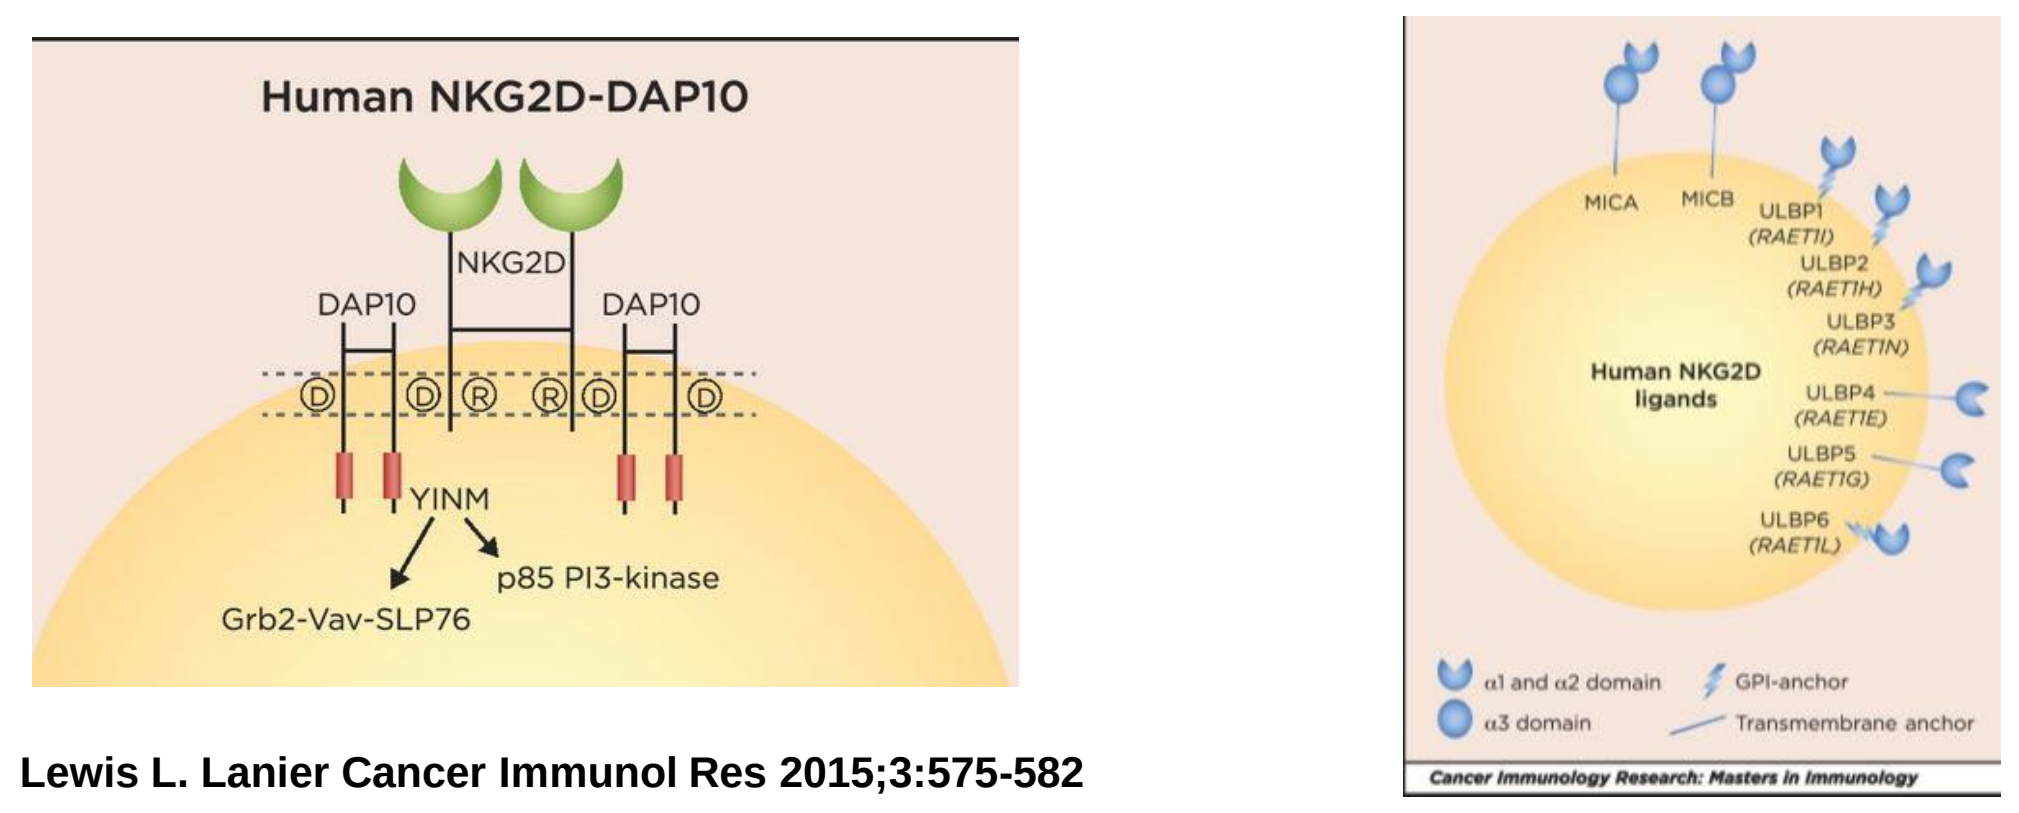
Human NKG2D-DAP10
NKG2D
DAP10
DAP10
YINM
p85 PI3-kinase
Grb2-Vav-SLP76
MICA
MICB
ULBP1 (RAET1I)
ULBP2 (RAET1H)
ULBP3 (RAET1N)
ULBP4 (RAET1E)
ULBP5 (RAET1G)
ULBP6 (RAET1L)
Human NKG2D ligands
α1 and α2 domain
α3 domain
GPI-anchor
Transmembrane anchor
Cancer Immunology Research: Masters in Immunology

Lewis L. Lanier Cancer Immunol Res 2015;3:575-582

- NKG2D is an activating receptor that is expressed on NK cells and subsets of T cells.
- NKG2D binds to numerous protein ligands that are upregulated and displayed on the surface in cells undergoing stress, excessive proliferation, or transformation, resulting in NK-mediated cytotoxicity of these abnormal cells and the secretion of cytokines and chemokines to enhance the immune response.

Figure 4: Characterization of K562-mb15-41BBL- expanded NK cells on Day7 and Day 12 post expansion

A.

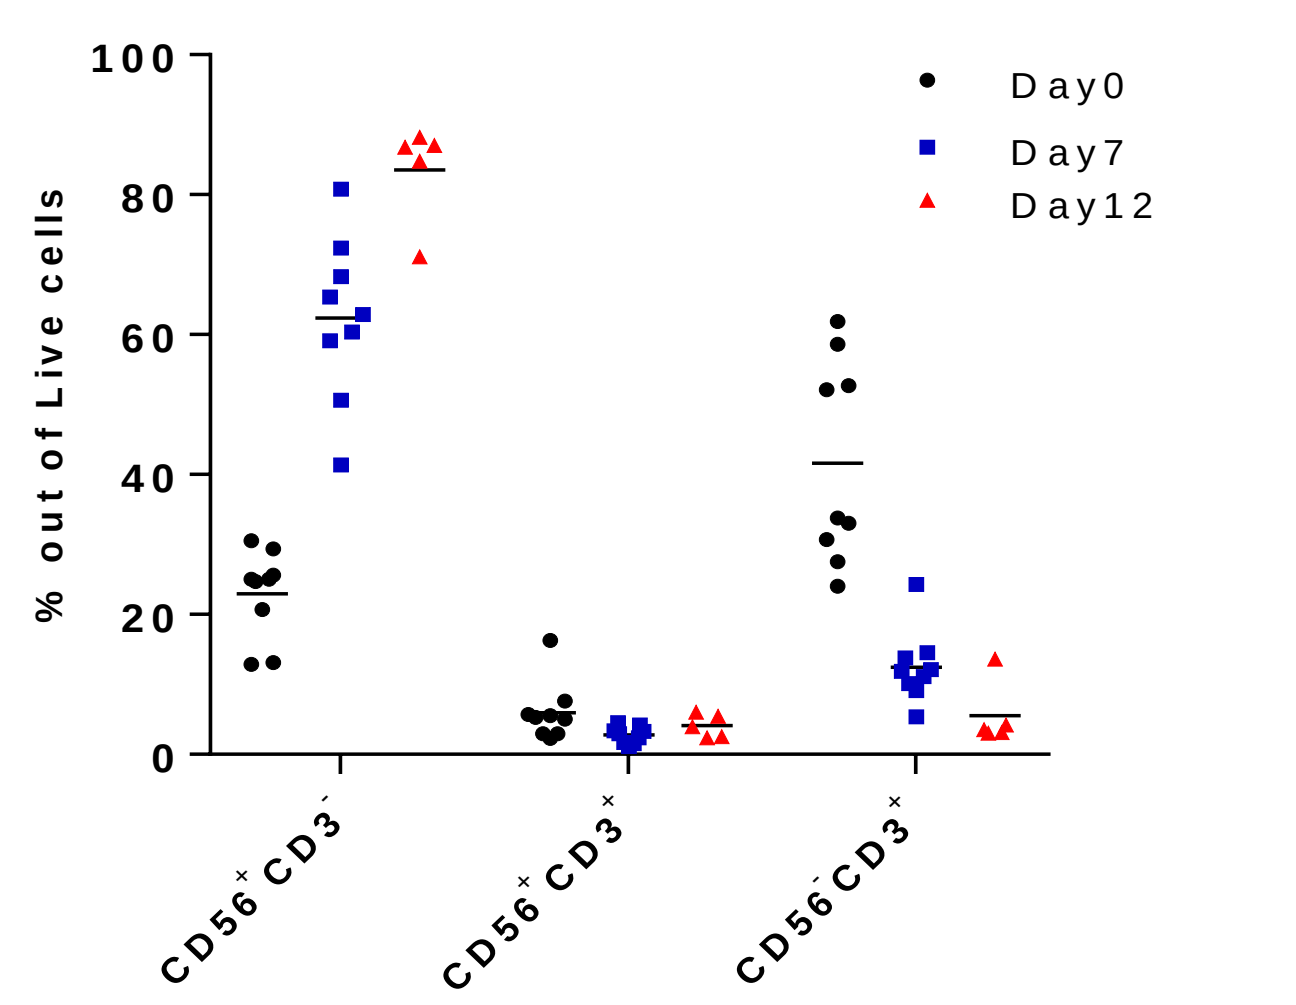
CD56 vs CD3
% out of Live cells
Day0
Day7
Day12
CD56+ CD3-
CD56+ CD3+
CD56- CD3+

Fig.4 Activated phenotype of expanded NK cells is maintained past day 7 to day 12 post expansion. ***(A-C)*** *NK cells from different donors were expanded with irradiated K562-mbIL15-41BBL stimulatory cells for 7 (n=9, blue squares) and 12 (n=5, red triangles) days. Thawed PBMCs (Day0, black dots) correspond to baseline control.*
(A) *Percentages of NK ($CD56^+CD3^-$), NKT ($CD56^+CD3^+$), and T ($CD56^-CD3^+$) cells in NK expansion cultures.*
Percentages of activating receptors CD16, NKG2D, NKp30, Nkp44, Nkp46, CD69, CD25 and DNAM-1 ***(B)*** *and of inhibitory receptors CD158b, KLRG1, NKG2a, PD1, TIGIT, and TIM-3* ***(C)*** *positive NK cells at day7 and day12 post expansion when compared to baseline.*

B.

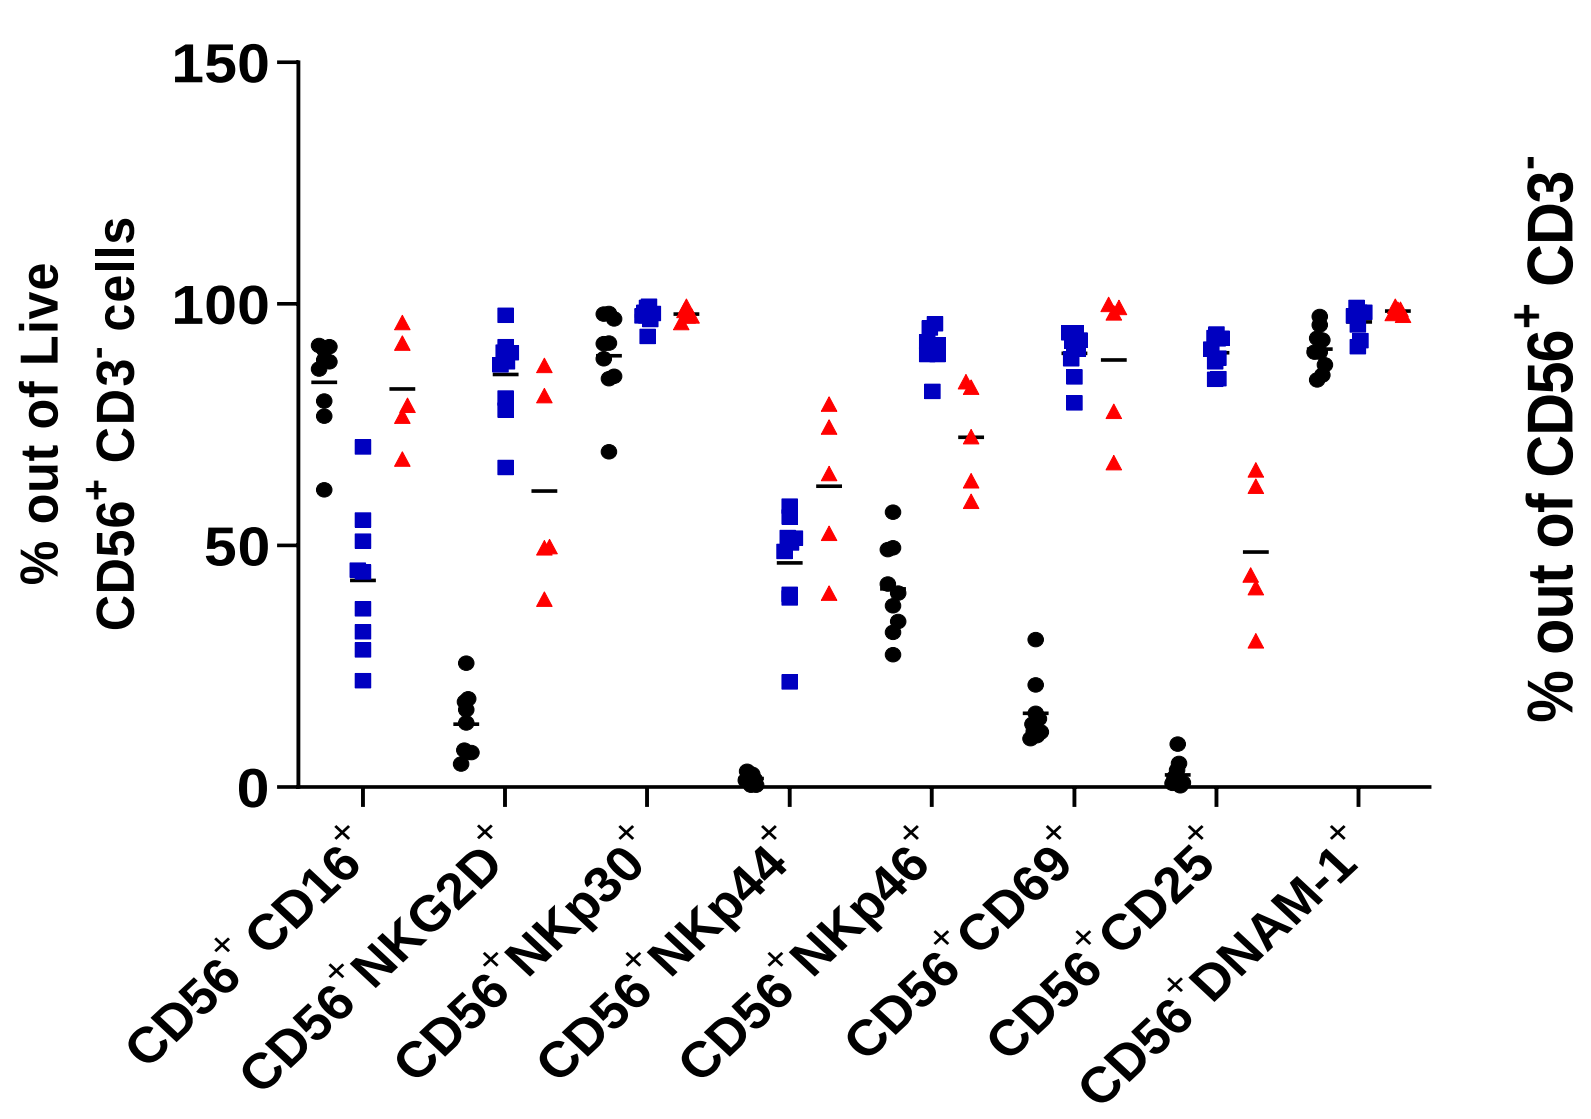
Activating Receptors
% out of Live CD56+ CD3- cells
CD56+ CD16+
CD56+ NKG2D+
CD56+ NKp30+
CD56+ NKp44+
CD56+ NKp46+
CD56+ CD69+
CD56+ CD25+
CD56+ DNAM-1+

C.

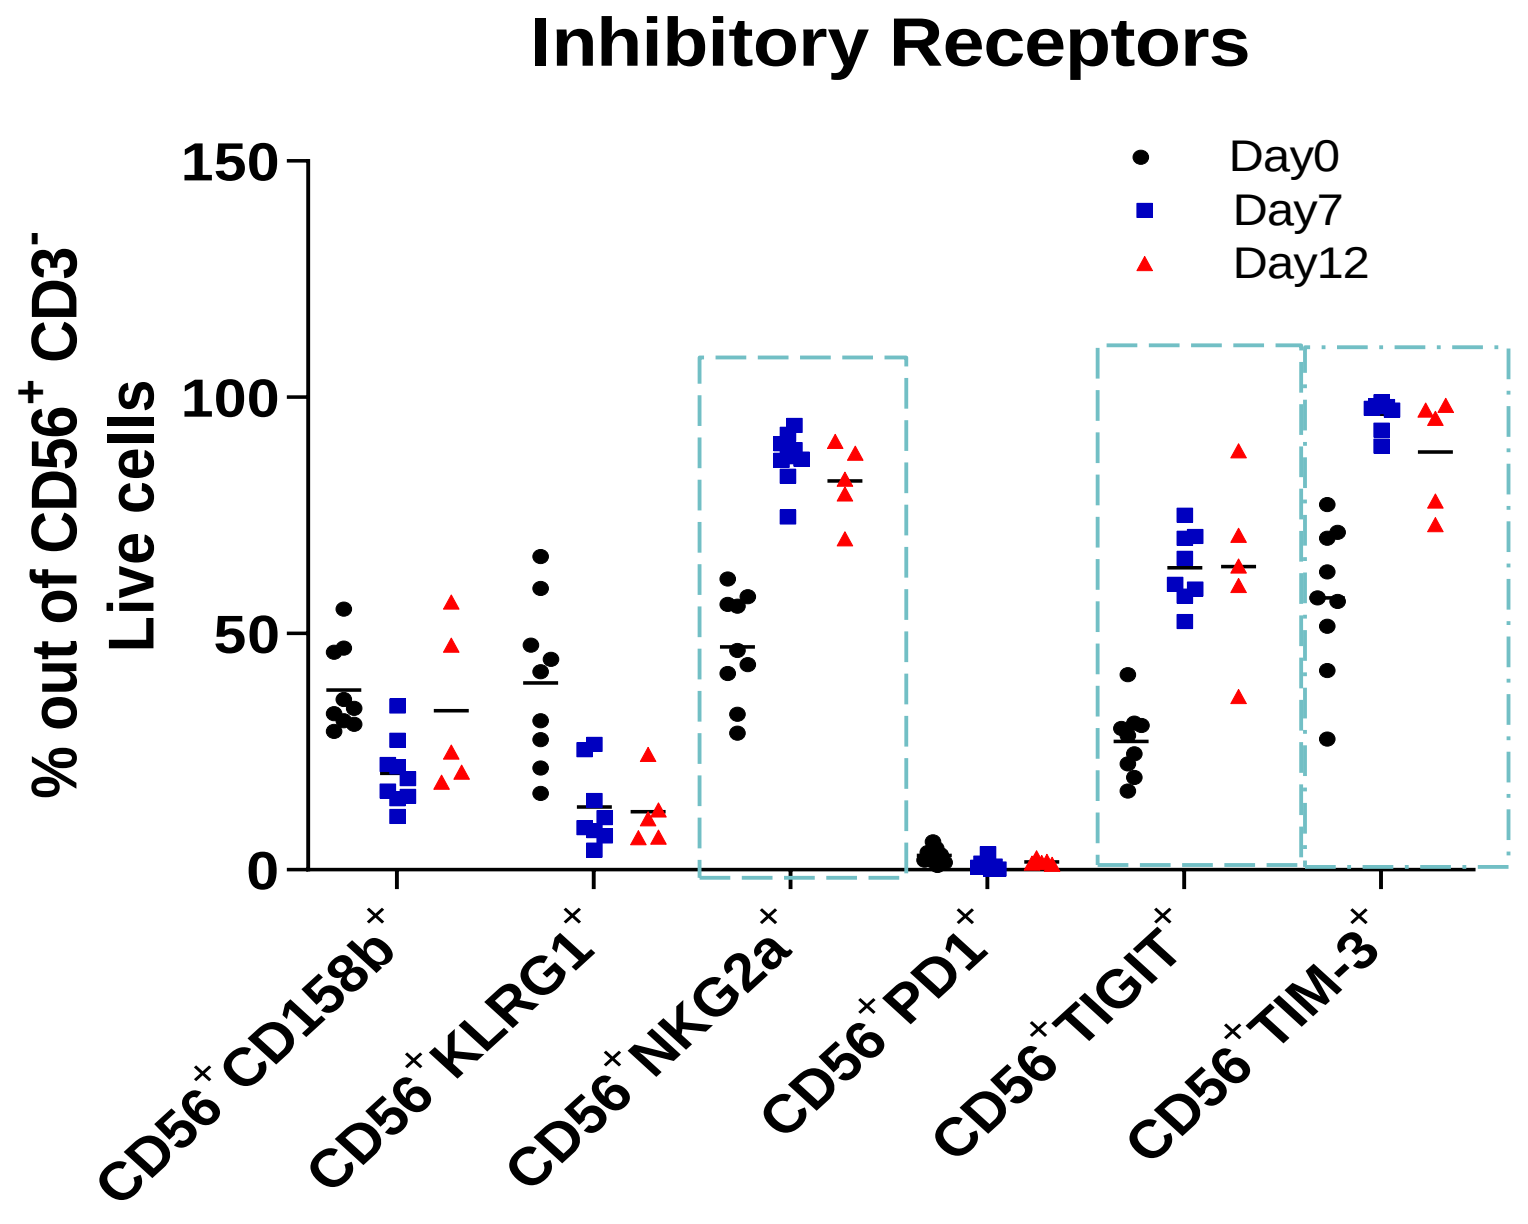
Inhibitory Receptors
% out of CD56+ CD3- Live cells
Day0
Day7
Day12
CD56+ CD158b+
CD56+ KLRG1+
CD56+ NKG2a+
CD56+ PD1+
CD56+ TIGIT+
CD56+ TIM-3+

Figure 5: Nkarta NKG2D based chimeras (NKG2D.aCR)

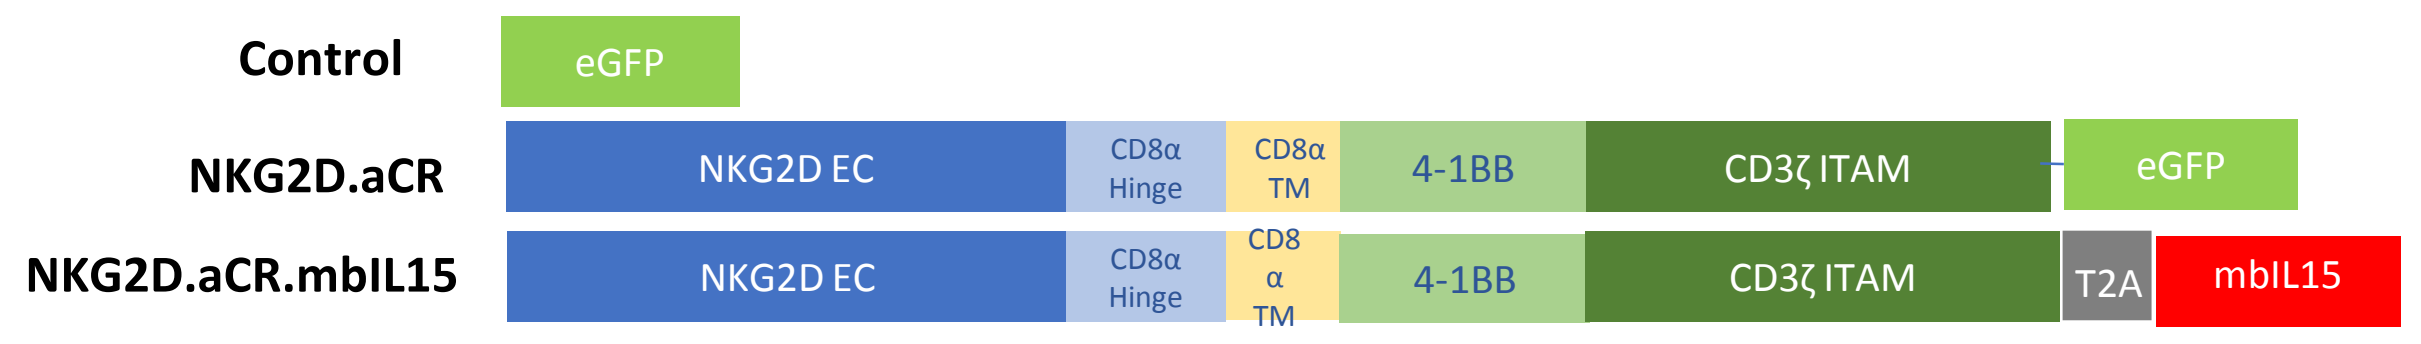
Control
eGFP
NKG2D.aCR
NKG2D EC
CD8α Hinge
CD8α TM
4-1BB
CD3ζ ITAM
eGFP
NKG2D.aCR.mbIL15
NKG2D EC
CD8α Hinge
CD8 α TM
4-1BB
CD3ζ ITAM
T2A
mbIL15

Figure 6: Co-expression of mbIL-15 and NKG2D.aCR induces upregulation of genes associated with cell survival and activation

A.

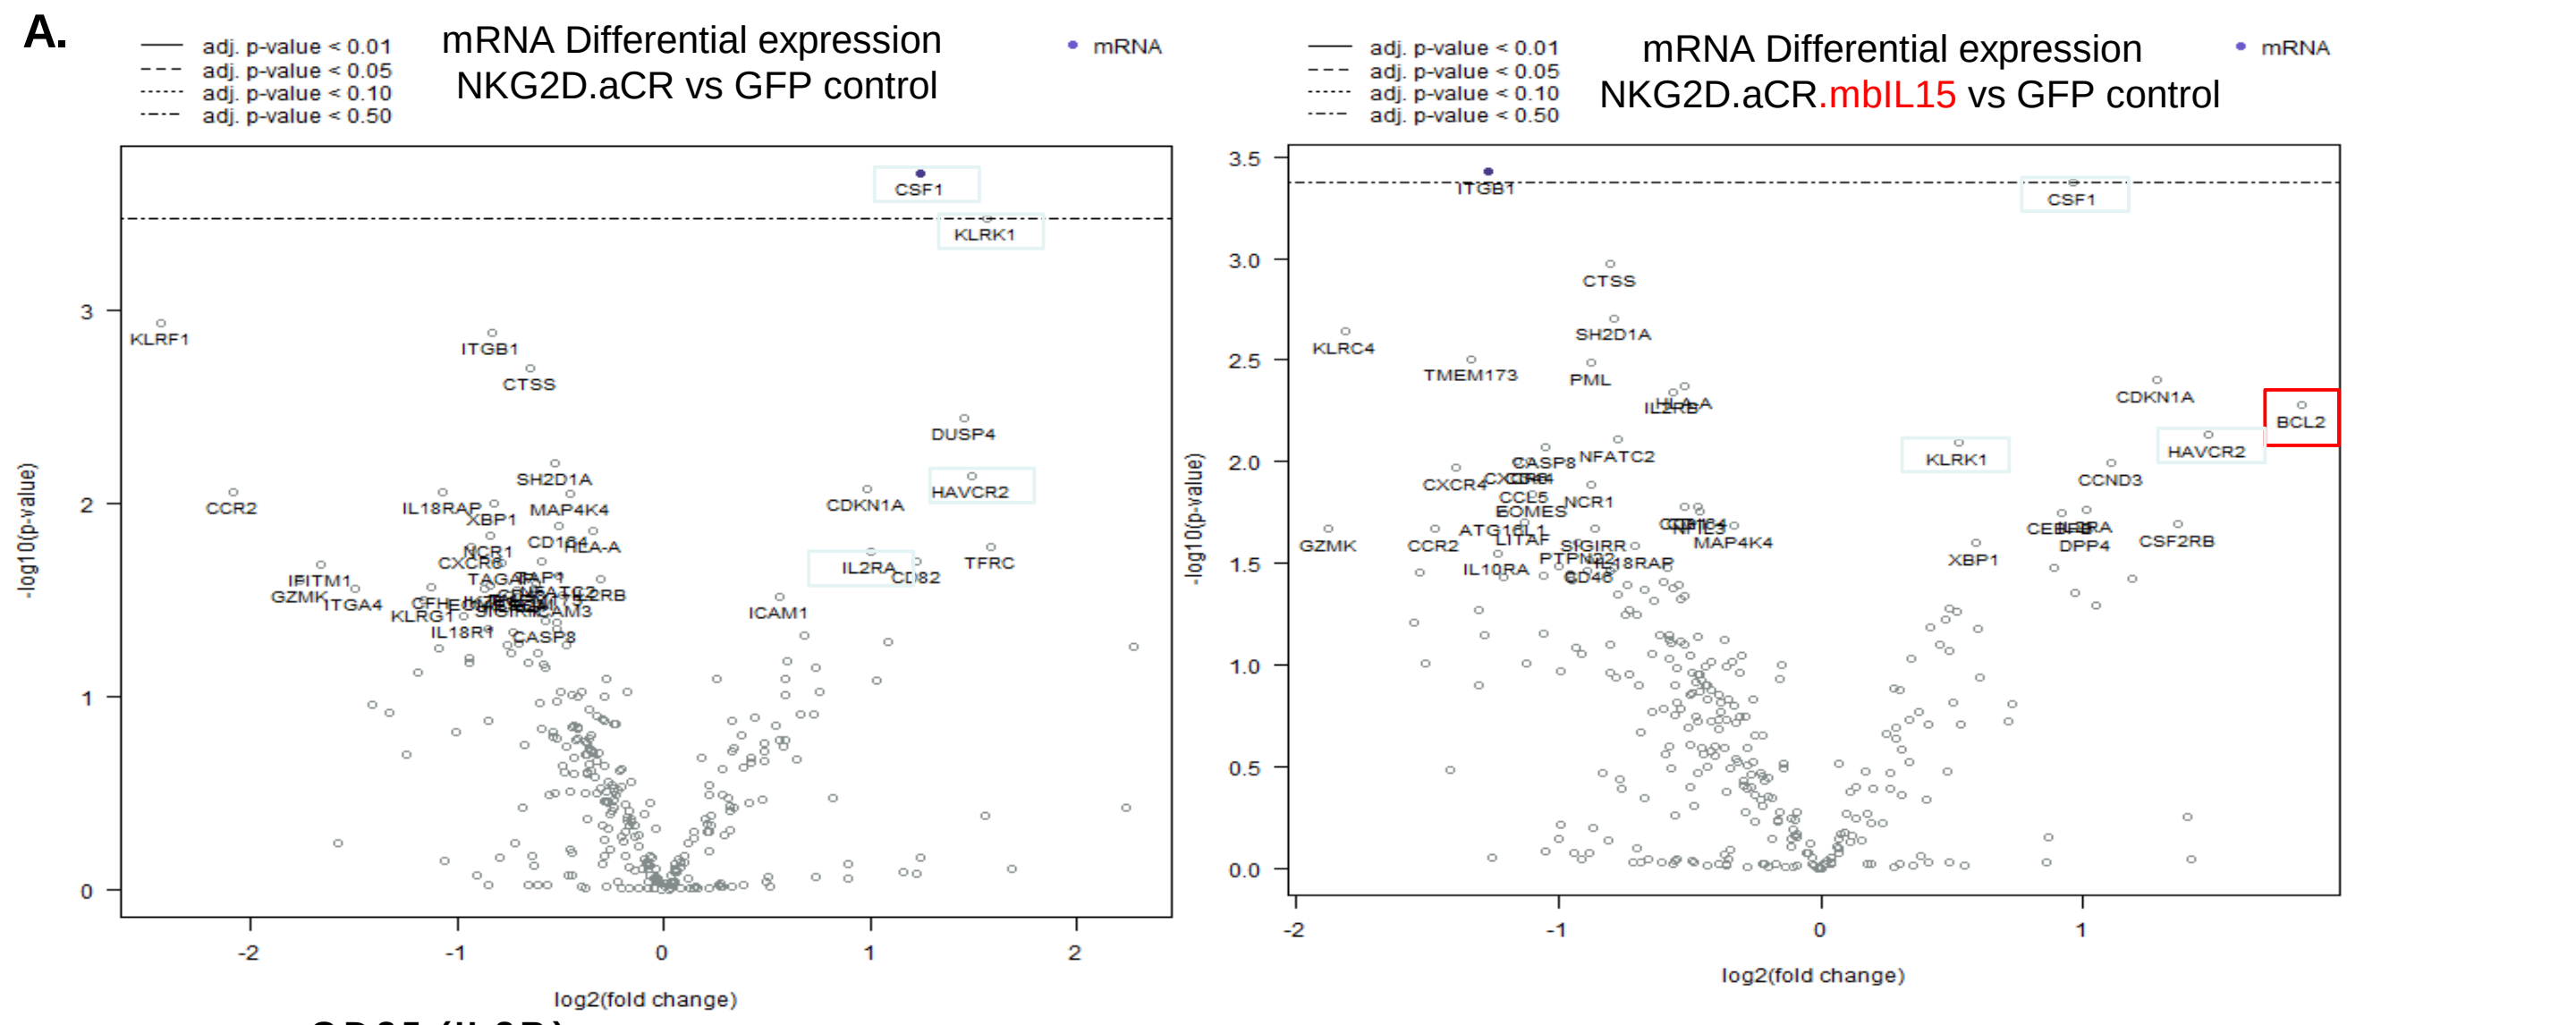
mRNA Differential expression
NKG2D.aCR vs GFP control
mRNA Differential expression
NKG2D.aCR.mbIL15 vs GFP control
adj. p-value < 0.01
adj. p-value < 0.05
adj. p-value < 0.10
adj. p-value < 0.50
mRNA
-log10(p-value)
log2(fold change)
CSF1
KLRK1
KLRF1
ITGB1
CTSS
DUSP4
HAVCR2
CDKN1A
IL2RA
TFRC
CD82
ICAM1
CCR2
BCL2
CDKN1A
HAVCR2
CCND3
CSF2RB
XBP1
KLRC4
TMEM173
SH2D1A
PML
GZMK

B.

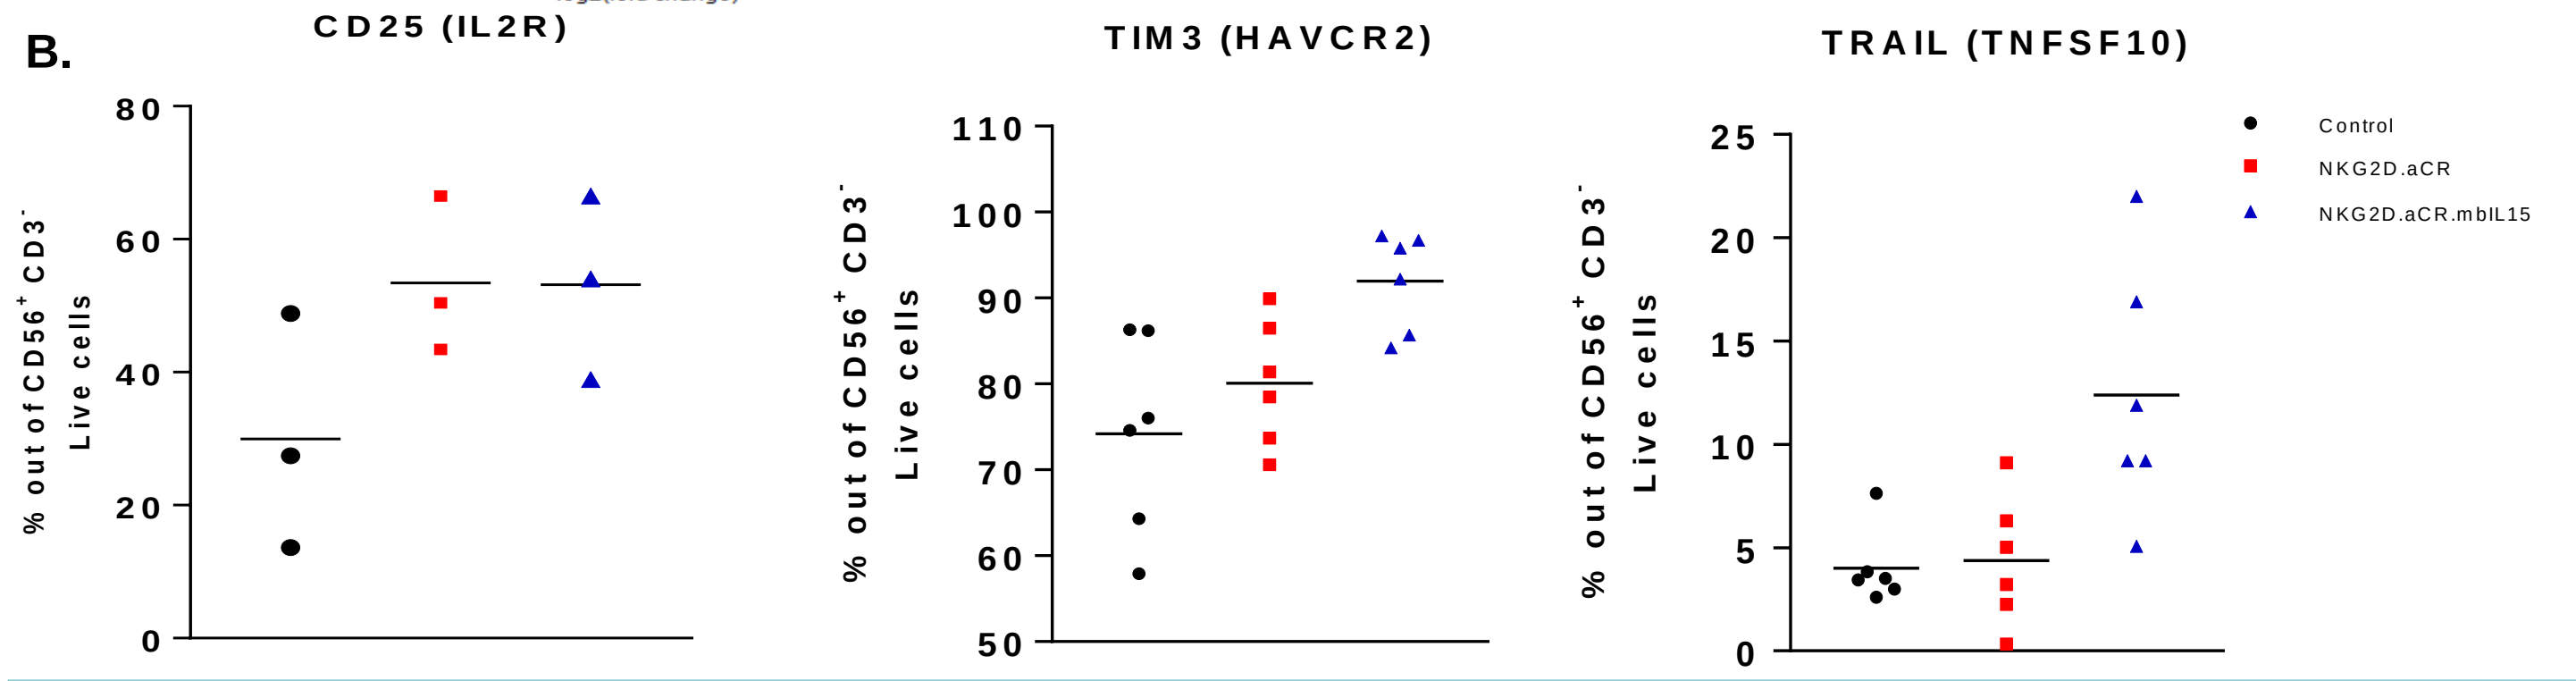
CD25 (IL2R)
TIM3 (HAVCR2)
TRAIL (TNFSF10)
% out of CD56+ CD3- Live cells
Control
NKG2D.aCR
NKG2D.aCR.mbIL15

Fig.6 (A-B) Upregulation of activation markers on engineered NK cells. ***A****. Differential gene expression in engineered NK cells. Volcano plots displaying gene's-log10 (p-value) and log2 fold change for NKG2D.aCR (Left) and NKG2D.aCR.mbIL15 (Right) compared to GFP control. Blue boxes represent common selected genes in both analyses. Gene in Red box (Bcl2) is uniquely overexpressed in NKG2D.aCR.mbIL15 NK cells* ***B.*** *Expanded NK cells from different donors (n=3-6) were transduced with GFP control and NKG2D.aCR constructs. Percentages of CD25, TIM3 and TRAIL positive engineered NK cells were assessed by flow cytometry four days post transduction.*

Figure 7: Increased activation and cytotoxicity of engineered NK cells upon NKG2D stimulation

A. NKG2D- vs isotype-stimulated NKG2D.aCR.mbIL15

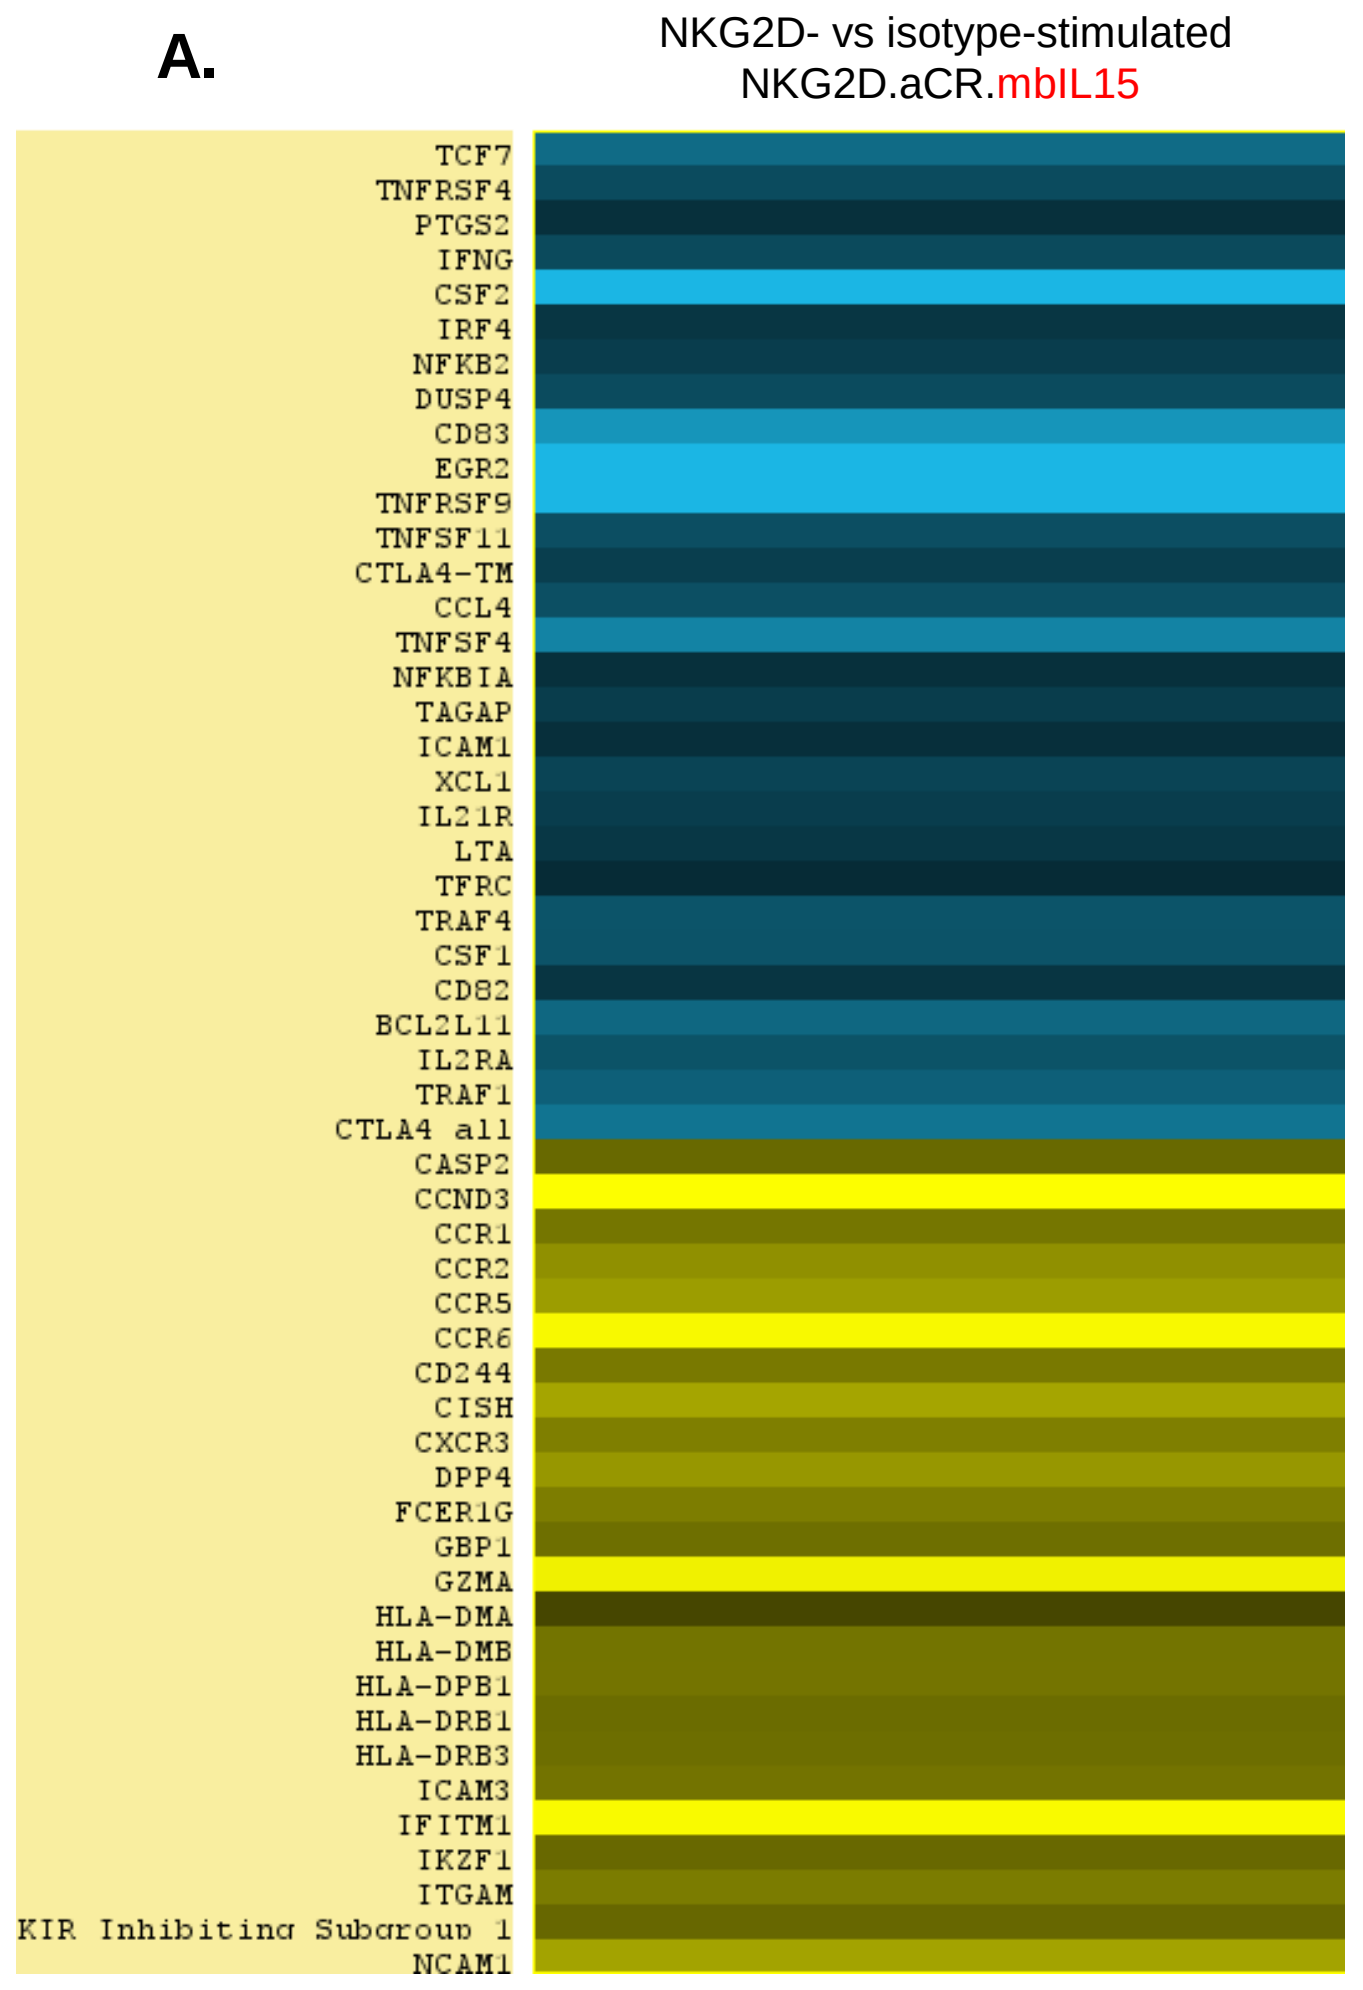
TCF7
TNFRSF4
PTGS2
IFNG
CSF2
IRF4
NFKB2
DUSP4
CD83
EGR2
TNFRSF9
TNFSF11
CTLA4-TM
CCL4
TNFSF4
NFKBIA
TAGAP
ICAM1
XCL1
IL21R
LTA
TFRC
TRAF4
CSF1
CD82
BCL2L11
IL2RA
TRAF1
CTLA4 all
CASP2
CCND3
CCR1
CCR2
CCR5
CCR6
CD244
CISH
CXCR3
DPP4
FCER1G
GBP1
GZMA
HLA-DMA
HLA-DMB
HLA-DPB1
HLA-DRB1
HLA-DRB3
ICAM3
IFITM1
IKZF1
ITGAM
KIR Inhibiting Subgroup 1
NCAM1

B. NKG2D- vs isotype-stimulated NKG2D.aCR

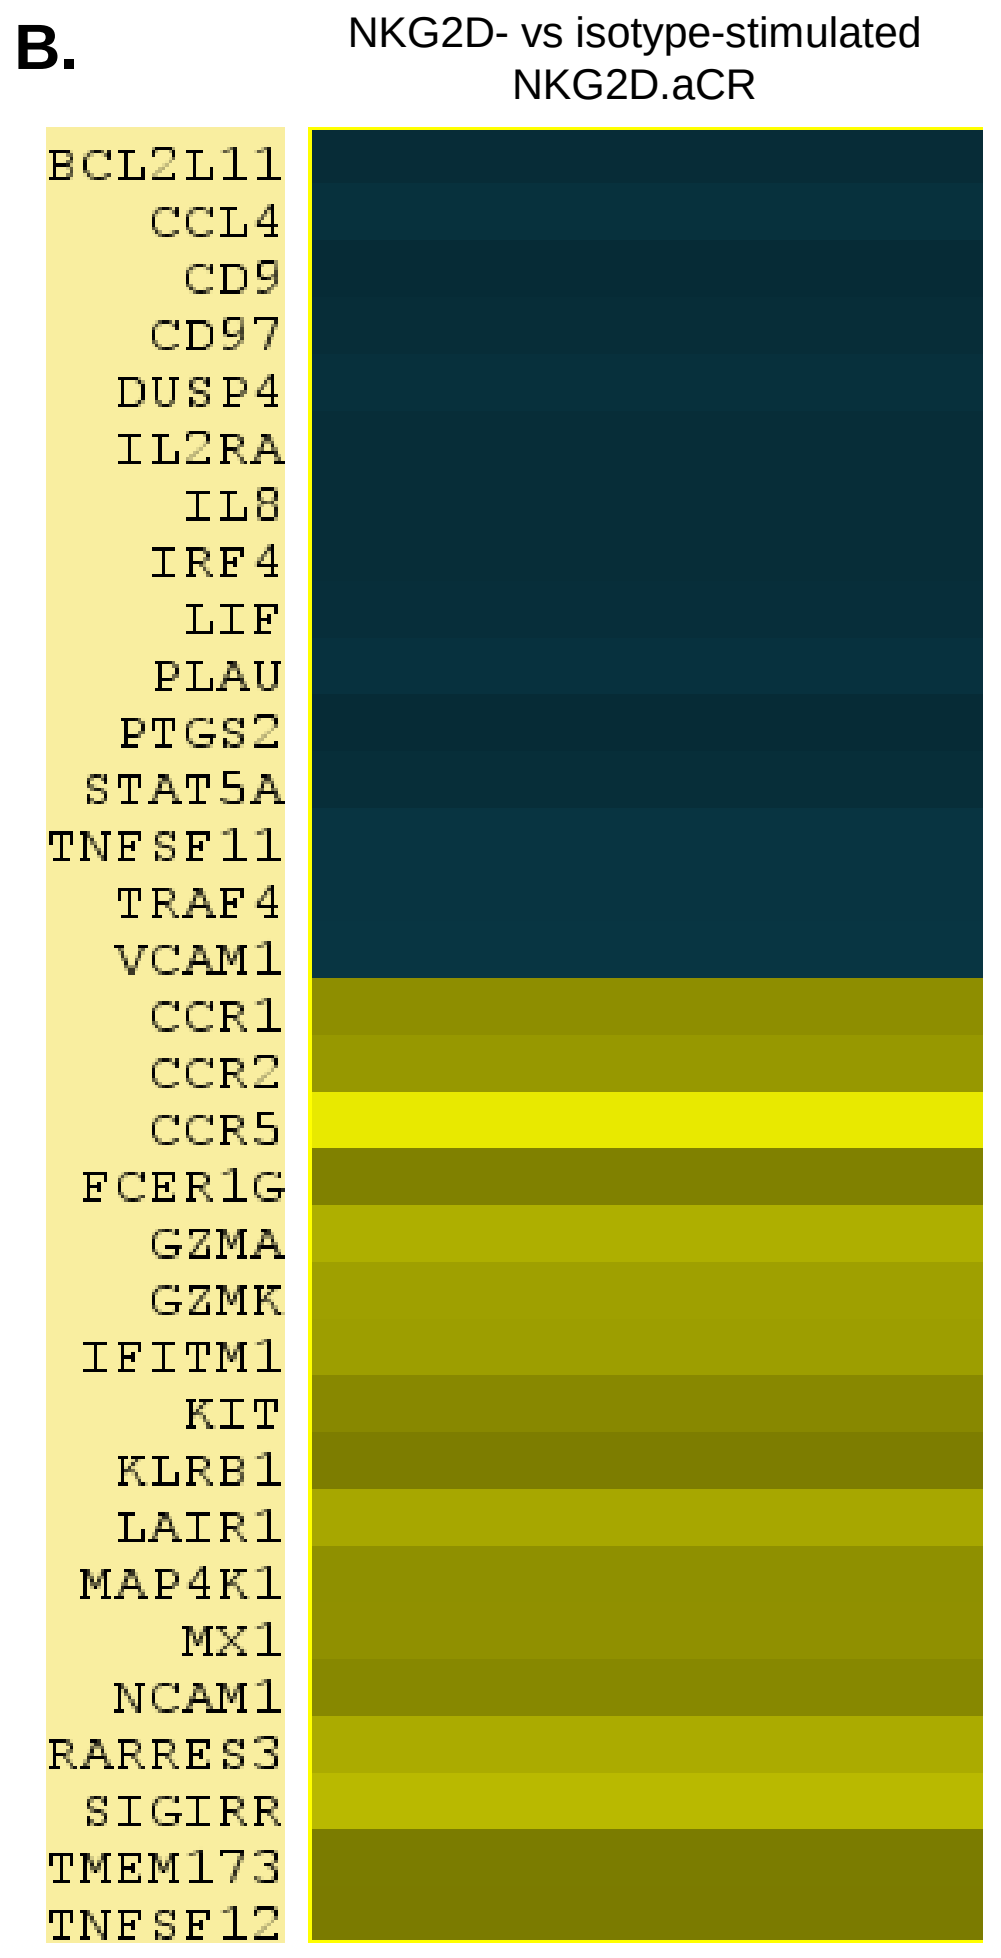
BCL2L11
CCL4
CD9
CD97
DUSP4
IL2RA
IL8
IRF4
LIF
PLAU
PTGS2
STAT5A
TNFSF11
TRAF4
VCAM1
CCR1
CCR2
CCR5
FCER1G
GZMA
GZMK
IFITM1
KIT
KLRB1
LAIR1
MAP4K1
MX1
NCAM1
RARRES3
SIGIRR
TMEM173
TNFSF12
-4.867
0.011
8.58

Fig.7 (A-B) *Heatmaps of selected genes differentially expressed in NKG2D-stimulated NKG2D.aCR.mbIL15* ***(A)*** *and NKG2D.aCR* ***(B)*** *compared to Isotype-stimulated engineered NK cells. The cutoff for Fold of change is >2 and for significance is P< 0.1. (n=2 per group).*

(C-E) *Engineered NK cells are stimulated with isotype (−) or NKG2D (+) coated beads at a Cell:Bead ratio of 1:1 for 16H. Expression of activation markers Ox40L, 41BB and TRAIL* ***(C)*** *and of LAMP-1 degranulation marker* ***(E)*** *was assessed by flow cytometry.* ***(D)*** *Quantification of IFN-γ, TNF-α, sTRAIL, and GM-CSF in culture supernatants using Luminex technology.*

(F) *NKG2D.aCR NK cell cytotoxicity on REH target tumor cells. Cytotoxicity was calculated based on live target cells after 2H co-culture at an Effector:Target cell ratio of 1:1.*

C.

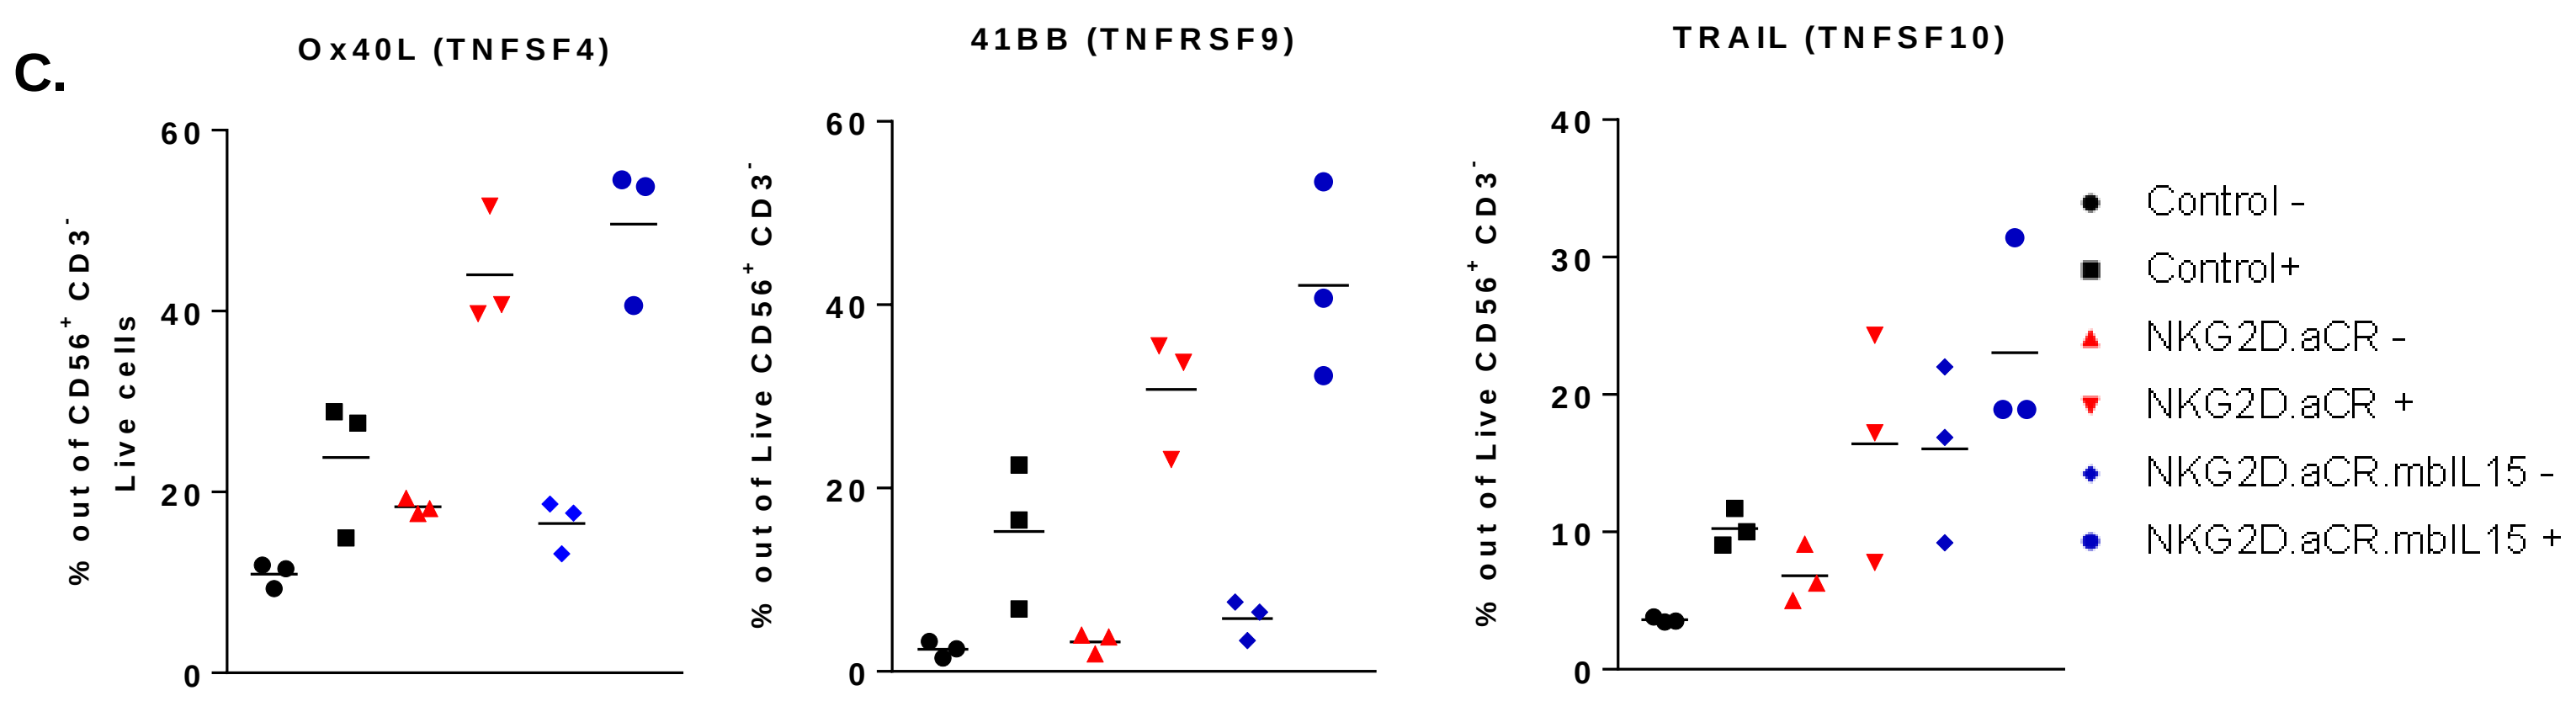
Ox40L (TNFSF4)
41BB (TNFRSF9)
TRAIL (TNFSF10)
% out of CD56+ CD3- Live cells
% out of Live CD56+ CD3-
Control -
Control+
NKG2D.aCR -
NKG2D.aCR +
NKG2D.aCR.mbIL15 -
NKG2D.aCR.mbIL15 +

D.

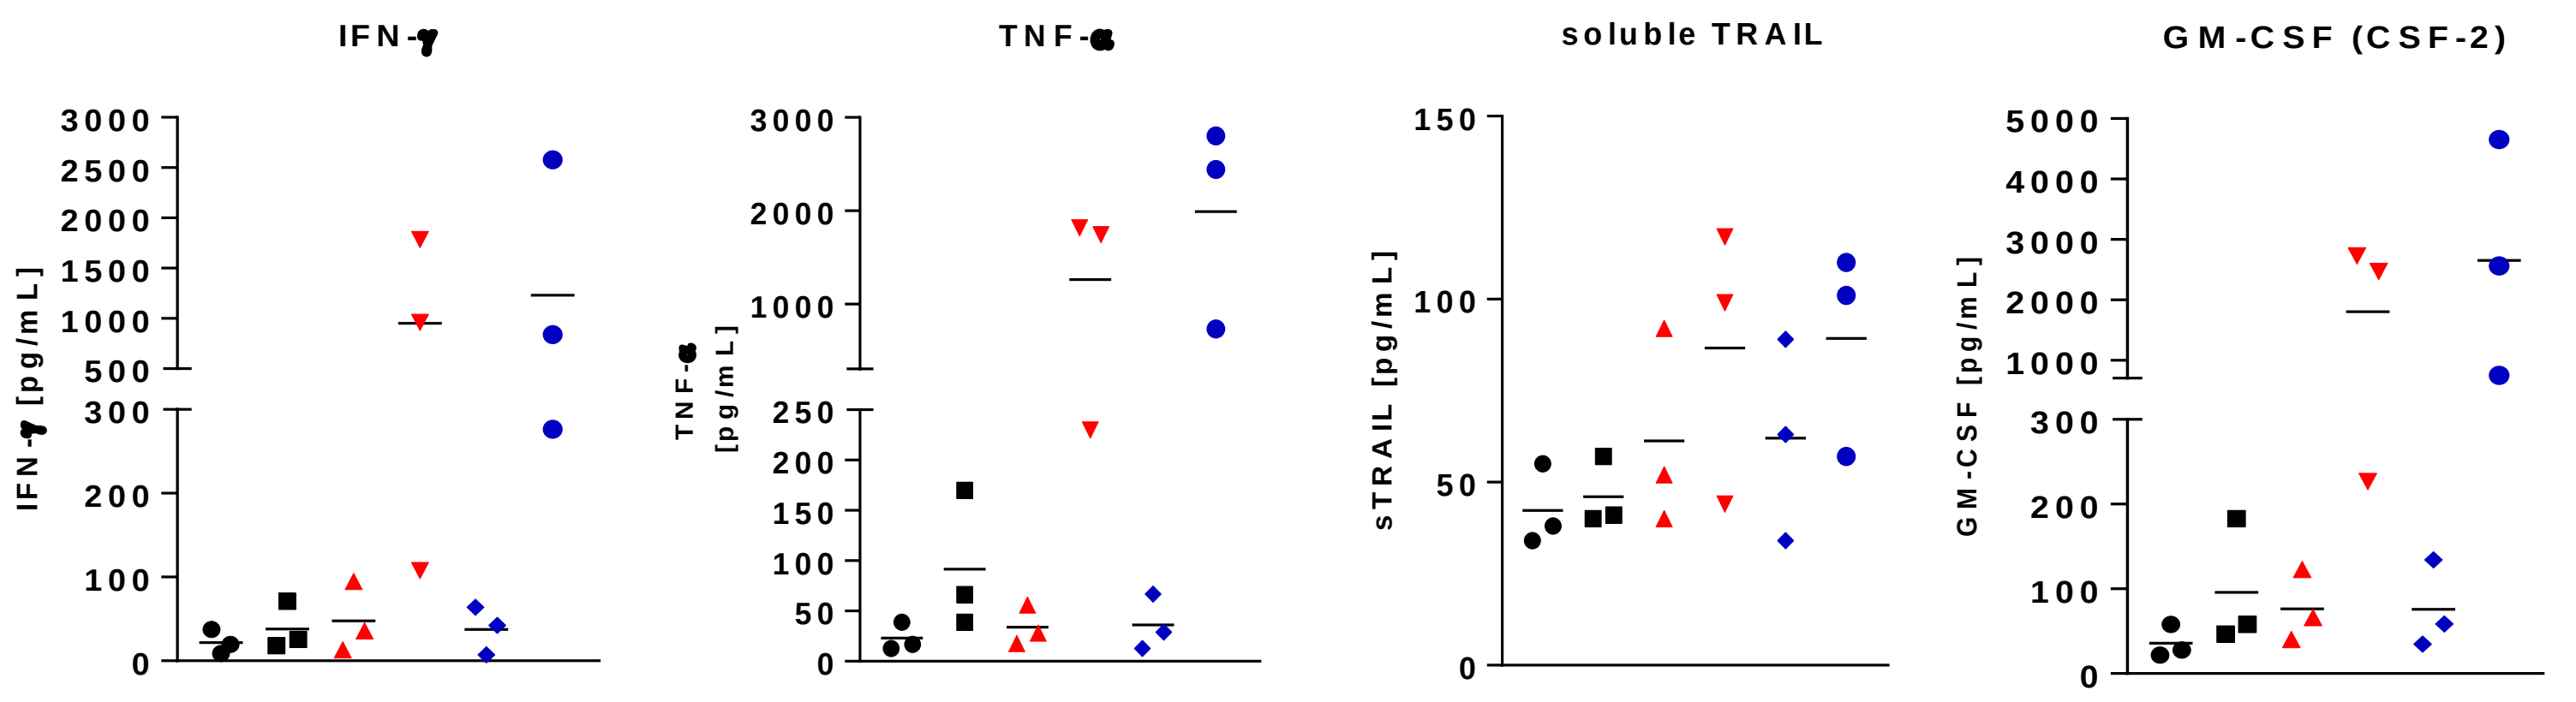
IFN-γ
TNF-α
soluble TRAIL
GM-CSF (CSF-2)
IFN-γ [pg/mL]
TNF-α [pg/mL]
sTRAIL [pg/mL]
GM-CSF [pg/mL]

E.

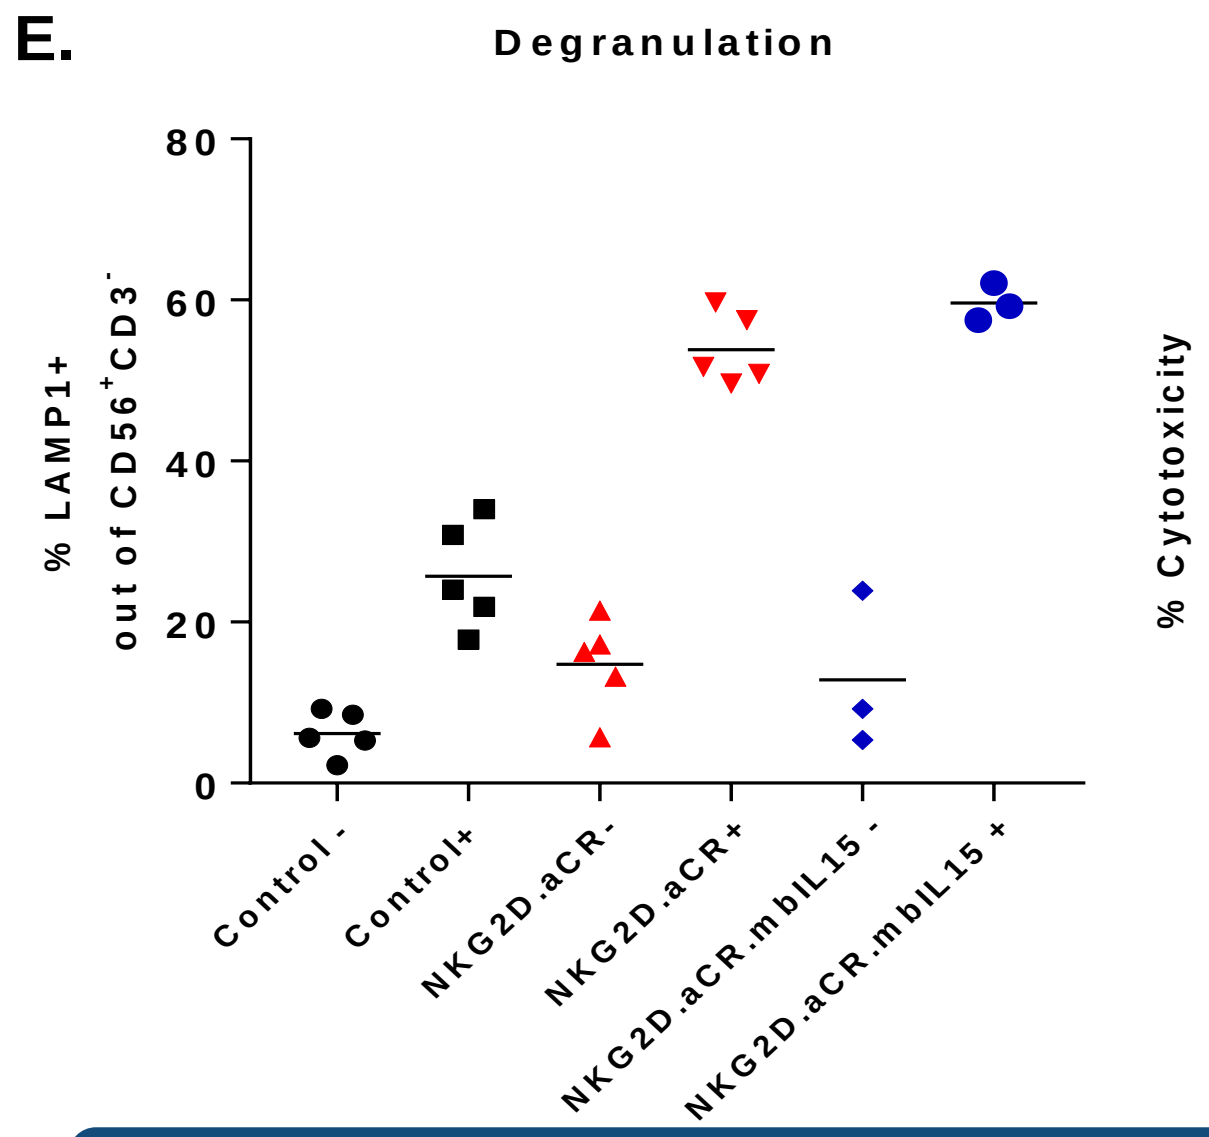
Degranulation
% LAMP1+ out of CD56+CD3-
Control -
Control+
NKG2D.aCR-
NKG2D.aCR+
NKG2D.aCR.mbIL15 -
NKG2D.aCR.mbIL15 +

F.

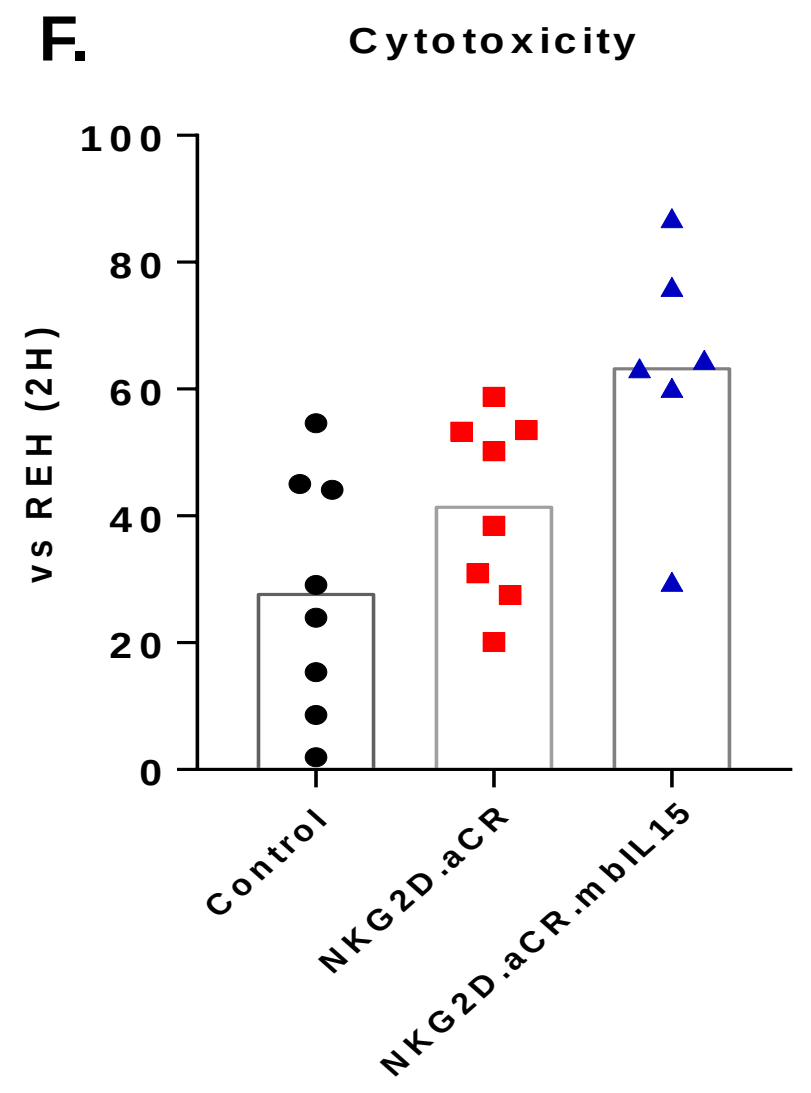
Cytotoxicity
% Cytotoxicity vs REH (2H)
Control
NKG2D.aCR
NKG2D.aCR.mbIL15

CONCLUSIONS

- Nkarta's expansion and transduction platform generates highly activated NKG2D.aCR engineered NK cells with greater longevity.
- Investigating NKG2D.aCR gene signatures allow the identification of biomarker candidates that reflect NKG2D.aCR potency.
- NKG2D.aCR provides a robust approach to cancer therapy that can be combined with existing therapies, and has potential for allogeneic or autologous applications to ultimately improve outcomes for patients in hematological malignancies and solid tumor settings.

REFERENCES

1. Leivas A., et al. Novel treatment strategy with autologous activated and expanded natural killer cells plus anti-myeloma drugs for multiple myeloma. *Oncoimmunology* (2016)
2. Virgin H.W., et al. Redefining chronic viral infection. *Cell* (2009)
3. Wherry E.J. T cell exhaustion. *Nat. Immunol*. (2011)

CONTACT James Trager, PhD jtrager@nkartatx.com www.nkartatx.com