ADAM17 knockout NK or CAR NK cells augment Antibody Dependent Cellular Cytotoxicity (ADCC) and antitumor activity

'Chao Guo, Ph.D.[1] , Yanying Fan, Ph.D.[2], Alexander Aronov[1], Nikki Kimura[1], Damie Juat[1], Jessica Sood[1], Ivan H. Chan, Ph.D.[1], James B. Trager, Ph.D.[1]*,
[1]Nkarta Therapeutics, South San Francisco, CA, USA [2]Current address: Adicet Bio, Redwood City, CA, USA *corresponding author

nkarta

Introduction

Natural killer (NK) cells are a critical component of the innate immune system. NK cells can be expanded from healthy donors and do not elicit graft-versus-host disease, making them an attractive source of 'off-the-shelf' allogeneic cell therapy. NK cells are also amenable to CRISPR and chimeric antigen receptor (CAR) genomic engineering for enhanced functions. Another important anti-tumor role of NK cells is via antibody-dependent cell mediated cytotoxicity (ADCC). The process is mediated by the CD16a receptor of NK cells, which binds the Fc portion of antibodies. After NK cell activation, the CD16a ectodomain is rapidly cleaved from the NK cell surface by a disintegrin, Metalloprotease-17 (ADAM17), that is produced by both tumor cells and activated NK cells[4]. This dampens NK cell activity by preventing NK cell attachment to antibody-coated target cells and diminishes CD16a signaling. Inhibiting ADAM17-mediated CD16a cleavage leads to augmented ADCC activity and increased cytokine production by human NK cells. Other NK cell activating receptors are known to be regulated by a similar mechanism[1].

Methods

In this study, we have utilized CRISPR-Cas9 ribonucleoproteins (RNPs) to disrupt the ADAM17 gene in isolated peripheral blood NK cells from healthy donors. FACS and Amplicon NGS Sequencing data confirmed successful disruption of the gene. Edited NK cells were expanded using IL-2 and stimulation with NKSTIM, a modified K562 stimulatory cell line expressing a membrane-bound form of IL-15 (mbIL-15) and 4-1BBL. IL-12 and IL-18 were added during expansion to drive memory-like NK cell differentiation. Furthermore, we transduced ADAM17 edited NK cells to express a CD70 CAR[2] or CD19 CAR[3] construct and mbIL-15. CAR expression was assessed by FACS. In vitro cytotoxicity was measured by IncuCyte®.

Results

Using an artificial activation system (phorbol 12-myristate 13-acetate [PMA]), we showed that ADAM17 KO NK cells maintain dramatically higher surface expression of CD16a and CD62L than control NK cells. In IncuCyte® cytotoxicity assays against Raji tumor cells in the presence of rituximab (anti-CD20), ADAM17 KO NK cells and ADAM17 KO CD19 CAR NK cells demonstrated higher cytotoxicity compared to control NK cells. Similarly, ADAM17 KO NK cells and ADAM17 KO CD70 CAR NK cells also showed enhanced cytotoxicity against 786-O tumor cells in the presence of anti-EGFR antibody, cetuximab. Furthermore, ADAM17/CISH/CBLB triple KO CD70 CAR NK cells exhibited enhanced antitumor efficacy in vivo in a HL60 AML xenograft model.

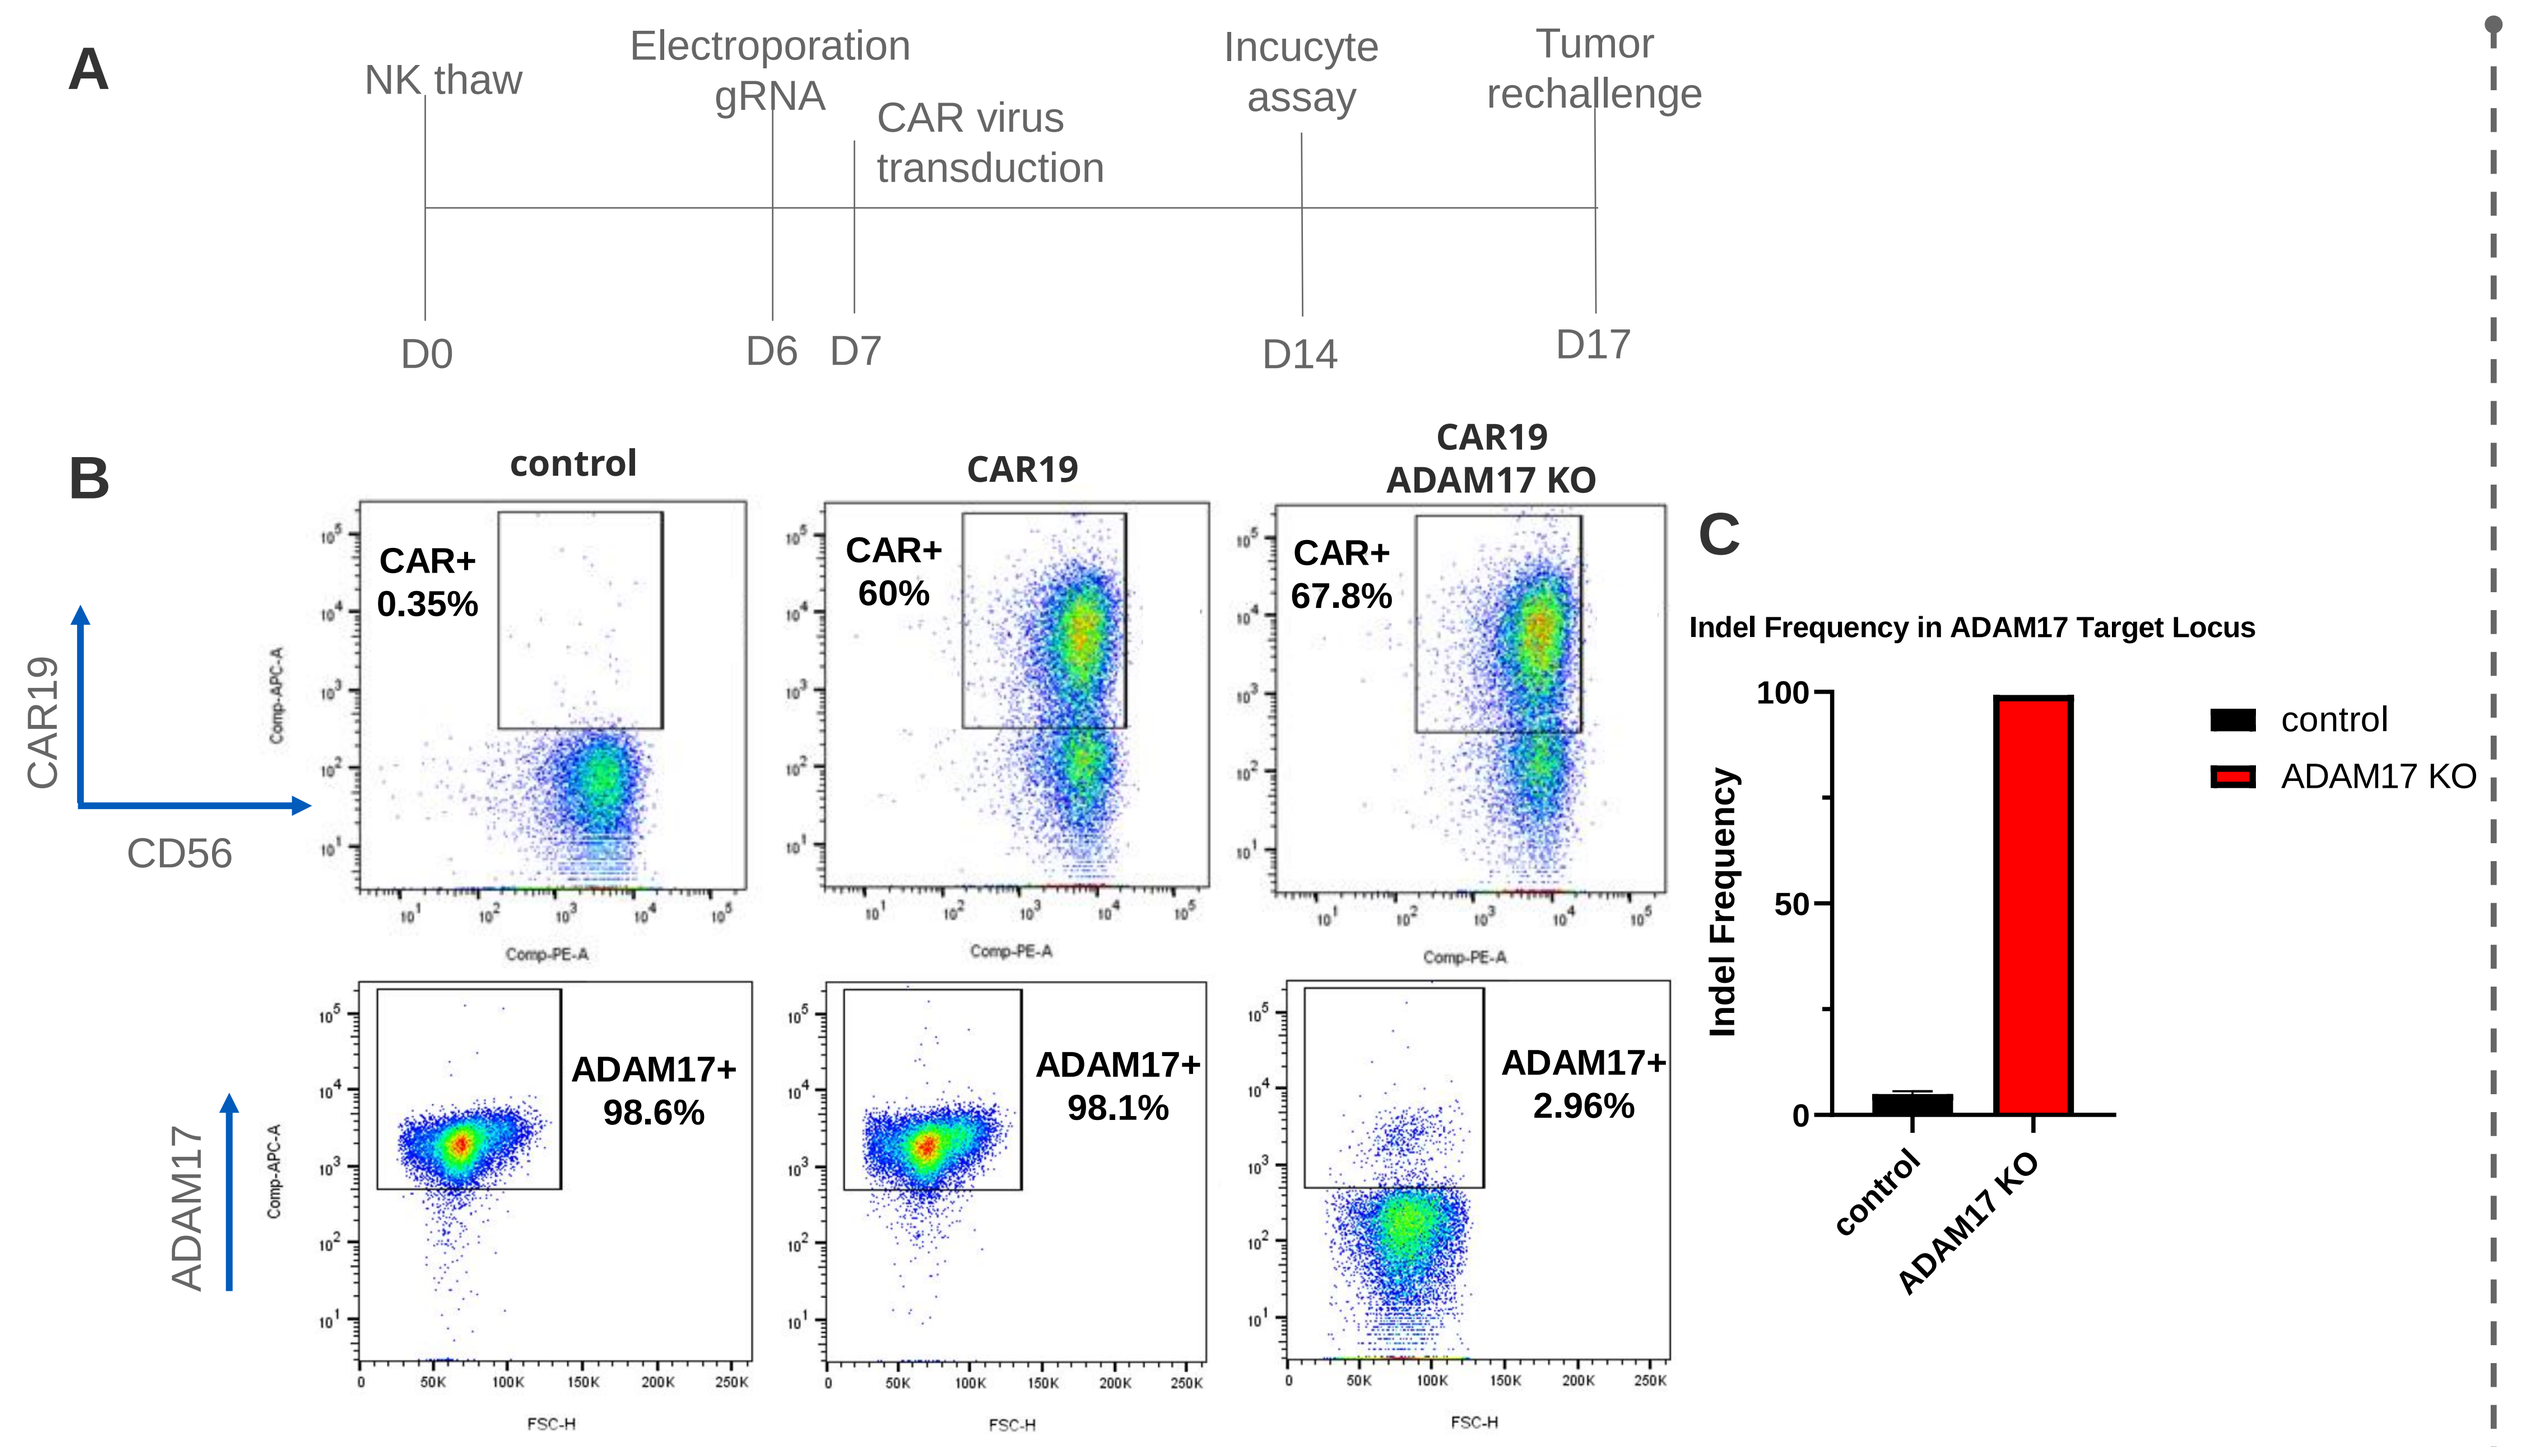
A
NK thaw
Electroporation gRNA
CAR virus transduction
Incucyte assay
Tumor rechallenge
D0
D6
D7
D14
D17
B
control
CAR19
CAR19 ADAM17 KO
CAR+ 0.35%
CAR+ 60%
CAR+ 67.8%
CAR19
CD56
ADAM17
ADAM17+ 98.6%
ADAM17+ 98.1%
ADAM17+ 2.96%
C
Indel Frequency in ADAM17 Target Locus
Indel Frequency
control
ADAM17 KO

Figure 1. Cas9-RNP mediated KO results in efficient ADAM17 deletion in primary human NK cells. (A) Schematic timeline for producing gene-knockout CAR NK cells. (B) FACS analysis to assess CAR19 expression on ADAM17 knockout NK cells (top row); ADAM17 FACS analysis evaluating knockout efficiency (bottom row). (C) Indel frequency analysis of ADAM17 KO based on targeted NGS of primary human NK cells. ADAM17 KO efficiency was approximately 97%.

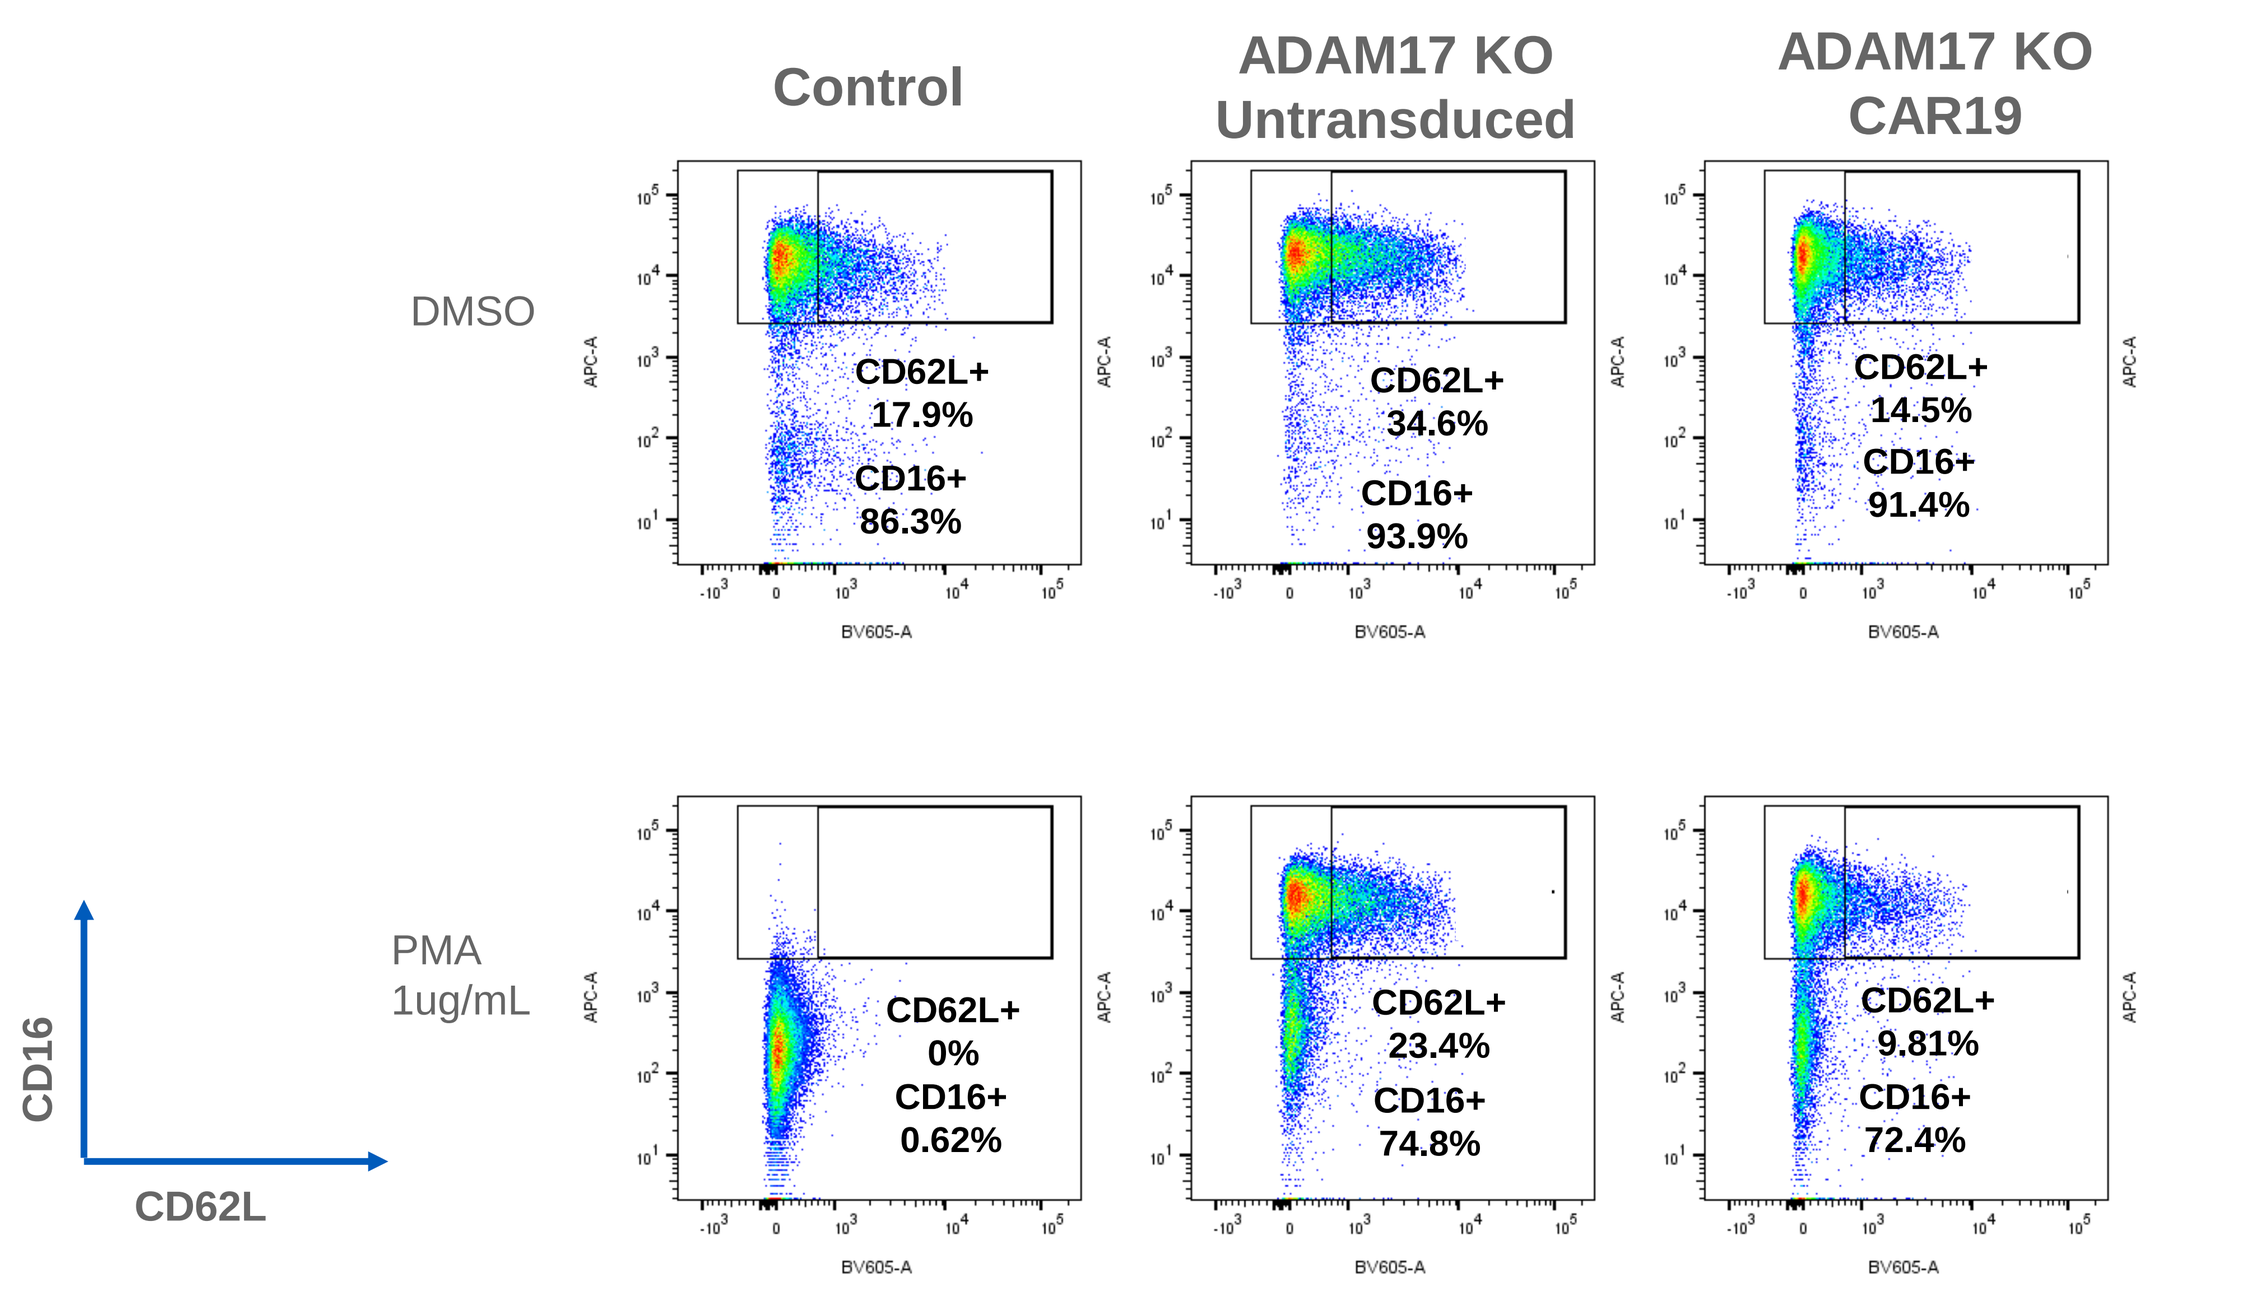
Control
ADAM17 KO Untransduced
ADAM17 KO CAR19
DMSO
PMA 1ug/mL
CD62L+ 17.9%
CD16+ 86.3%
CD62L+ 34.6%
CD16+ 93.9%
CD62L+ 14.5%
CD16+ 91.4%
CD62L+ 0%
CD16+ 0.62%
CD62L+ 23.4%
CD16+ 74.8%
CD62L+ 9.81%
CD16+ 72.4%
CD16
CD62L

Figure 2. ADAM17 KO maintains CD16 surface expression on CD19 CAR NK cells. NK cells with or without ADAM17 KO were stimulated with PMA (1 ug/mL) for 1 h, CD16a and CD62L expression were analyzed by flow cytometry.

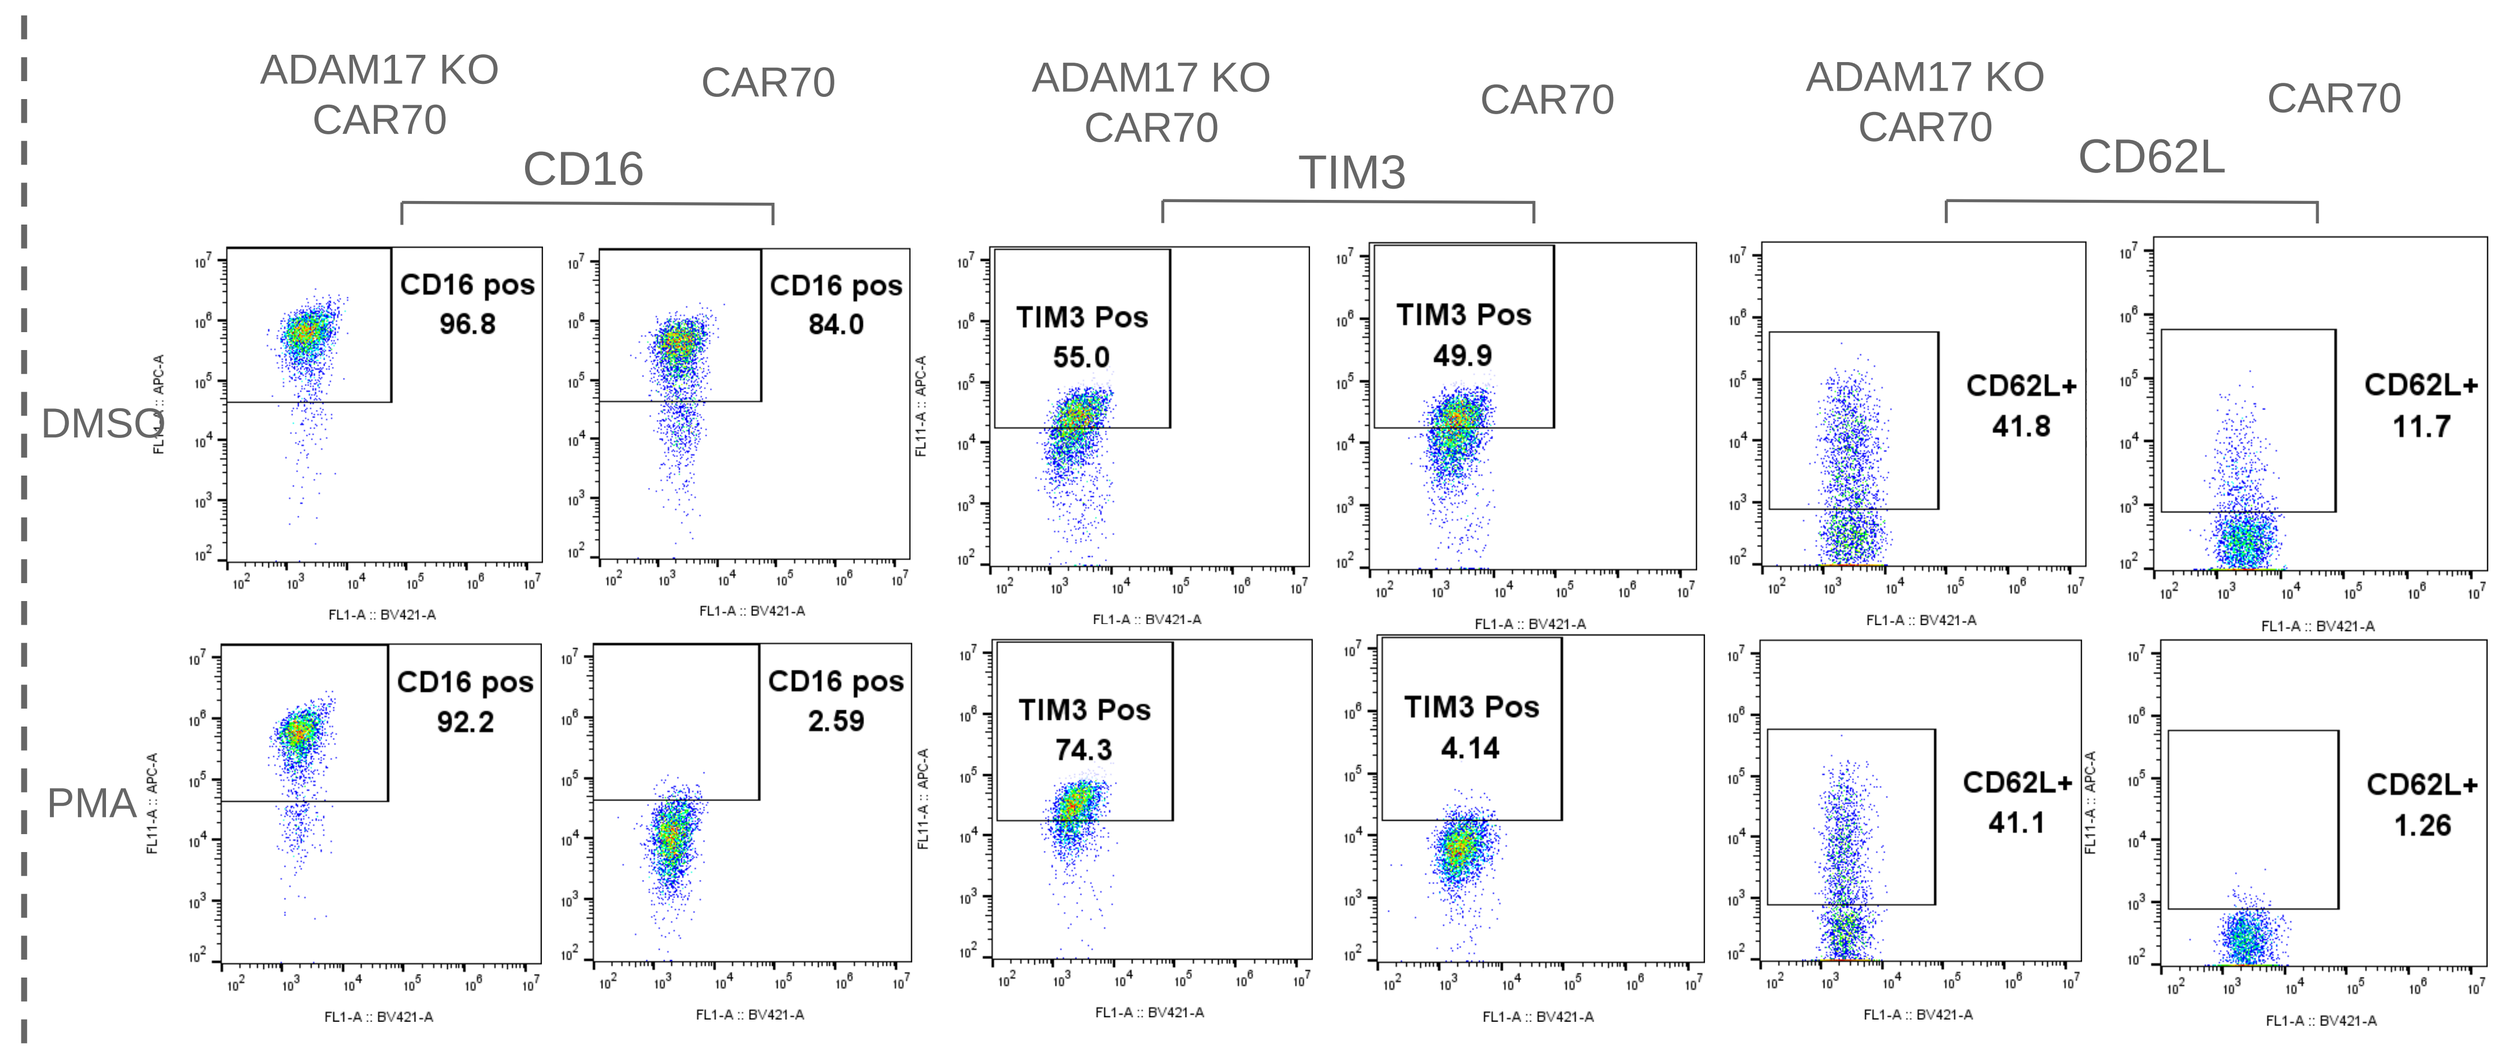
ADAM17 KO CAR70
CAR70
CD16
TIM3
CD62L
DMSO
PMA
CD16 pos 96.8
CD16 pos 84.0
CD16 pos 92.2
CD16 pos 2.59
TIM3 Pos 55.0
TIM3 Pos 49.9
TIM3 Pos 74.3
TIM3 Pos 4.14
CD62L+ 41.8
CD62L+ 11.7
CD62L+ 41.1
CD62L+ 1.26

Figure 3. ADAM17 deletion results in increased surface levels of CD62L and TIM3, which are additional substrates of ADAM17. CD70 CAR NK cells with or without ADAM17 KO were stimulated with PMA (1 ug/mL) for 1 h, CD16a, CD62L and TIM3 expression were analyzed by flow cytometry.

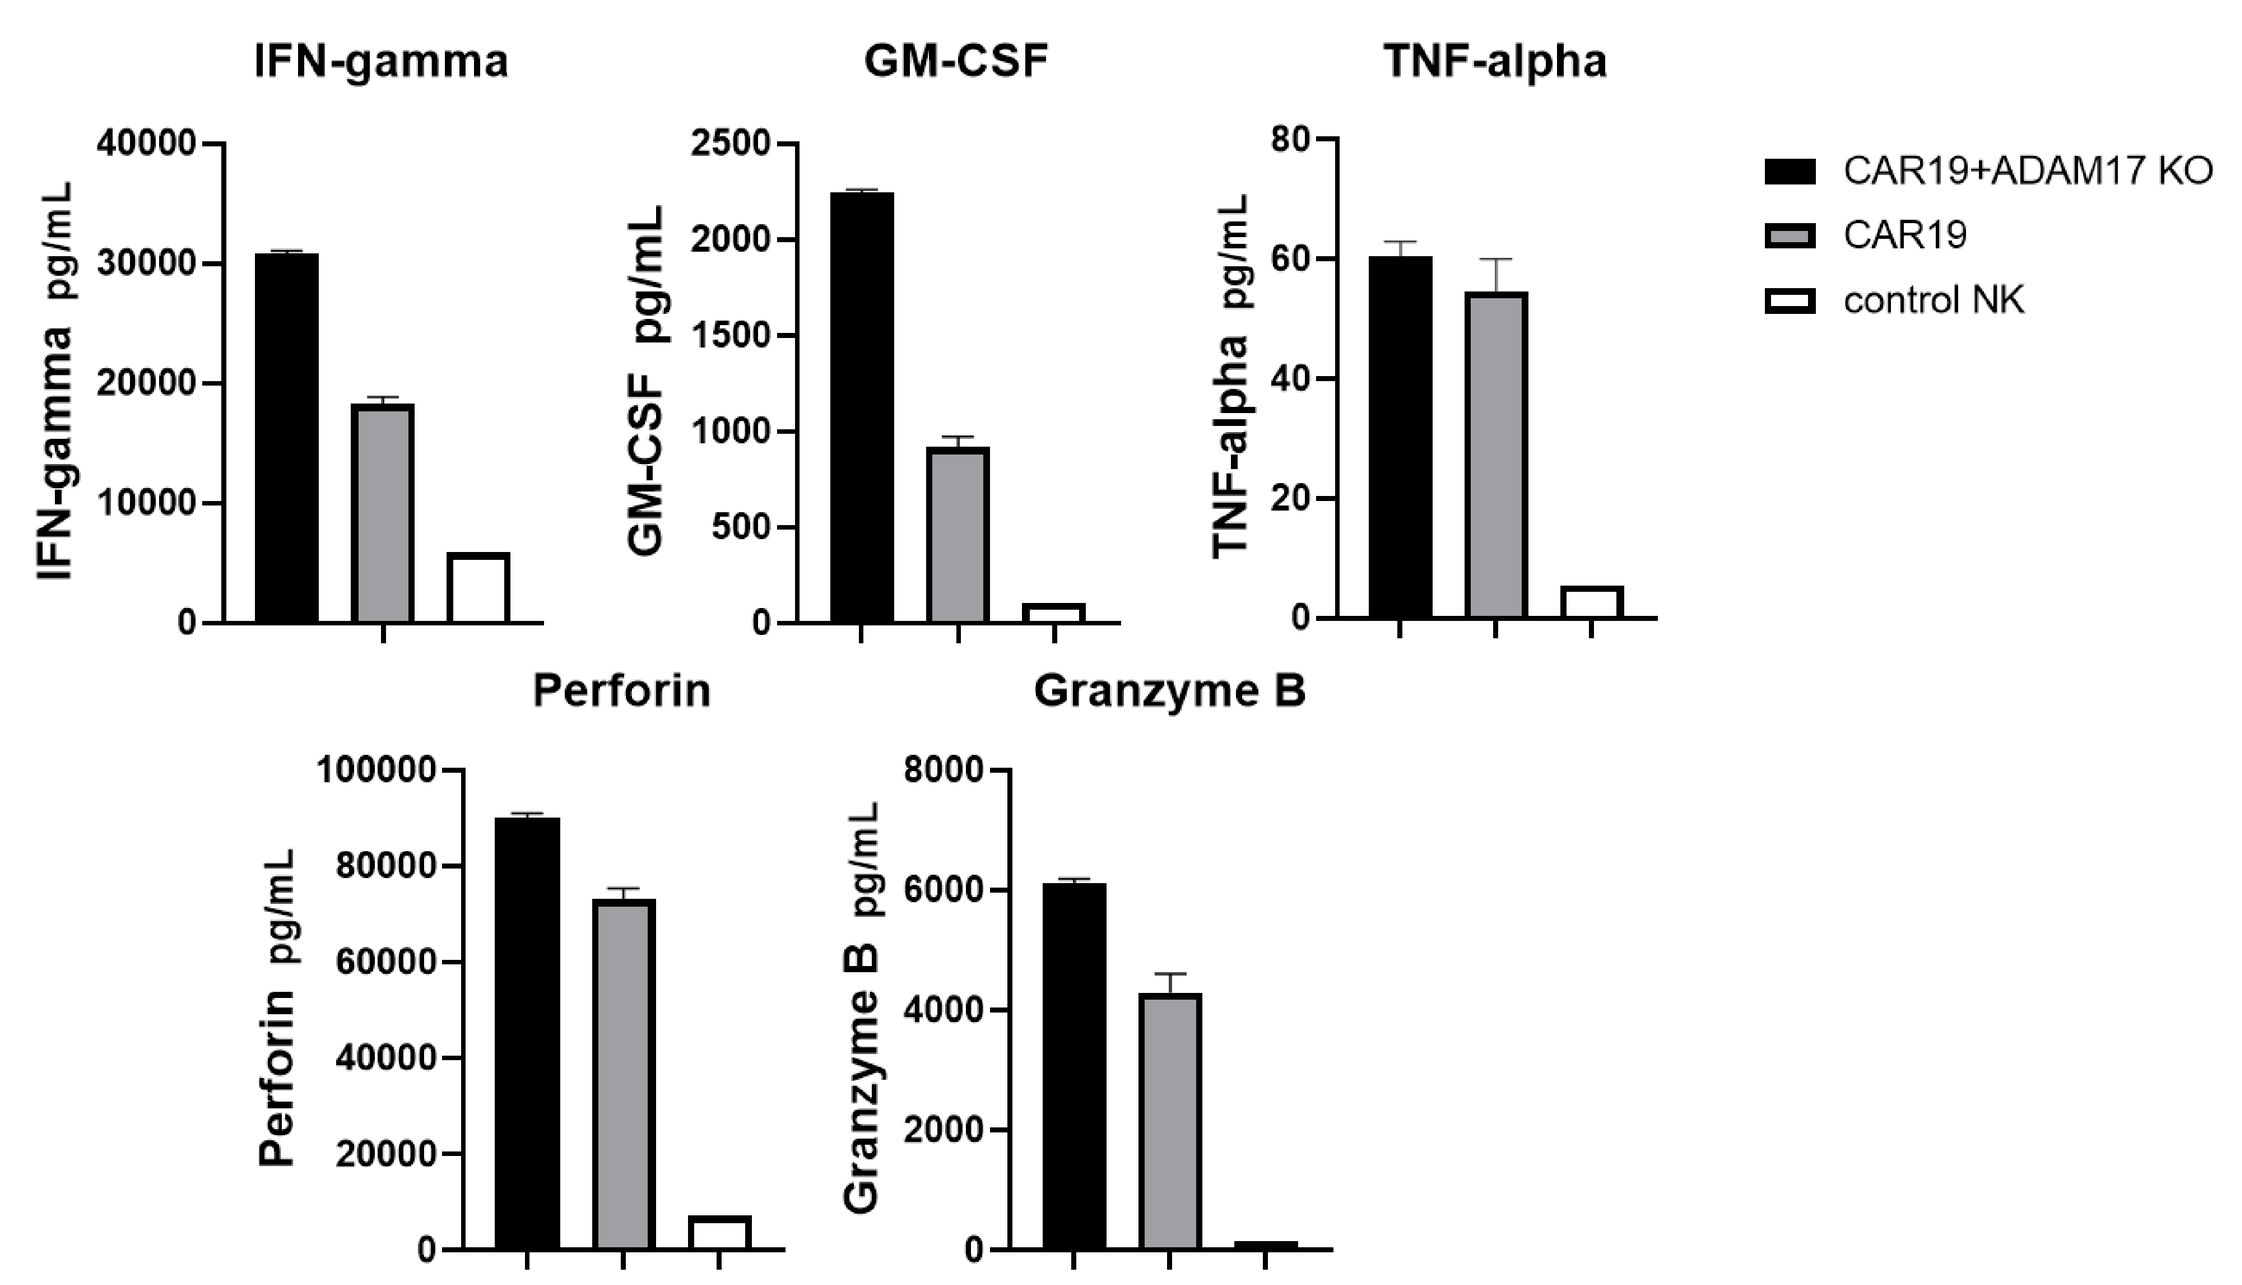
IFN-gamma
GM-CSF
TNF-alpha
Perforin
Granzyme B
IFN-gamma pg/mL
GM-CSF pg/mL
TNF-alpha pg/mL
Perforin pg/mL
Granzyme B pg/mL
CAR19+ADAM17 KO
CAR19
control NK

Figure 4. ADAM17 knockout increases secretion of cytokines and cytotoxic proteins from CAR19 NK cells. Luminex® analysis of ADAM17 KO CD19 CAR NK pro-inflammatory cytokine release in response to 72h co-culture with Raji leukemia cell line at a 1:1 E:T ratio.

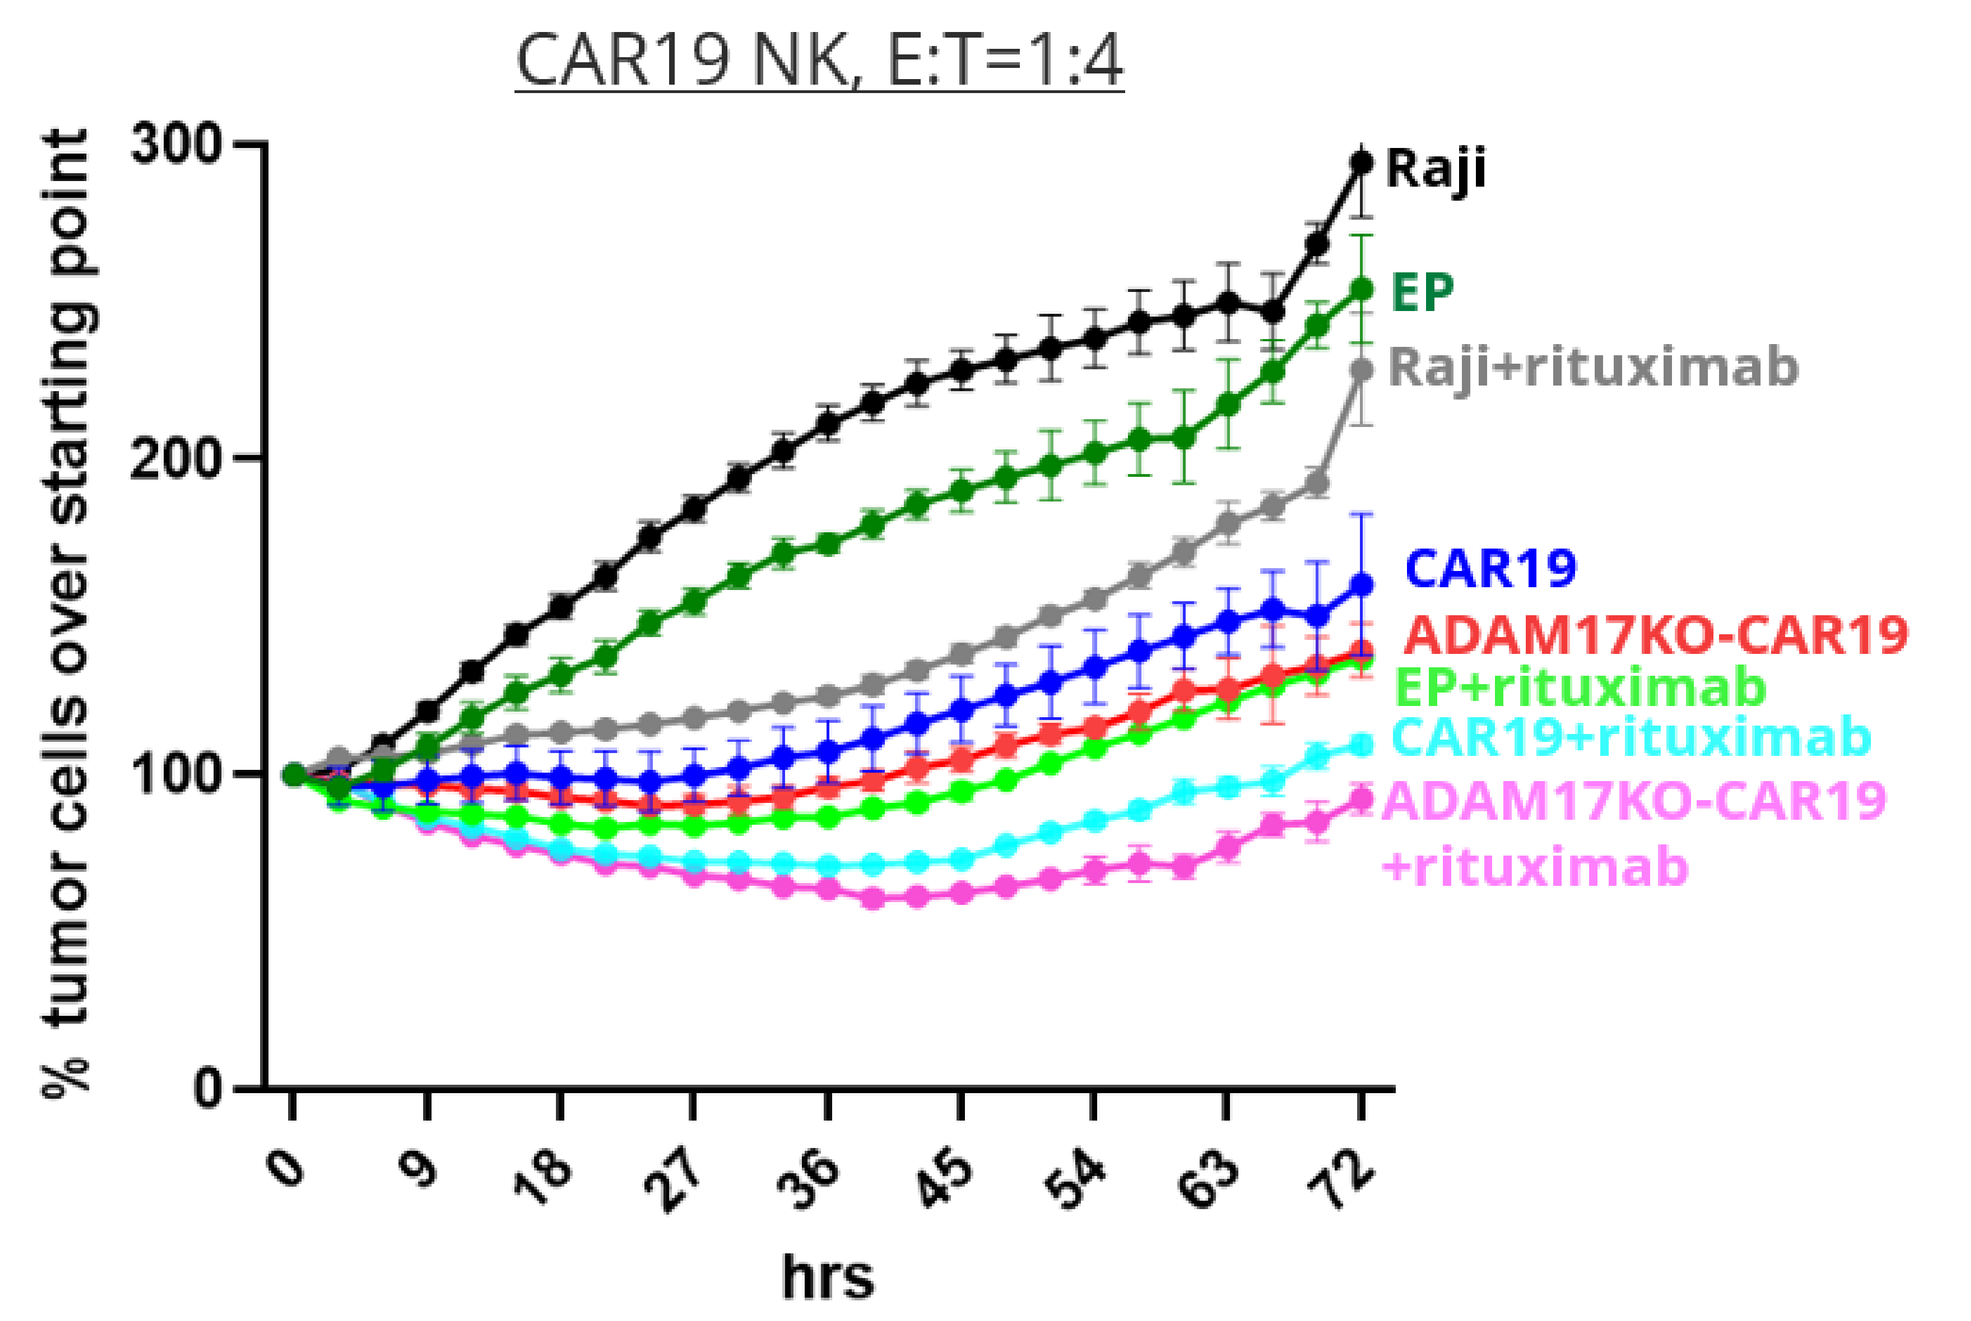
CAR19 NK, E:T=1:4
% tumor cells over starting point
hrs
Raji
EP
Raji+rituximab
CAR19
ADAM17KO-CAR19
EP+rituximab
CAR19+rituximab
ADAM17KO-CAR19 +rituximab

Figure 5. ADAM17 KO enhances combinatorial potency of CAR19 NK cells and rituximab. Cytotoxicity of ADAM17 knockout CD19 CAR NK cells against Raji cell line and co-incubated with +/- 10µg/mL of anti-CD20 rituximab at 1:4 E:T ratio via IncuCyte® S3 live cell analysis at day 14. EP: mock-electroporated and untransduced control NK cells

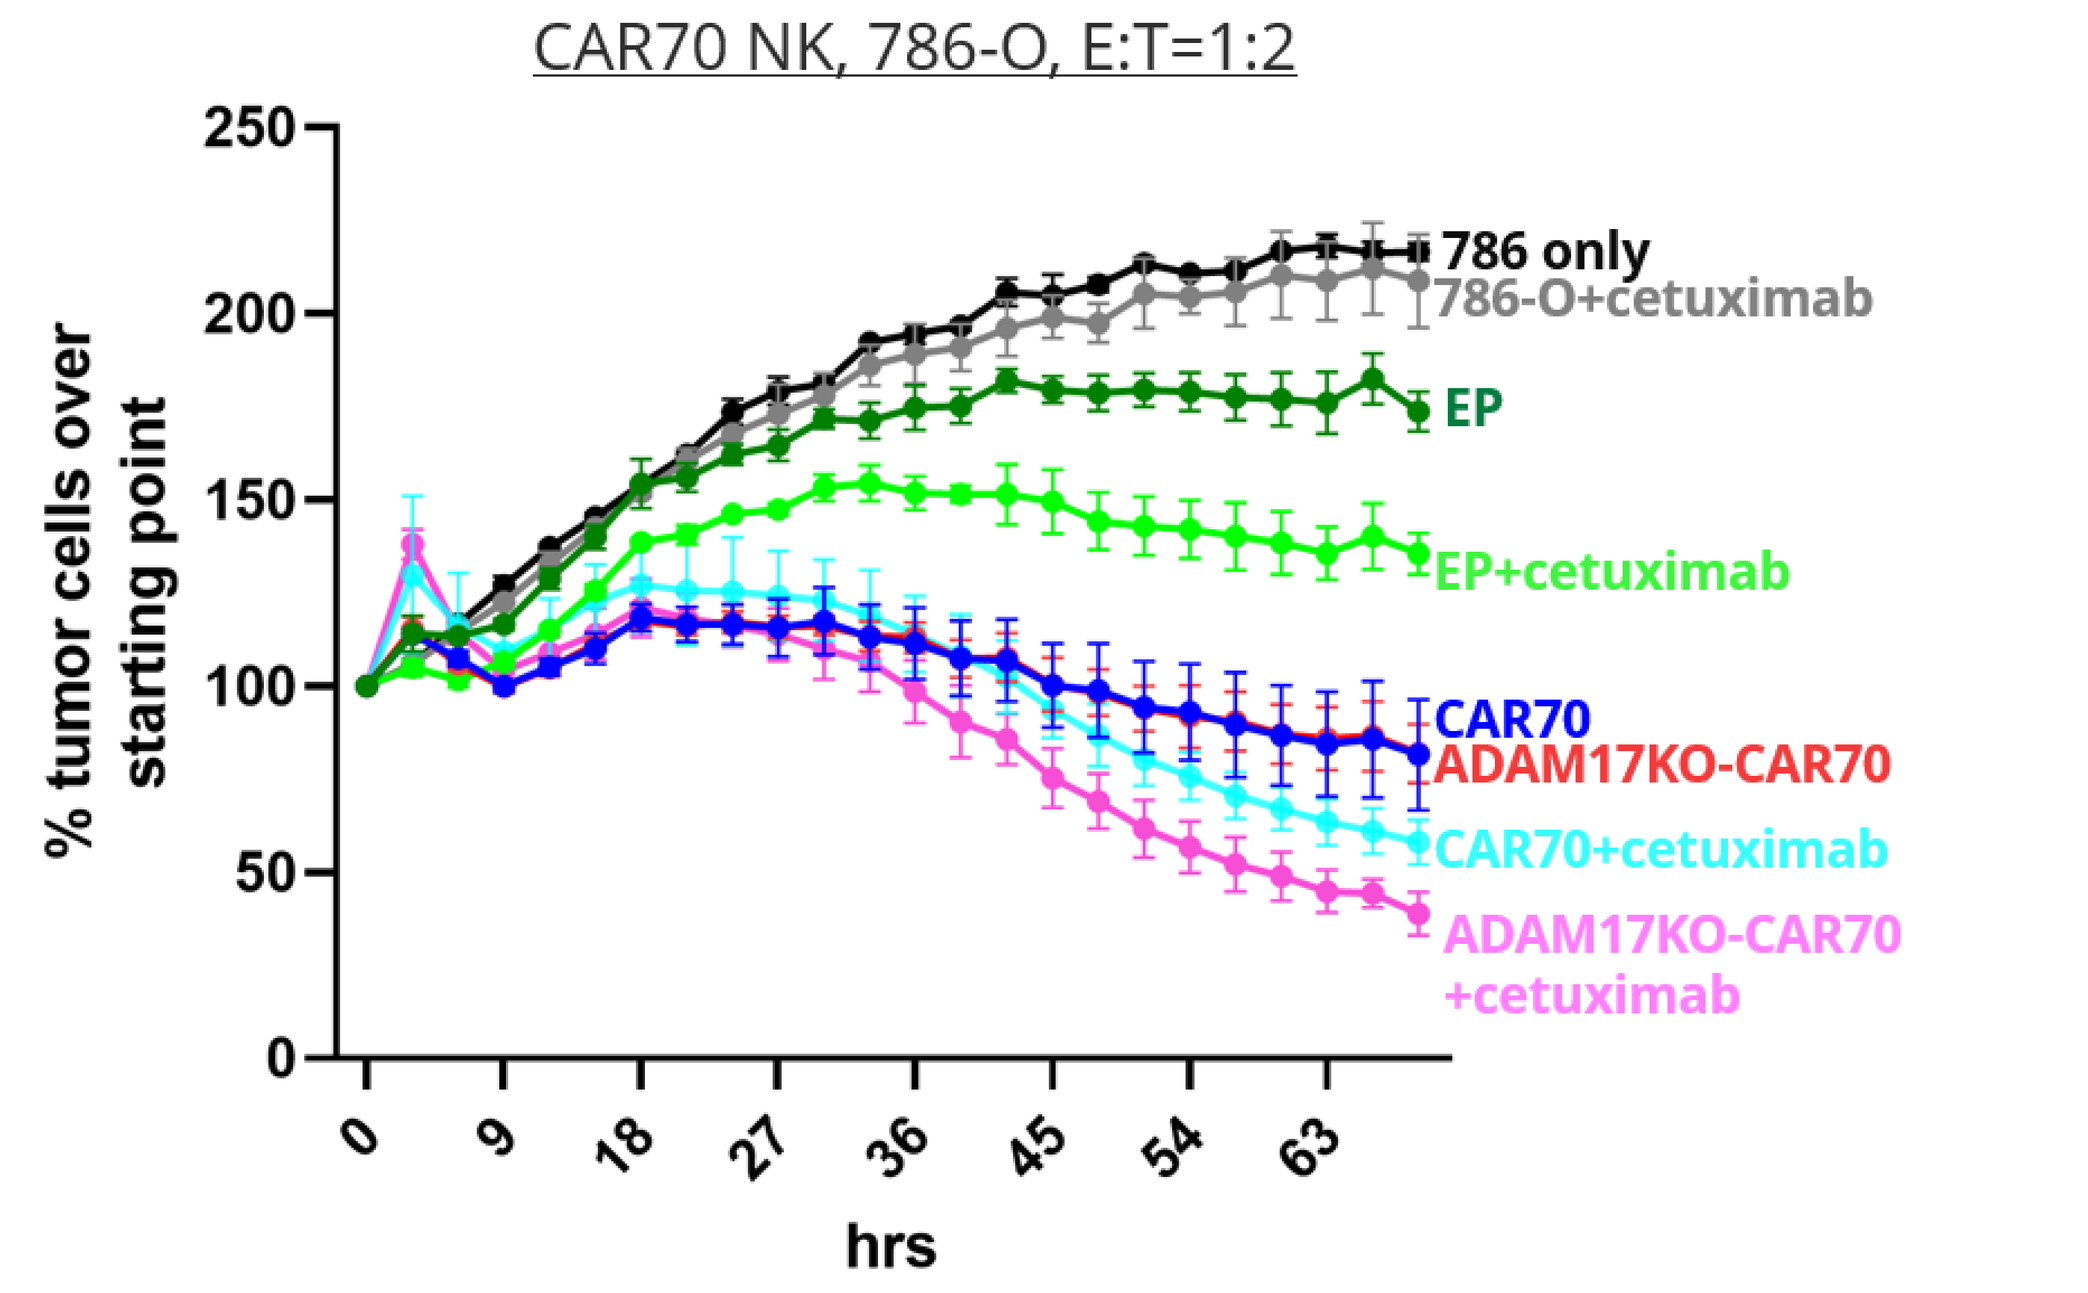
CAR70 NK, 786-O, E:T=1:2
% tumor cells over starting point
hrs
786 only
786-O+cetuximab
EP
EP+cetuximab
CAR70
ADAM17KO-CAR70
CAR70+cetuximab
ADAM17KO-CAR70 +cetuximab

Figure 6. ADAM17 KO enhances combinatorial potency of CAR70 NK cells and cetuximab.

Cytotoxicity of ADAM17 KO CD70 CAR NK cells against 786-O RCC cell line and co-incubated with +/- 2µg/mL of anti-EGFR cetuximab at 1:2 E:T ratio via IncuCyte® S3 live cell analysis at day 14. EP: mock-electroporated and untransduced control NK cells

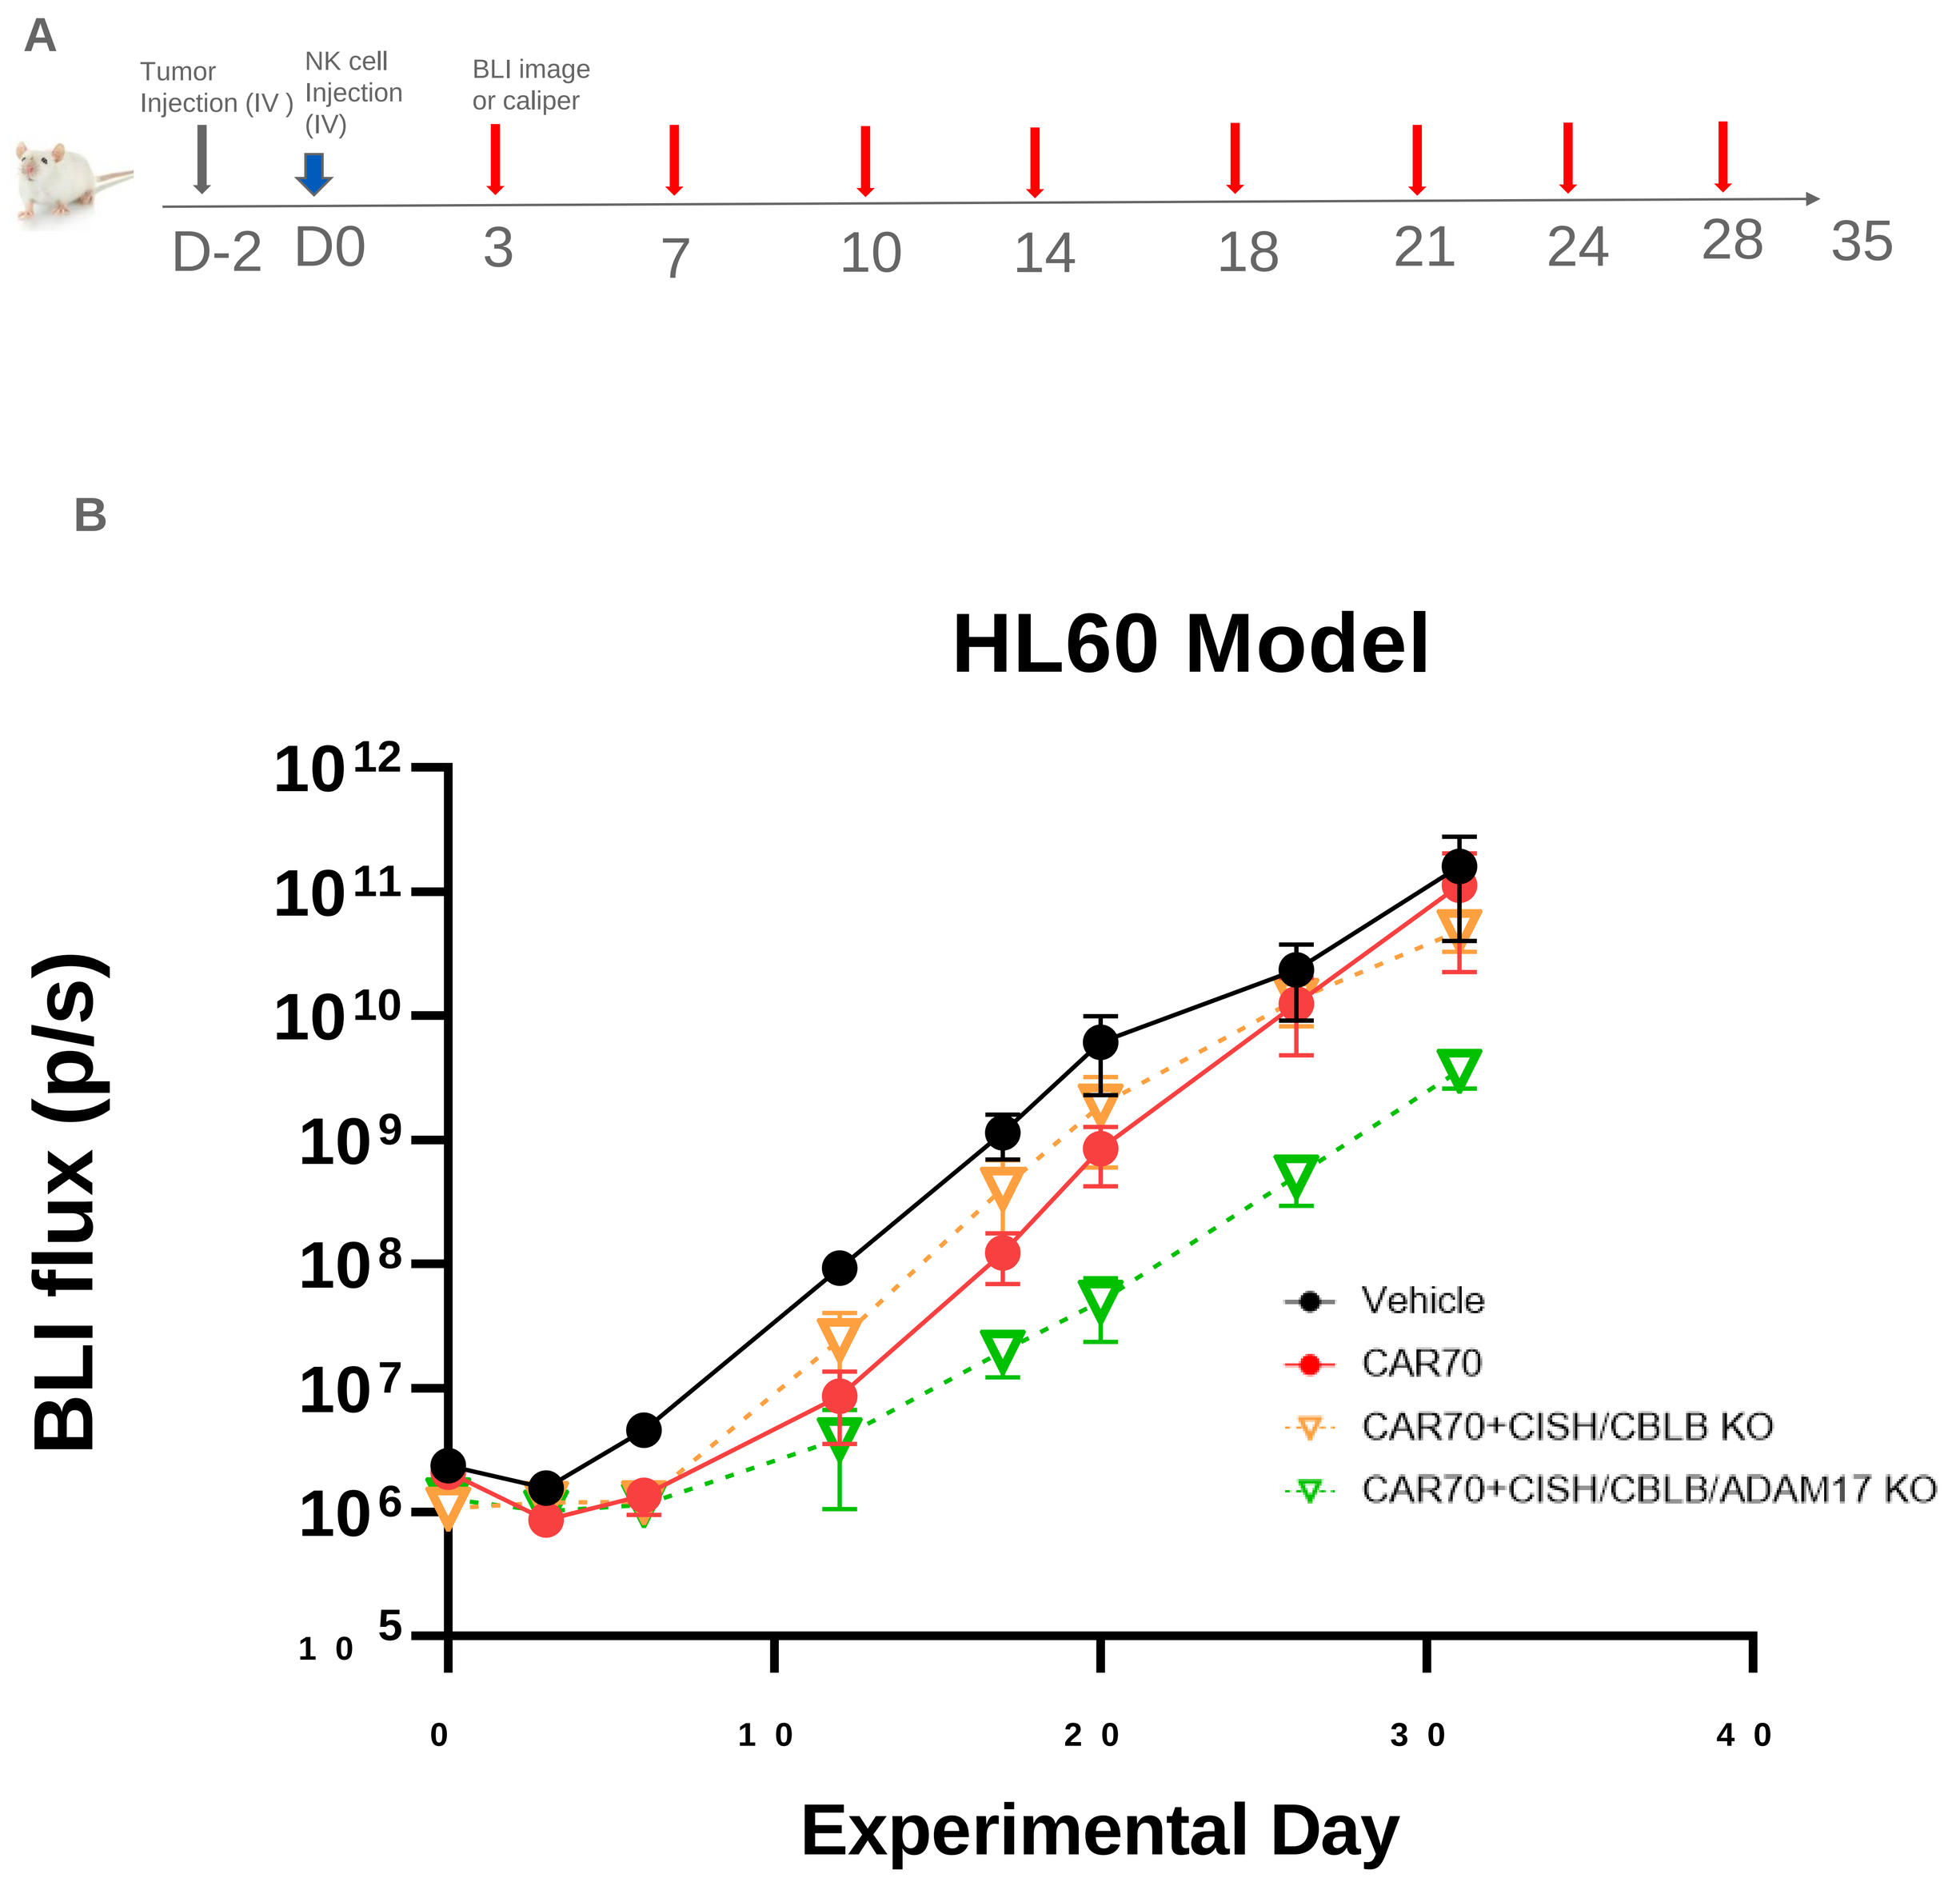
A
Tumor Injection (IV)
NK cell Injection (IV)
BLI image or caliper
D-2
D0
B
HL60 Model
BLI flux (p/s)
Experimental Day
Vehicle
CAR70
CAR70+CISH/CBLB KO
CAR70+CISH/CBLB/ADAM17 KO

Figure 7. ADAM17 KO enhances in vivo anti-tumor activity of CISH/CBLB KO CD70 CAR NK cells. (A) Schematic of NK cell treatment in a HL60 (low CD70 expression cell line) AML xenograft model (B). Tumor measurement of the mice was performed twice per week. On Day 0, the following groups received $1x10^7$ NK cells: CAR70, CAR70+CISH/CBLB KO and CAR70+CISH/CBLB/ADAM17 KO.

Conclusion

Taken together, we show ADAM17 KO NK or ADAM17 KO CAR NK cells demonstrate improved ADCC and ADAM17 KO can enhance in vivo anti-tumor activity of CISH/CBLB KO CD70 CAR NK cells against relevant tumor models. These data support the further exploration of ADAM17 gene knockout NK cells for clinical application and to improve efficacy of therapeutic antibodies in combination with adoptive transfer of engineered NK cells.

References

1. Khan et al, *Front. Immunol* 2020 Feb 13.
2. Guo et al, *Journal for ImmunoTherapy of Cancer* 2021 Nov; 9 (Suppl 2):A123-A123.
3. Buren et al, *Cancer Immunol Res* (2020) 8 (3_Supplement): B64.
4. Walcheck et al, *Expert Opinion on Biological Therapy* 2019 19:12, 1229-1232.

Contact
James Trager, PhD
jtrager@nkartatx.com
www.nkartatx.com