Combination of anti-EGFR antibody cetuximab with NKX101, an allogeneic NKG2D-L targeting NK cell therapy, enhances potency and *in vitro* cytotoxicity against solid tumors

Cynthia Cho, Max Zhang, Carmel Chan, Ph.D., Kyle Hansen, James Trager, Ph.D.

Abstract Presentation Number: 3183

Introduction

Antibody-dependent cellular cytotoxicity (ADCC) is a mechanism of cell-mediated immune defense against opsonized target cells wherein the Fc-fragment of an antibody binds to CD16a (FcγRIIIa) on immune effectors such as NK cells, triggering their activation and targeted killing. Cetuximab is a human IgG1 antibody targeting epidermal growth factor receptor (EGFR) that is approved for the treatment of colorectal and head and neck cancers. Binding of cetuximab to EGRF^{+} cell lines has been shown to trigger ADCC-mediated killing by NK cells. NKX101 is a donor-derived Natural Killer Group 2D ligand (NKG2D-L) targeting allogeneic NK cell therapy in development for the treatment of cancer. NKX101 is engineered to express an NKG2D-based chimeric antigen receptor (CAR) and membrane-bound IL-15. To investigate the combination of NKX101 and cetuximab, we developed *in vitro* assays to (i) determine the activity of NKX101 against cancer cells via CD16-mediated ADCC, (ii) compare ADCC activity of NKX101 cells generated from high-affinity CD16 and low-affinity CD16 donors, and (iii) evaluate the synergistic interaction of NKX101 combined with cetuximab.

Methods

NKX101 cells or untransduced NK cell controls were generated from peripheral blood drawn from six healthy donors. NucRed™ labeled EGFR^{+} NKG2D-L^{+} human cancer cell lines were pre-incubated with cetuximab or isotype control antibody and co-cultured with NKX101. Specific cell killing of target cells was measured using an Incucyte® instrument. Potency was determined using IC_{50} values generated from four-parameter dose-response curves in GraphPad Prism. For synergy assessments, a dose response matrix of NKX101 cells and cetuximab was used to generate cytotoxicity values. Synergy scores for combinations were determined using the SynergyFinder Plus web application (https://synergyfinderplus.org/). To determine donor CD16 V/F 158 genotypes, genomic DNA was isolated from each donor and assayed using a TaqMan™ CD16 genotyping kit.

Expression of EGFR and NKG2D-L by flow cytometry

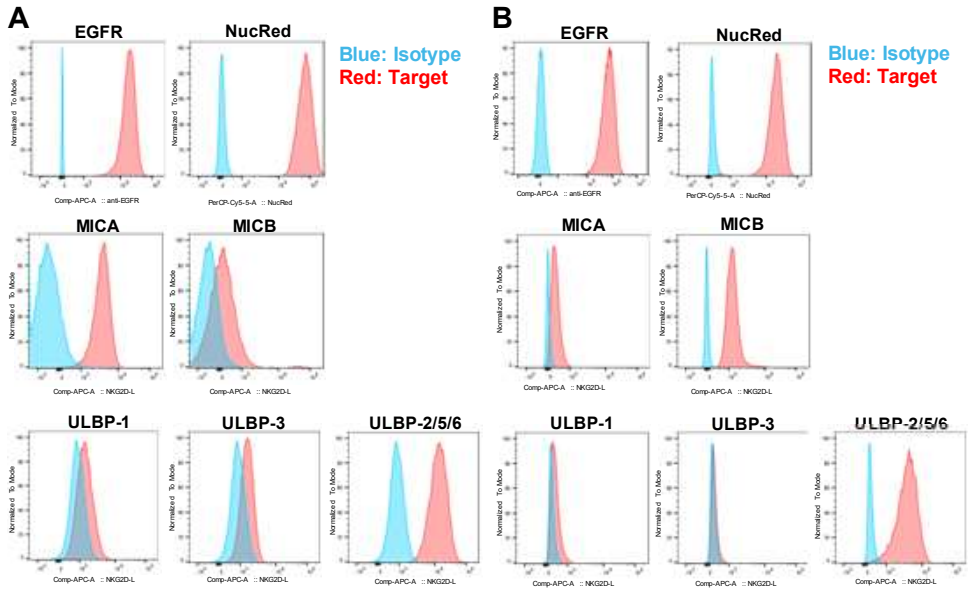
A
EGFR
NucRed
Blue: Isotype
Red: Target
MICA
MICB
ULBP-1
ULBP-3
ULBP-2/5/6
B
EGFR
NucRed
Blue: Isotype
Red: Target
MICA
MICB
ULBP-1
ULBP-3
ULBP-2/5/6

Figure 1. A) NKG2D ligand expression on A-431 NucRed™ labelled epidermoid carcinoma target cell line. **B)** NKG2D ligand expression on NCI-H2228 NucRed™ labelled non-small cell lung cancer target cell line.

Results

To assess the effect of NKX101 in combination with cetuximab, we generated NucRed™ labeled human cancer cell lines that expressed both EGFR and NKG2D ligands. Evaluation of the interaction between NKX101 and cetuximab *in vitro* revealed that the two agents can combine to potently kill EGFR^{+} A-431 tumor cells in a synergistic manner. NKX101 combined with cetuximab produced a nearly 8-fold increase in potency compared to untransduced control NK cells alone. Blocking CD16 on NKX101 cells with a neutralizing antibody decreased the potency of the combination by 48%, suggesting that the ADCC effect observed was CD16-mediated. To determine if common CD16 polymorphisms influence NKX101 ADCC, we compared NKX101 cells generated from CD16 158 V/V, V/F, and F/F genotype donors. No significant differences in potency were observed with NKX101 cells expressing the high affinity CD16 158 V/V genotype compared with those derived from the low affinity 158 CD16 V/F or 158 F/F donors. Improvements in potency with the addition of cetuximab were observed for all 6 donors tested, ranging from an increase of ~10% to an increase of ~40%. We next employed a dose-response matrix to assess synergy scores of NKX101 combined with cetuximab for all 6 donors. HSA and Loewe mean synergy scores ranged from ~2.0 to ~12.5 for each donor, further supporting the ability of NKX101 cells to combine with cetuximab in a synergistic manner.

Cetuximab enhances NKX101 *in vitro* anti-tumor killing through ADCC activity

A

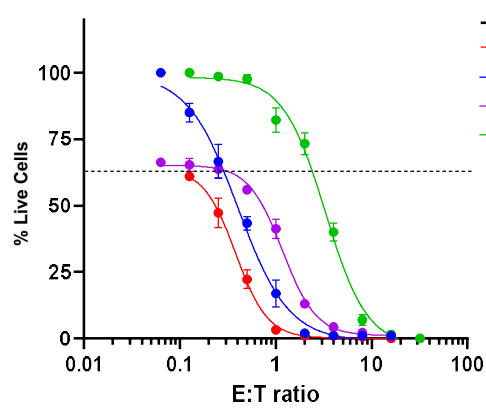
cetuximab alone
NKX101 + cetuximab
NKX101 + isotype
UT NK + cetuximab
UT NK + isotype
% Live Cells
E:T ratio

Condition	% Increase in Potency Over UT + Isotype
NKX101 + cetuximab	758%
NKX101 + isotype	712%
UT + cetuximab	180%

B

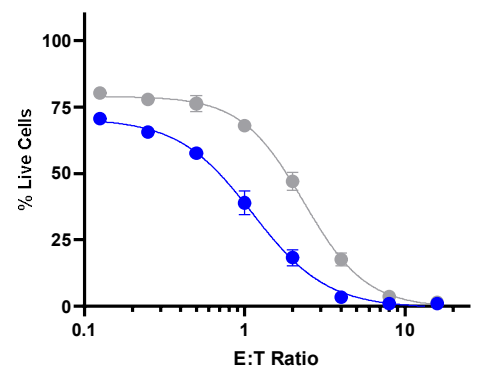
NKX101 + cetuximab + isotype
NKX101 + cetuximab + anti-CD16
% Live Cells
E:T Ratio

Condition	% Decrease in Potency Compared to Isotype Control
NKX101 + cetuximab + anti-CD16	48%

Figure 2. A) Specific killing of A-431 tumor cells *in vitro* by NKX101 or untransduced NK cell controls (UT). Cells were combined with either 10 ug/mL cetuximab or 10 ug/mL hIgG1 isotype control antibody. **B)** Specific killing of A-431 cells with antibody blockade of CD16. NKX101 cells were pre-incubated with 20 ug/mL anti-CD16 or 20 ug/mL mIgG1 isotype antibody and combined with cetuximab.

CD16 expression on NKX101 cells from 6 donors

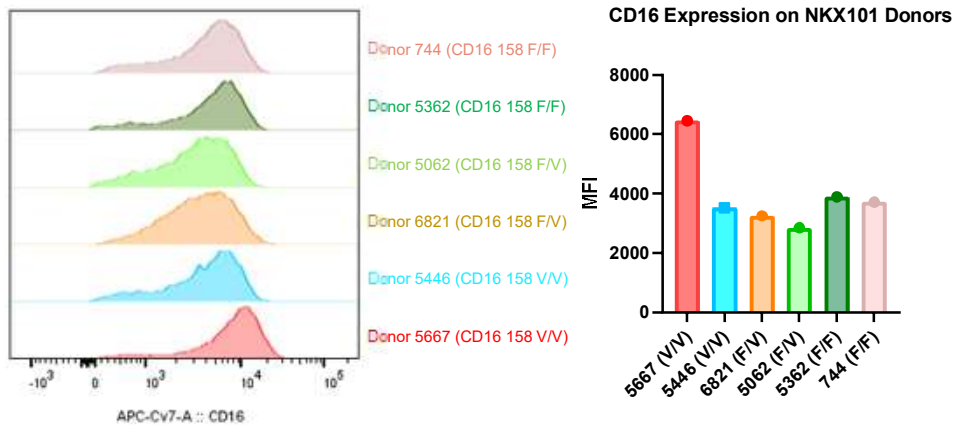
Donor 744 (CD16 158 F/F)
Donor 5362 (CD16 158 F/F)
Donor 5062 (CD16 158 F/V)
Donor 6821 (CD16 158 F/V)
Donor 5446 (CD16 158 V/V)
Donor 5667 (CD16 158 V/V)
APC-Cy7-A :: CD16
CD16 Expression on NKX101 Donors
MFI
5667 (V/V)
5446 (V/V)
6821 (F/V)
5062 (F/V)
5362 (F/F)
744 (F/F)

Figure 3. CD16 expression on NKX101 donors. Mean fluorescent intensity (MFI) was measured by flow cytometry analysis.

No significant differences in potency observed between high affinity CD16 V158 donors compared to low affinity CD16 V158 donors

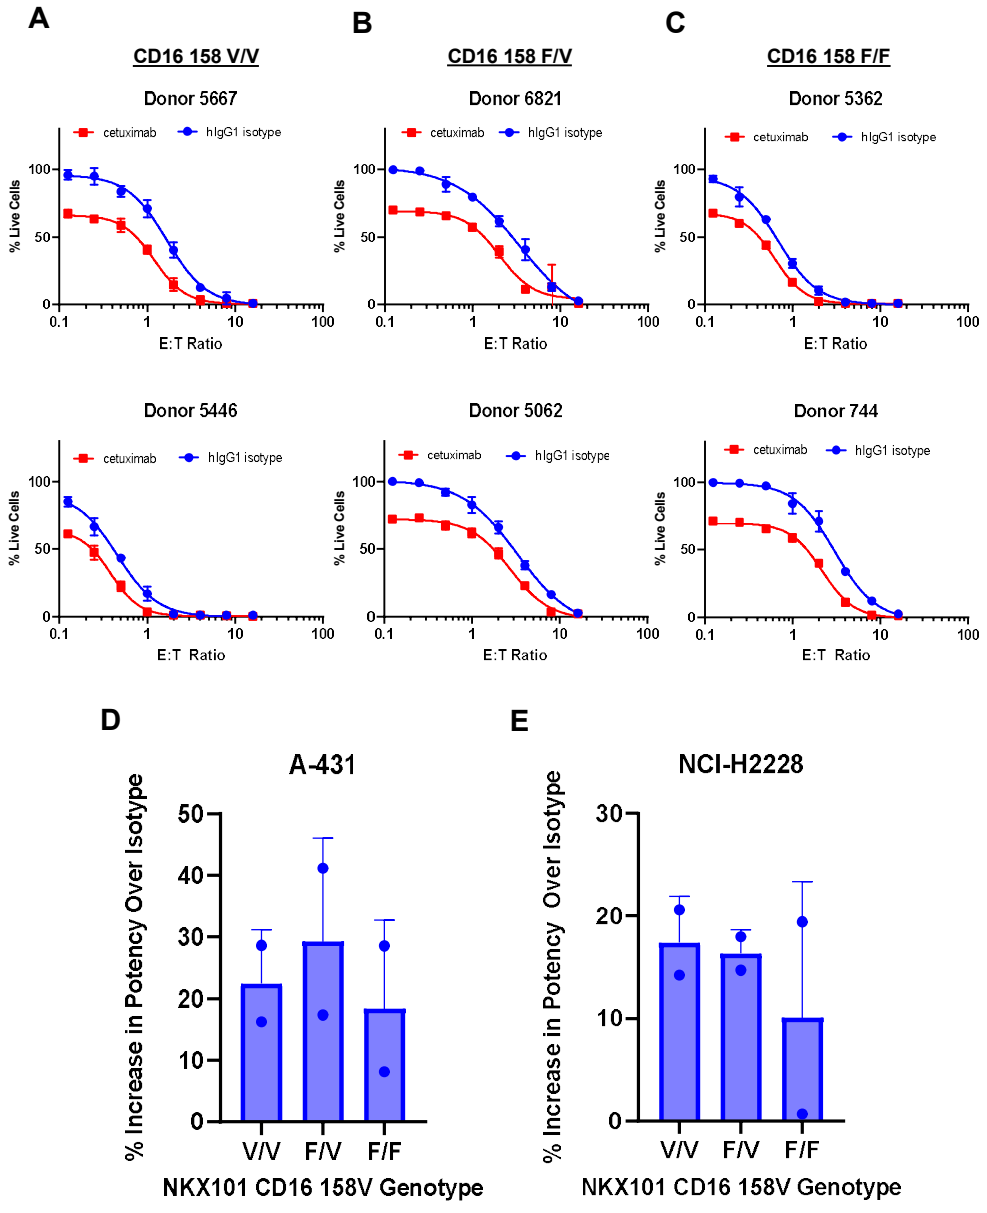
A
CD16 158 V/V
Donor 5667
Donor 5446
B
CD16 158 F/V
Donor 6821
Donor 5062
C
CD16 158 F/F
Donor 5362
Donor 744
cetuximab
hIgG1 isotype
% Live Cells
E:T Ratio
D
A-431
E
NCI-H2228
% Increase in Potency Over Isotype
V/V
F/V
F/F
NKX101 CD16 158V Genotype

Figure 4. A-C) IC_{50} curves from specific killing of A-431 cells cultured with NKX101 cells in combination with 10 ug/mL cetuximab or hIgG1 isotype. NKX101 cells were generated from donors with high-affinity CD16 V/V genotype (A), lower-affinity CD16 V/F genotype (B), and low-affinity CD16 F/F genotype (C). **D)** Percent increase in killing potency over isotype of NKX101 cells with cetuximab when co-cultured with A-431 tumor cells. **E)** Percent killing potency increase of NKX101 cells with cetuximab when co-cultured with NCI-H2228 tumor cells.

NKX101 and cetuximab combine to kill cancer cells in a synergistic manner

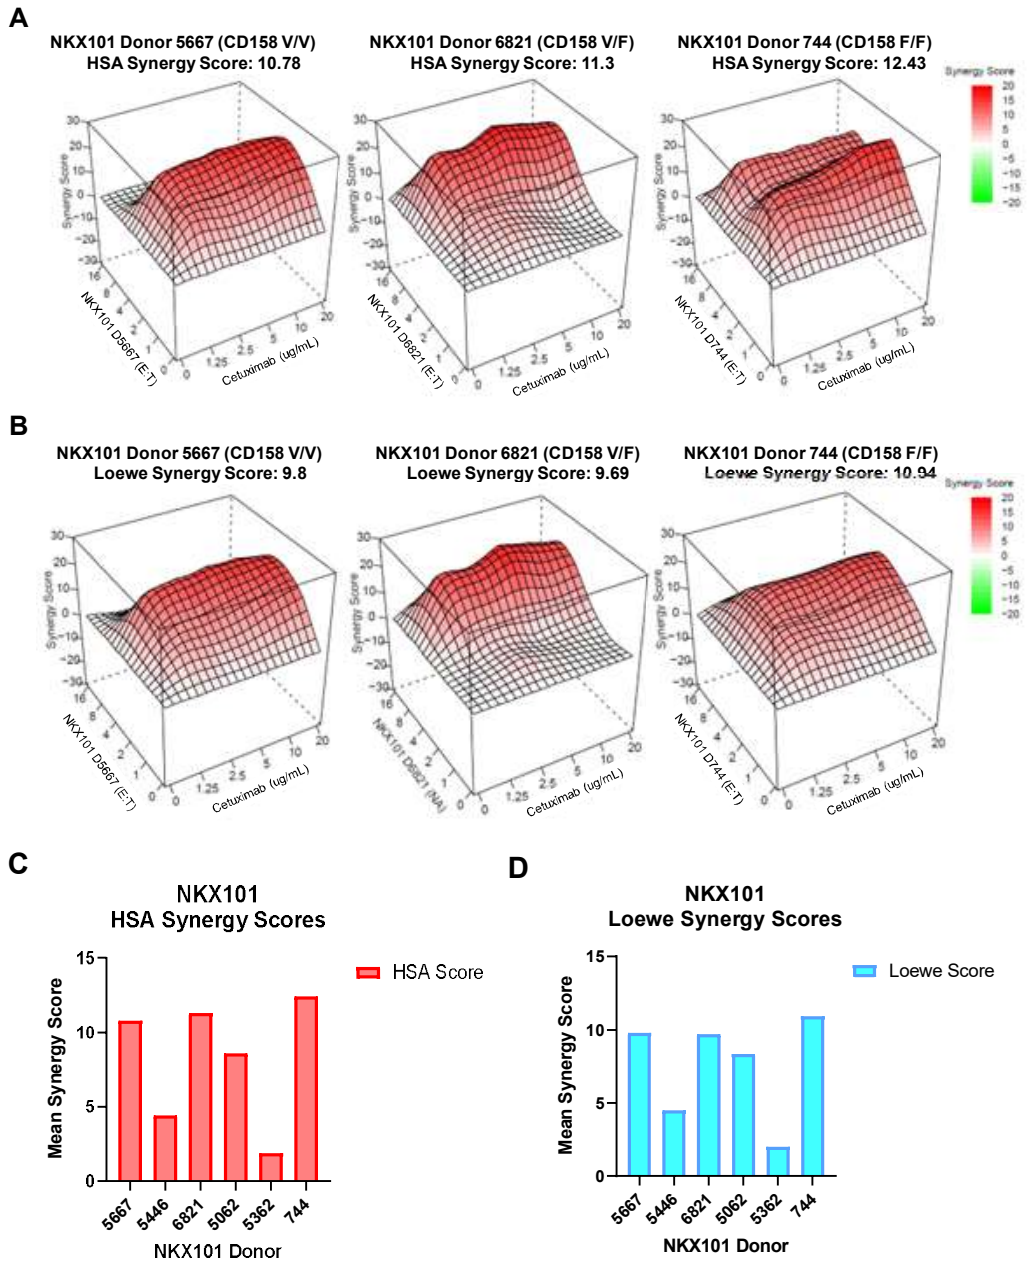
A
NKX101 Donor 5667 (CD158 V/V)
HSA Synergy Score: 10.78
NKX101 Donor 6821 (CD158 V/F)
HSA Synergy Score: 11.3
NKX101 Donor 744 (CD158 F/F)
HSA Synergy Score: 12.43
B
NKX101 Donor 5667 (CD158 V/V)
Loewe Synergy Score: 9.8
NKX101 Donor 6821 (CD158 V/F)
Loewe Synergy Score: 9.69
NKX101 Donor 744 (CD158 F/F)
Loewe Synergy Score: 10.94
Cetuximab (ug/mL)
C
NKX101
HSA Synergy Scores
HSA Score
D
NKX101
Loewe Synergy Scores
Loewe Score
Mean Synergy Score
NKX101 Donor

Figure 5. A-B) HSA and Loewe synergy maps of a dose-response matrix for the specific killing of A-431 cells by NKX101 combined with cetuximab. **C)** HSA synergy scores for all 6 donors tested. **D)** Loewe synergy scores for all 6 donors tested.

Conclusion

NKX101 cells carry out ADCC *in vitro* when used in combination with cetuximab, enhancing NKX101 cytotoxicity against solid tumor-derived cell lines. NKX101 in combination with cetuximab led to increased killing potency compared to NKX101 alone or untransduced NK cells plus cetuximab. Utilizing dose-response matrices and the SynergyFinder Plus web application, we determined that NKX101 can combine synergistically with cetuximab to eliminate EGFR^{+} cancer cells. Notably, we found no significant differences in anti-tumor activity when cetuximab was combined with NKX101 cells generated from donors with high or low affinity CD16 polymorphisms, suggesting that ADCC mediated by NKX101 and cetuximab is not strongly influenced by donor CD16 genotype. In summary, we show the combination of an engineered allogeneic NK cell therapy, NKX101, with cetuximab leads to an increase in the specific killing of cancer cells *in vitro*, supporting further investigation into this combination for the treatment of solid tumors.

Contact

James Trager, PhD
jtrager@nkartatx.com
www.nkartatx.com