A combined strategy of CD70 CAR co-expression with membrane bound IL-15 and CISH knockout results in enhanced NK cytotoxicity and persistence

Luxuan Buren[1], Yanying Fan[1], Meriam Vejiga[1], Chao Guo[1], Yu Jeong Kim[1], Alexander Aronov[1], Don-Hong Wang[1], Ming-Hong Xie[1], Katherine Jamboretz[1], Sasha Lazetic[1], Ivan H. Chan[1], James Trager[1]

Jointly presented by Nkarta Therapeutics and CRISPR Therapeutics

[1]Nkarta Inc, South San Francisco, CA, USA

Introduction

Chimeric antigen receptors (CARs) have been used successfully to retarget T cells in patients with hematologic malignancies. Natural killer (NK) cells offer an alternative to T cells for cellular immunotherapy. NK cells are well-suited for allogeneic use as they are not HLA-restricted and patients that receive NK cell treatment do not develop graft-versus-host disease (GVHD). Further, NK cells can be highly expanded, engineered, edited, and cryopreserved, thus lending themselves to 'off-the-shelf' production of a potent, targeted cell therapies.

Here, we investigated multiple approaches to modify NK anti-tumor targeting and function via CD70 targeted chimeric antigen receptor (CAR) expression and CISH gene knockout (KO). CD70 (CD27 ligand) is highly expressed in multiple hematological malignancies and in solid tumors, such as renal cell carcinoma. CISH, cytokine-inducible SH2 containing protein, is a critical checkpoint regulator in NK cell-mediated tumor immunity. We show that a combined strategy of CD70 CAR engineering with CISH KO in NK cells resulted in superior anti-tumor potency.

Methods

An scFv library was screened to identify CD70 binders as illustrated in Figure 1A. 74 CD70 binders were tested in a Jurkat cell screen to evaluate CAR expressions, tonic signaling and activation (Figure 1B-C). CAR candidates were selected based in part on the activation to tonic signal ratio (Figure 1D) in addition to CAR expression. The top 10 binders from the Jurkat screen were further characterized in primary NK cells. For NK expansion, NK cells were generated by co-culture of purified NK cells from healthy donors with genetically modified irradiated K562 feeder cells. CISH KO and CD70 KO were performed using CRISPR/Cas9 ribonucleoprotein (RNP) complexes. NK cells were transduced with a gamma-retrovirus encoding a CD70 CAR and membrane bound IL-15 (mbIL-15). CAR expression was evaluated by flow cytometry. *In vitro* cytotoxicity of transduced NK cells was measured using the IncuCyte S3 live cell analysis system.

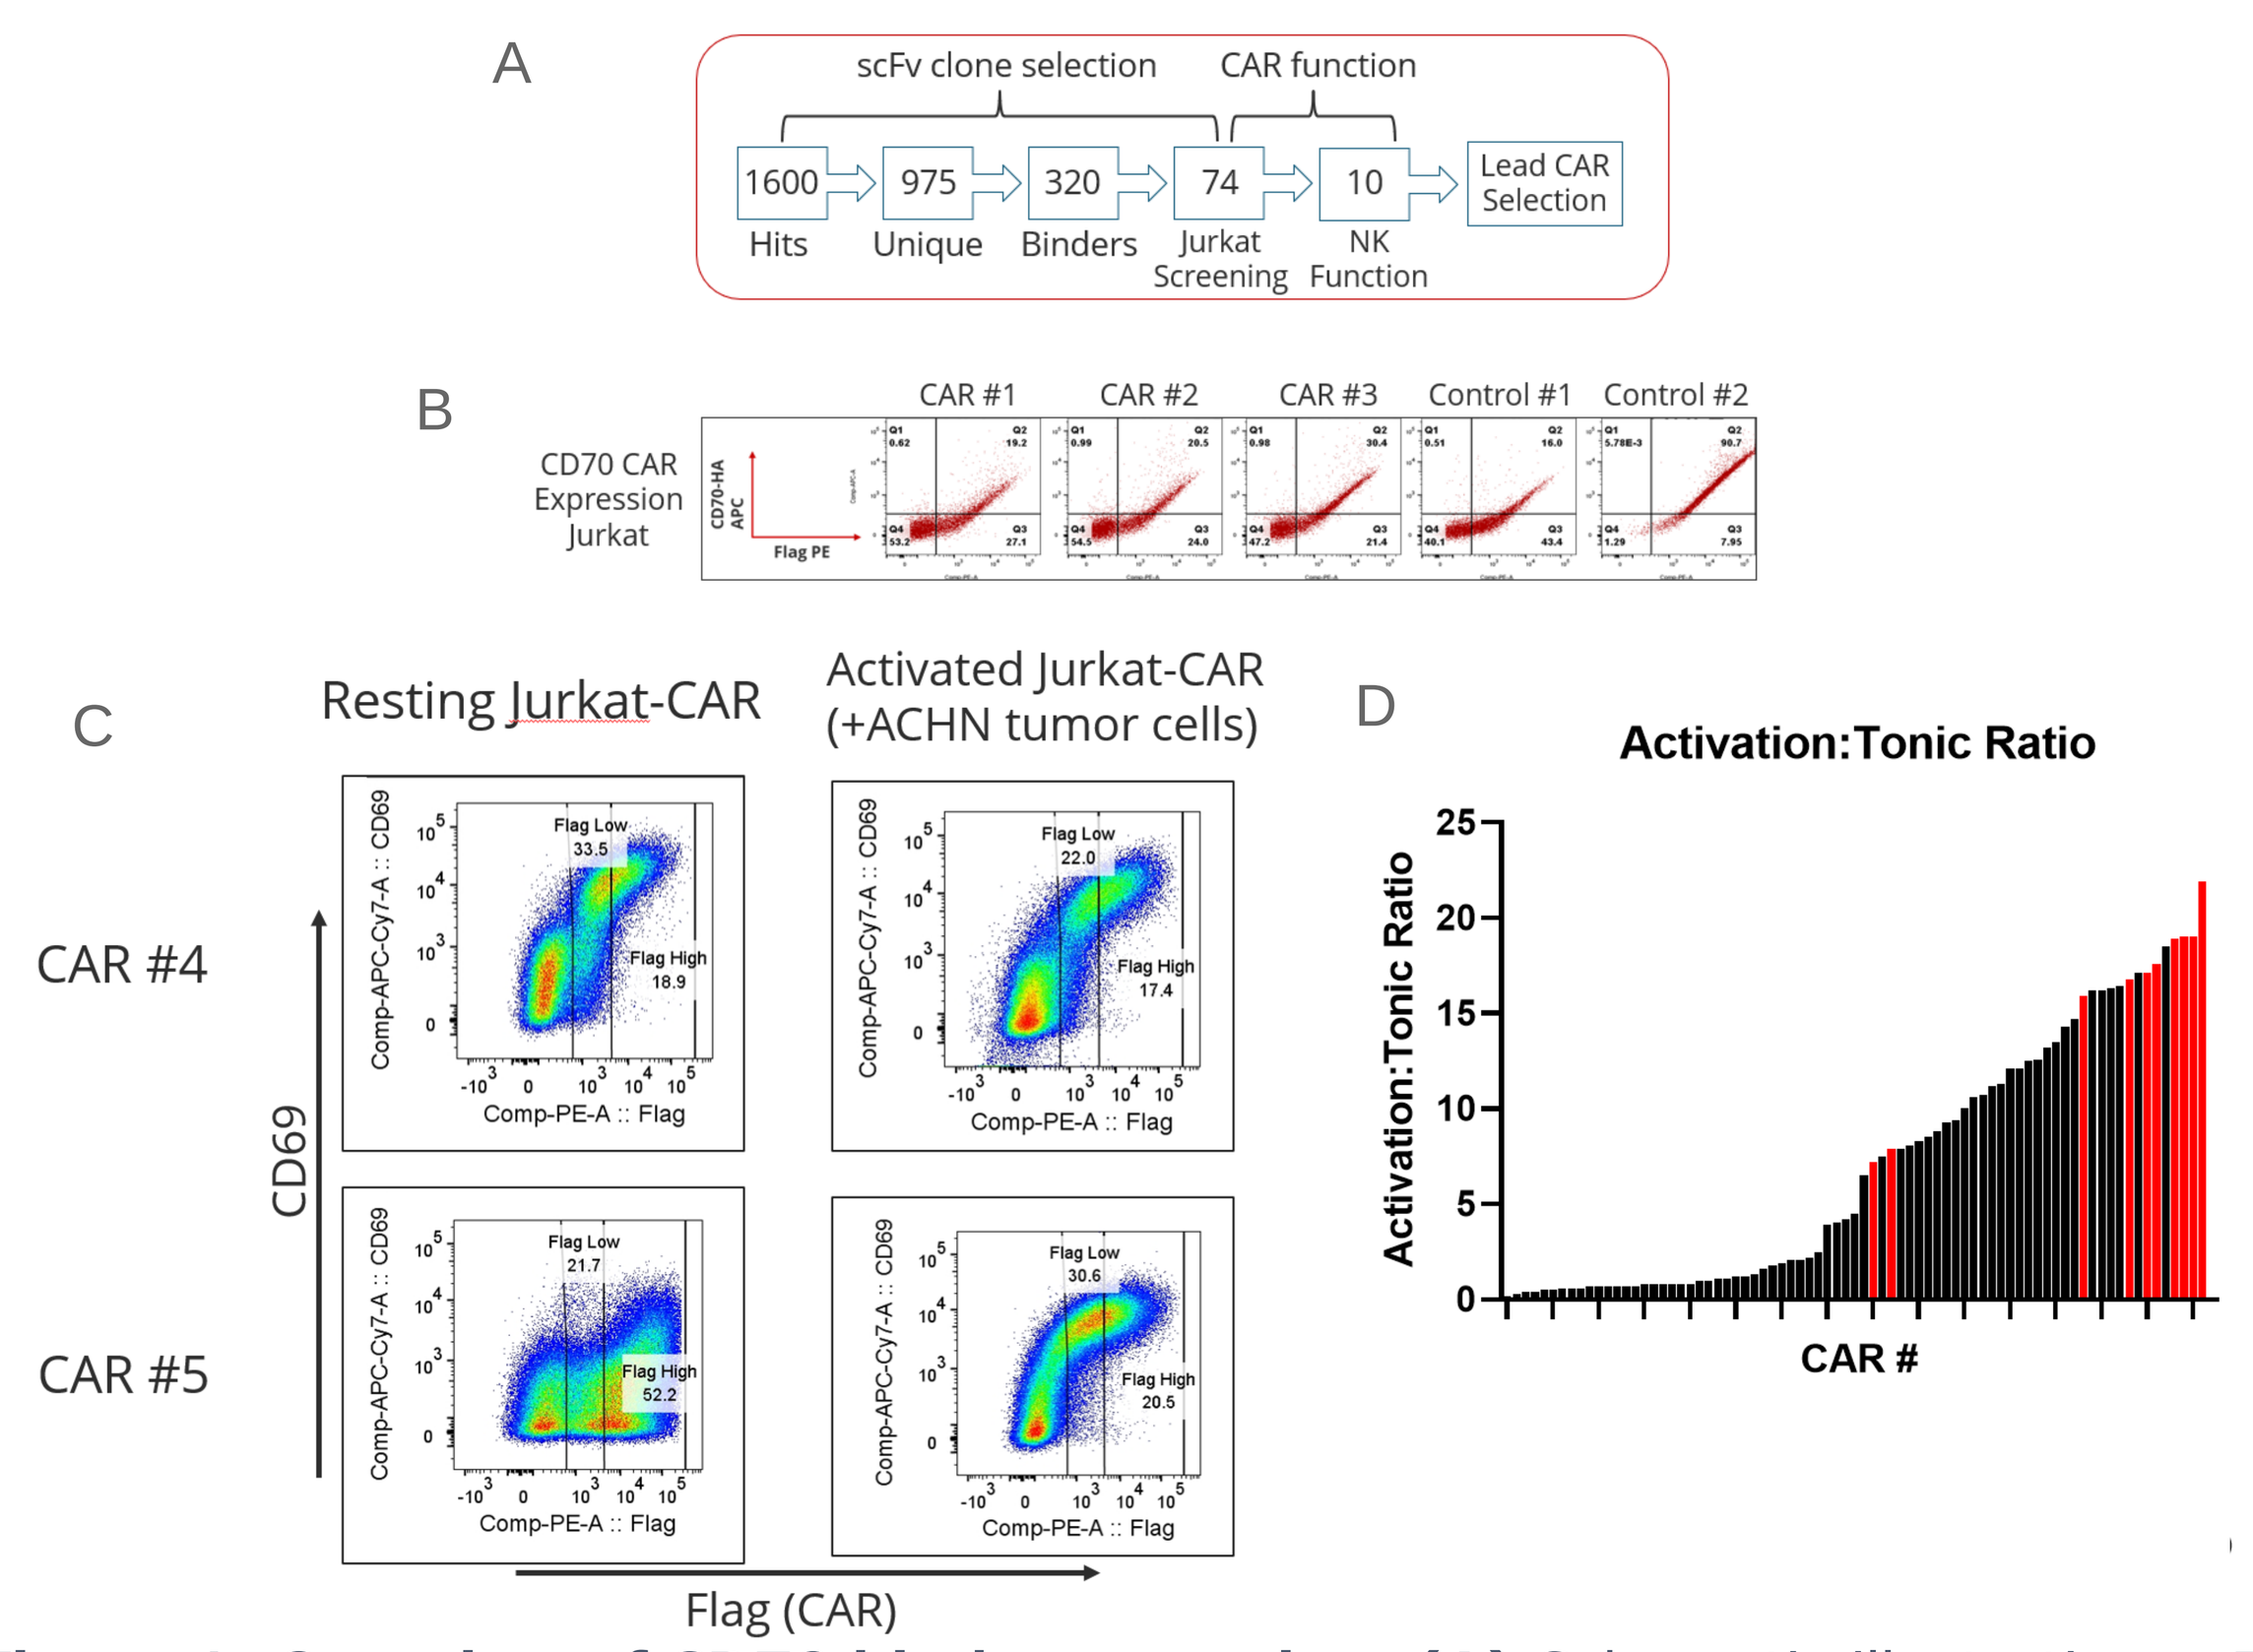
A
scFv clone selection
CAR function
1600 Hits
975 Unique
320 Binders
74 Jurkat Screening
10 NK Function
Lead CAR Selection
B
CD70 CAR Expression Jurkat
CAR #1
CAR #2
CAR #3
Control #1
Control #2
C
Resting Jurkat-CAR
Activated Jurkat-CAR (+ACHN tumor cells)
CAR #4
CAR #5
CD69
Flag (CAR)
D
Activation:Tonic Ratio
Activation:Tonic Ratio
CAR #

Figure 1: Overview of CD70 binder screening. (A) Schematic illustrating scFv clone selection of CD70 CAR constructs. **(B)** CD70 CAR expression on Jurkat cells. **(C)** Tonic signaling and activation of Jurkat-CAR with and without ACHN tumor cells, respectively, as measured by CD69 expression. **(D)** Activation to Tonic Ratio of CD70 CAR candidates. CAR candidates labeled red were selected for characterization in primary NK cells.

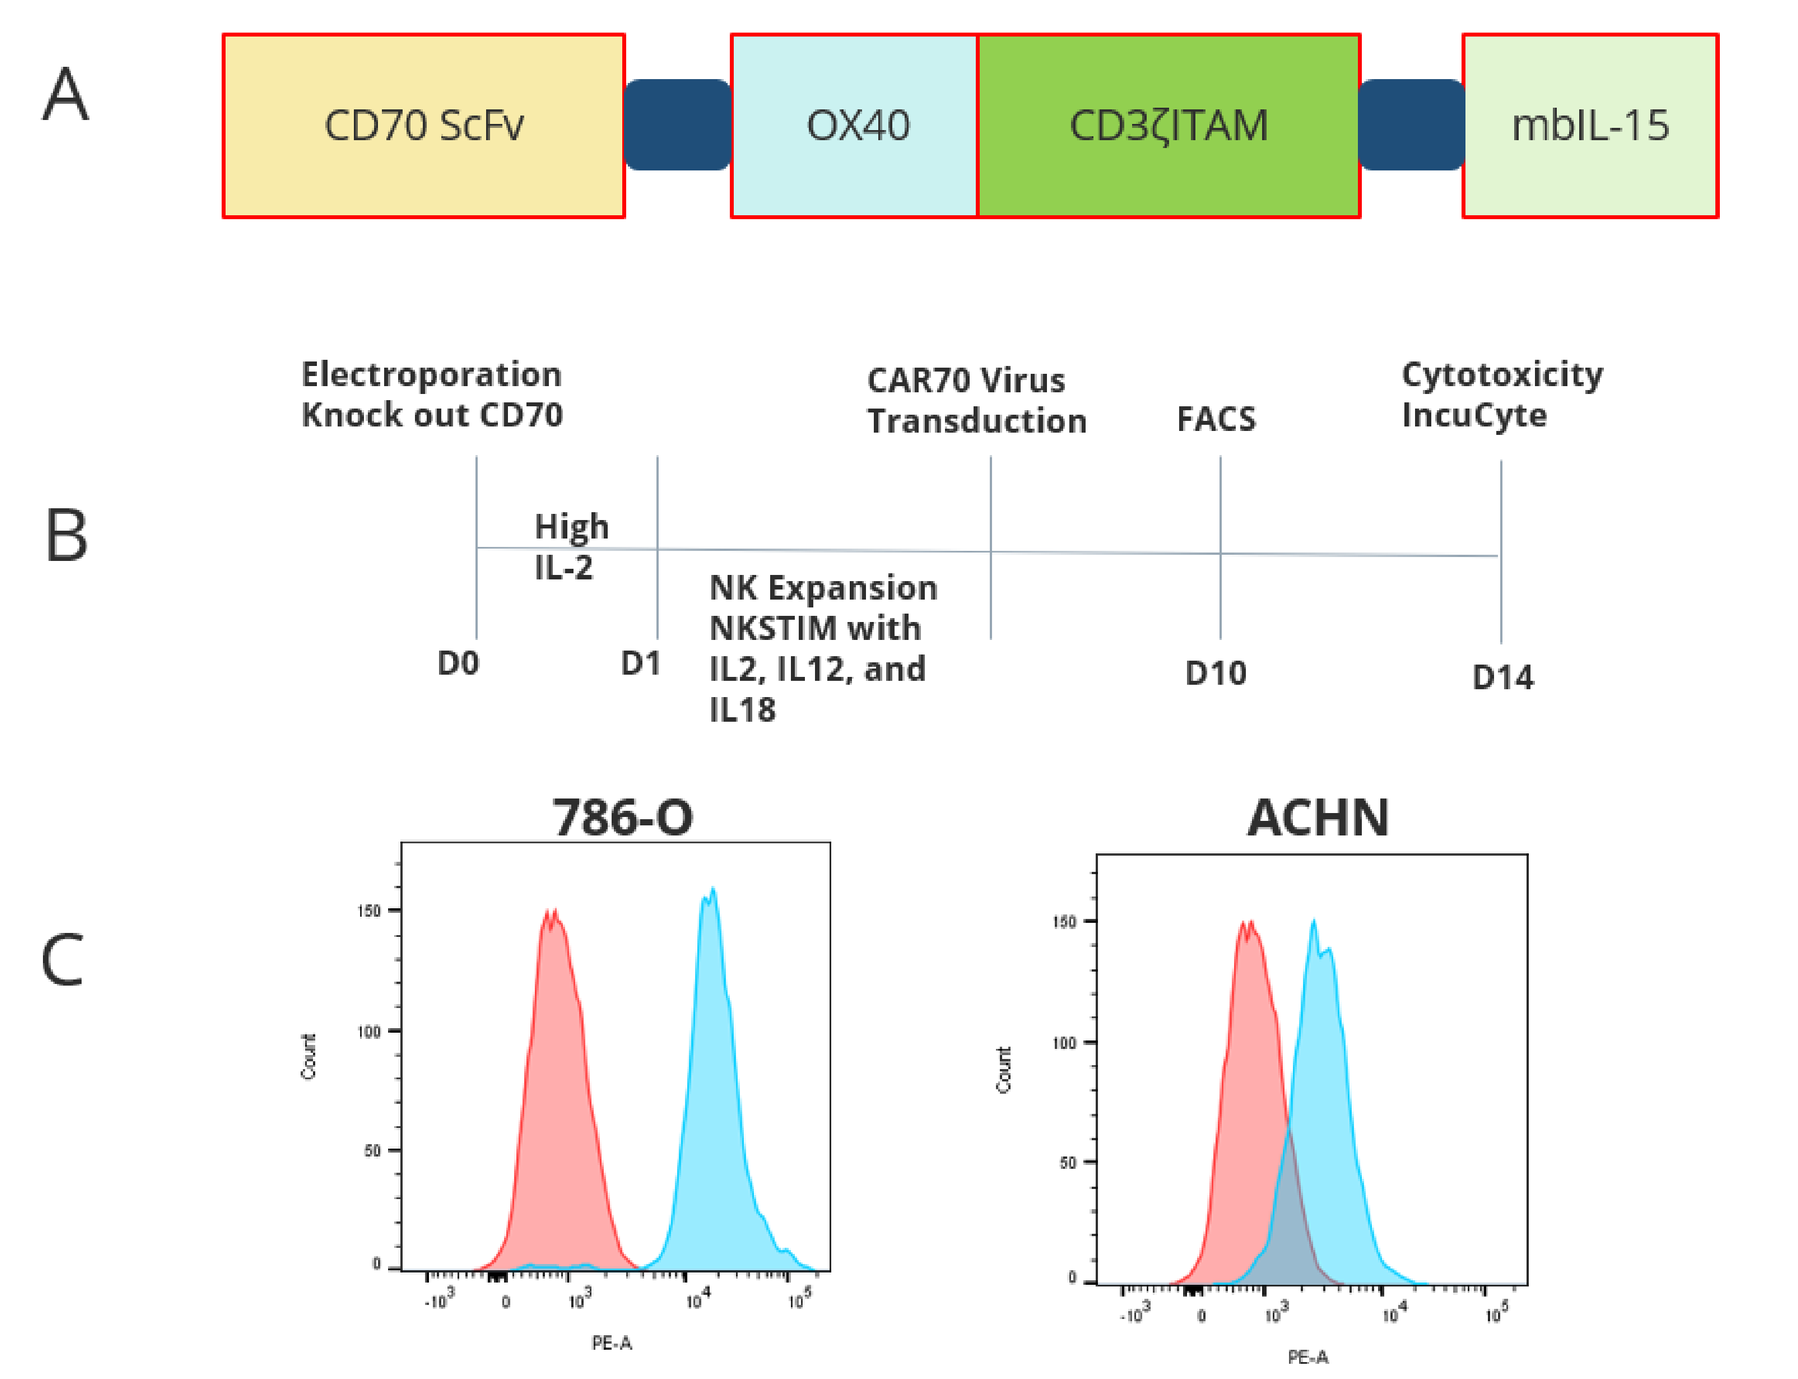
A
CD70 ScFv
OX40
CD3ζITAM
mbIL-15
B
Electroporation Knock out CD70
High IL-2
NK Expansion NKSTIM with IL2, IL12, and IL18
CAR70 Virus Transduction
FACS
Cytotoxicity IncuCyte
D0
D1
D10
D14
C
786-O
ACHN

D

Target	Cell Line	MFI	
CD70	786-O	22163	High
CD70	ACHN	3326	Medium/Low

Figure 2: Workflow and RCC cell lines used to characterize top CD70 CAR candidates. (A) Diagram illustrating the design of the CD70 CAR construct. **(B)** Schematic for generating CD70 CAR NK cells and testing. **(C)** Surface CD70 protein expression of 786-O and ACHN RCC cell lines (red, isotype; blue, CD70). **(D)** Expression level of CD70 molecules present on the surface of 786-O and ACHN RCC lines and Mean Fluorescence Intensity (MFI).

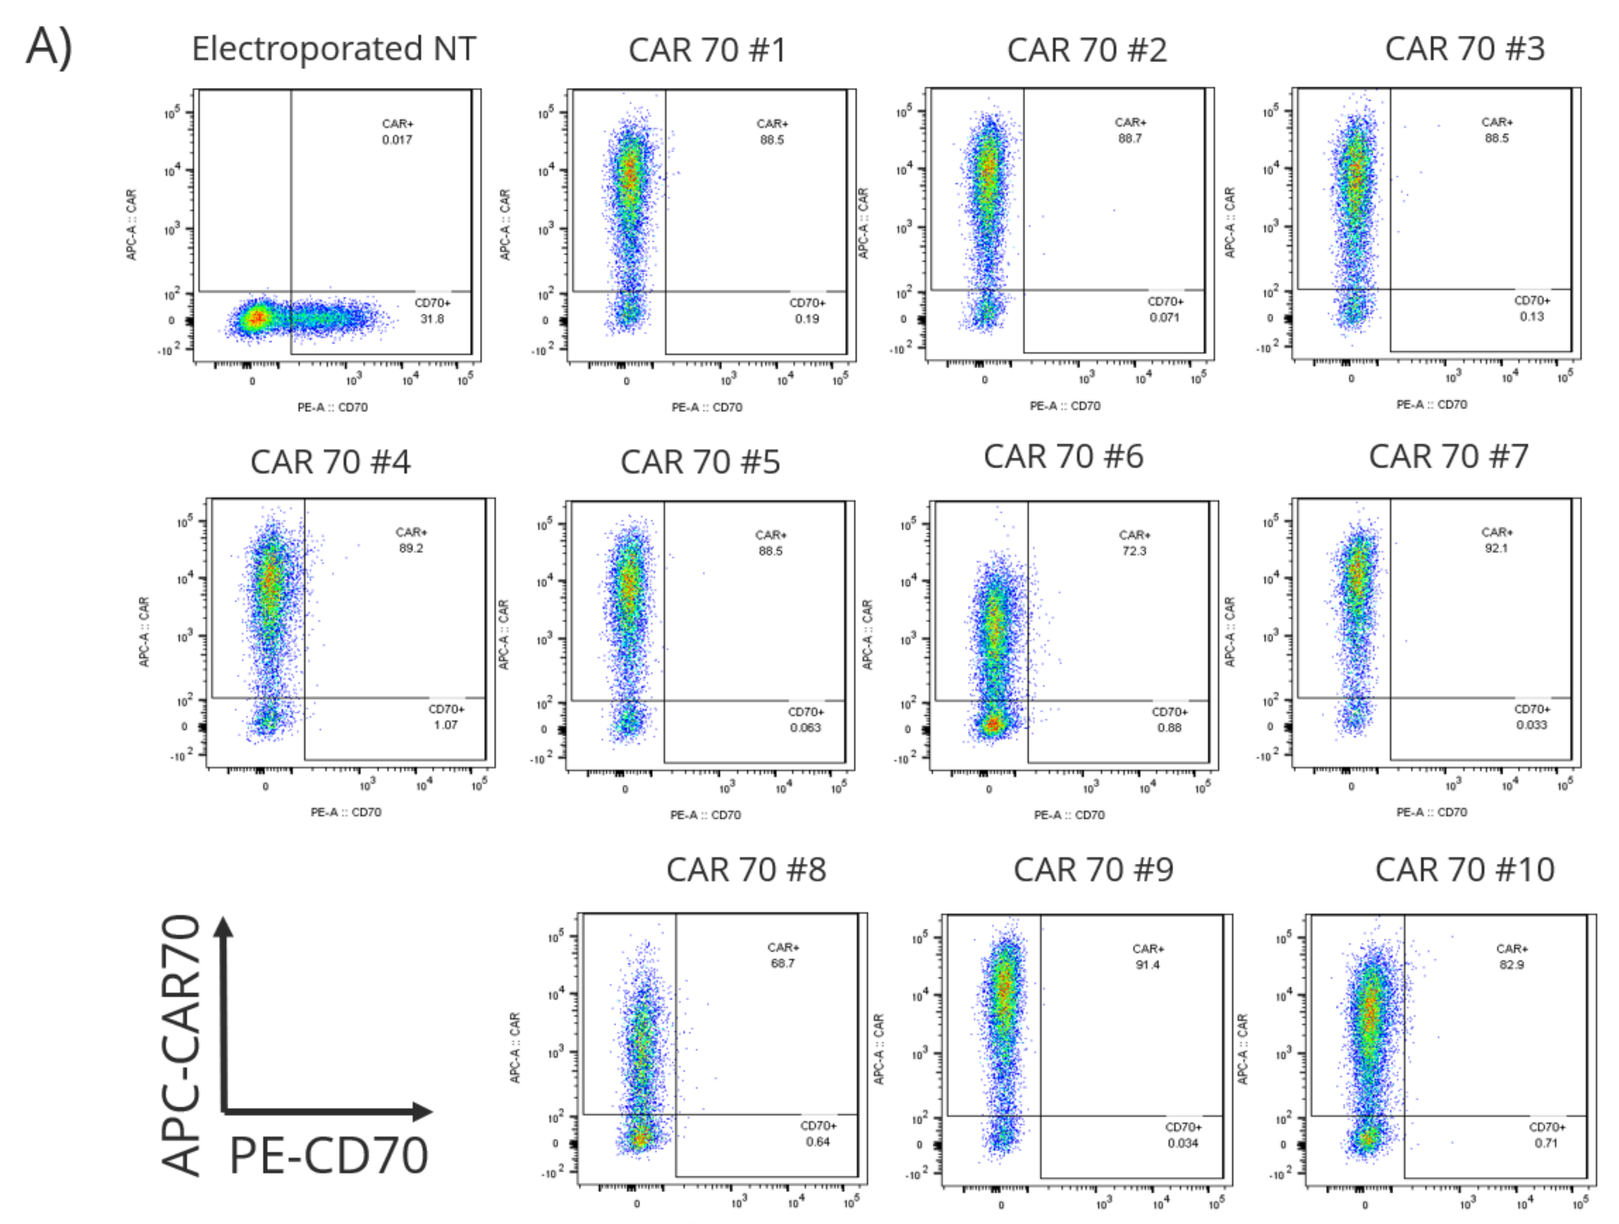
A)
Electroporated NT
CAR 70 #1
CAR 70 #2
CAR 70 #3
CAR 70 #4
CAR 70 #5
CAR 70 #6
CAR 70 #7
CAR 70 #8
CAR 70 #9
CAR 70 #10
APC-CAR70
PE-CD70

B)

Construct	CD70 expression (Day 10)	CAR Expression (Day 10)
EPNT	31.8%	-
CAR 70 #1	0.19%	88.5%
CAR 70 #2	0.07%	88.7%
CAR 70 #3	0.13%	88.5%
CAR 70 #4	1.07%	89.2%
CAR 70 #5	0.063%	88.5%
CAR 70 #6	0.88%	72.3%
CAR 70 #7	0.033%	92.1%
CAR 70 #8	0.64%	68.7%
CAR 70 #9	0.034%	91.4%
CAR 70 #10	0.71%	82.9%

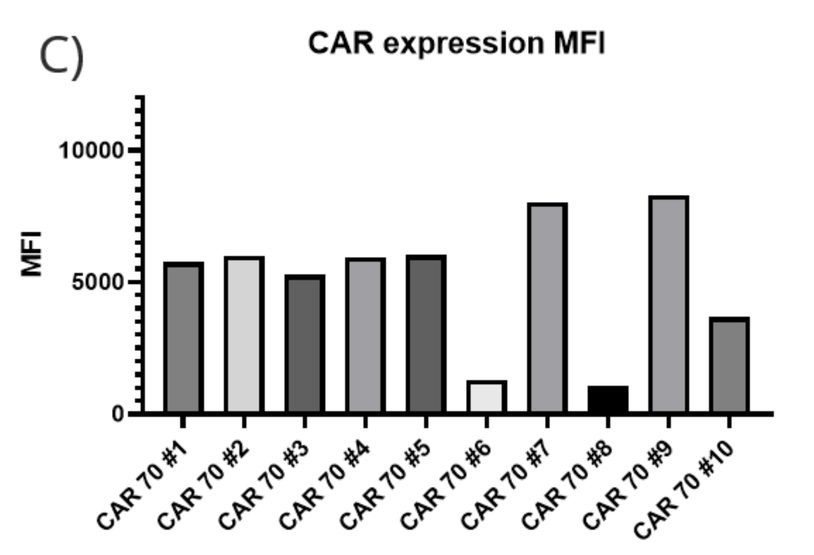
C)
CAR expression MFI
MFI
CAR 70 #1
CAR 70 #2
CAR 70 #3
CAR 70 #4
CAR 70 #5
CAR 70 #6
CAR 70 #7
CAR 70 #8
CAR 70 #9
CAR 70 #10

Figure 3: CD70 CAR candidates are well expressed in primary NK cells. (A) Top 10 CD70 CAR expression, as assessed by flow cytometry, in expanded NK cells transduced with CD70 CAR at MOI of 3 and maintained in low IL-2 culturing conditions; NT, non-transduced **(B)** Summary table of CD70 and CAR expression post transduction. **(C)** MFI of CD70 CAR expression.

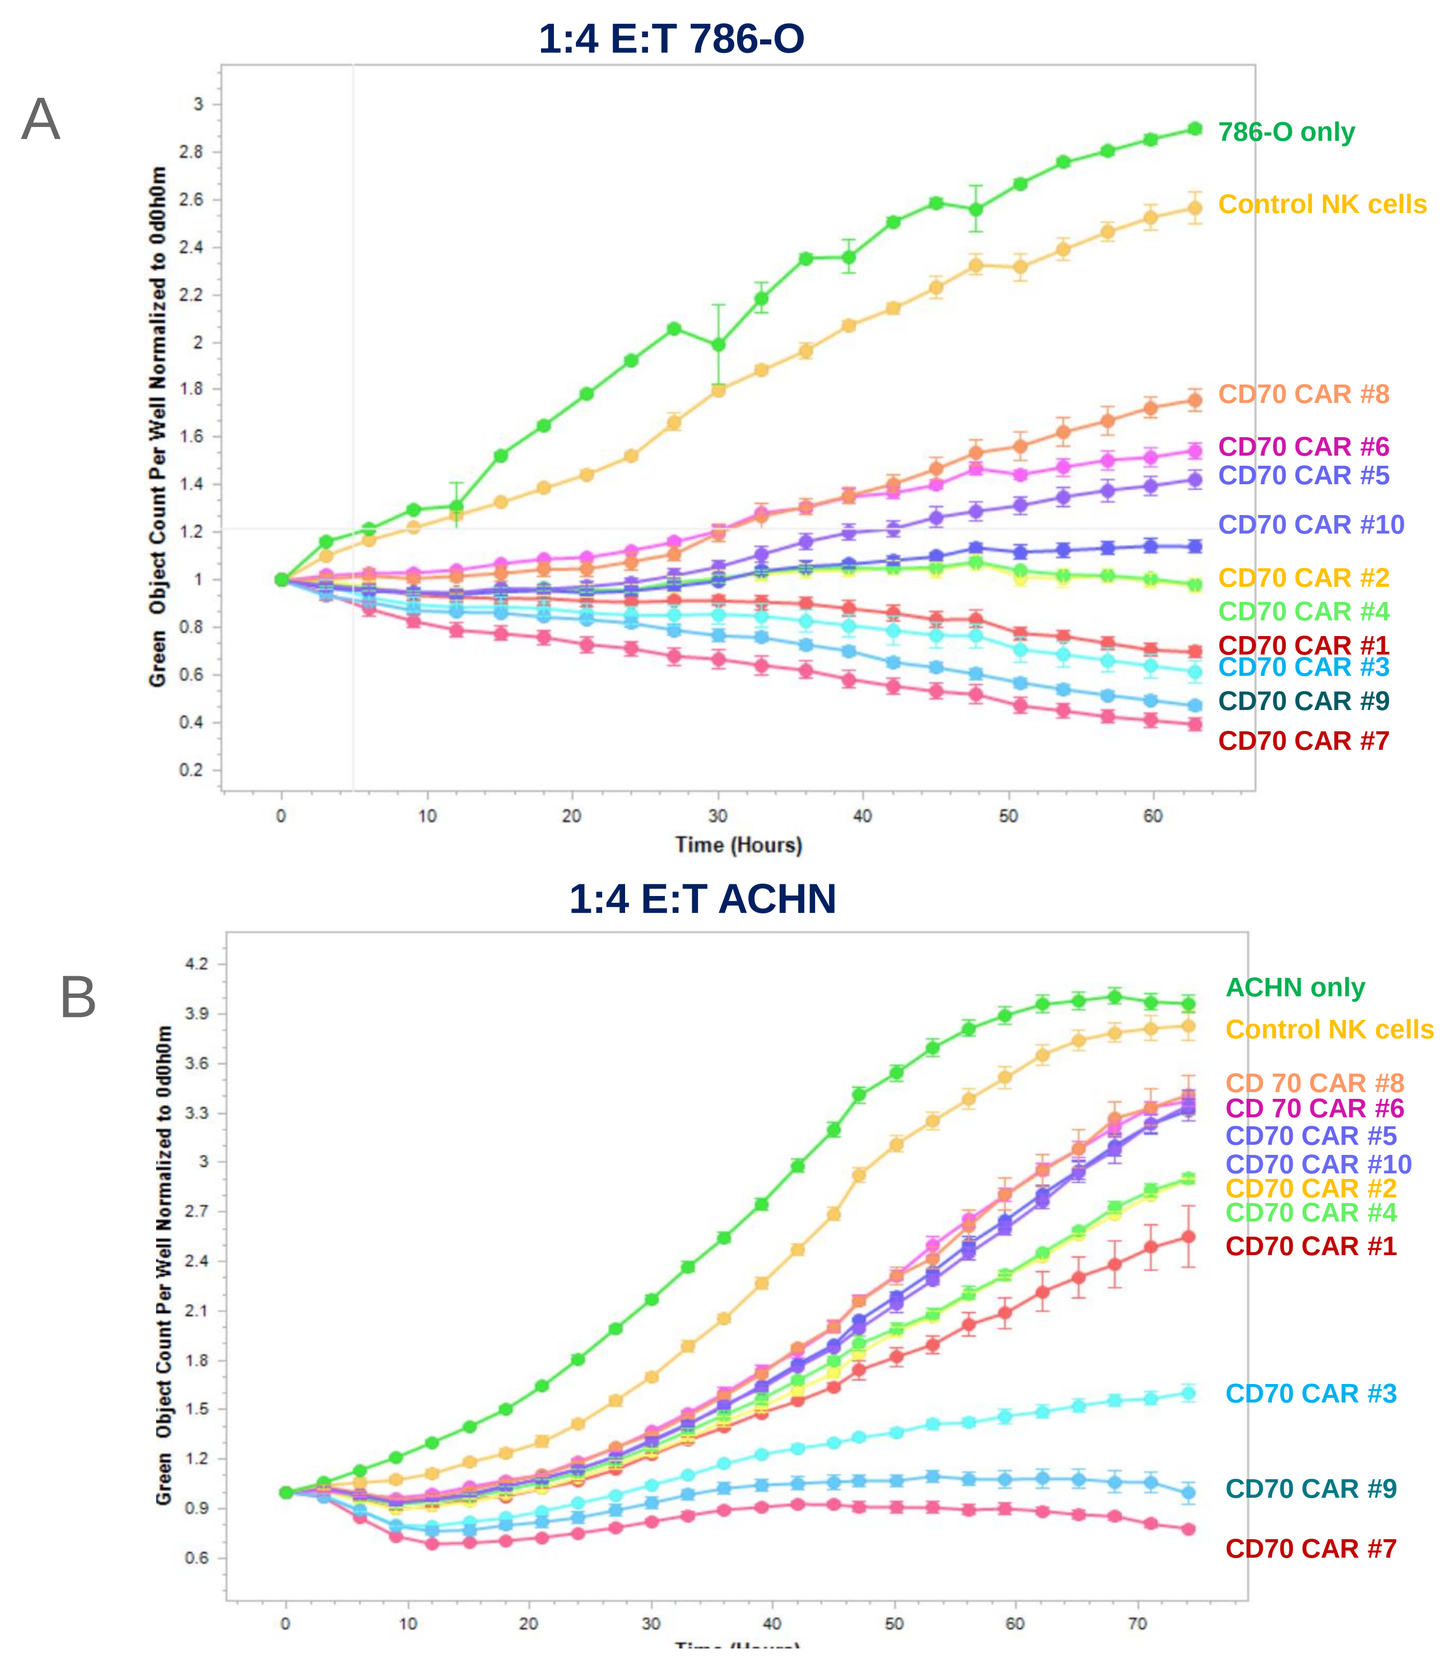
A
1:4 E:T 786-O
786-O only
Control NK cells
CD70 CAR #8
CD70 CAR #6
CD70 CAR #5
CD70 CAR #10
CD70 CAR #2
CD70 CAR #4
CD70 CAR #1
CD70 CAR #3
CD70 CAR #9
CD70 CAR #7
Time (Hours)
B
1:4 E:T ACHN
ACHN only
Control NK cells

Figure 4: CD70 CAR candidates rank similarly in NK cytotoxicity against both 786-O and ACHN tumor cell lines. (A) Cytotoxicity at E:T=1:4 against 786-O Luciferase GFP and **(B)** against ACHN Luciferase GFP cell lines post transduction via Incucyte S3 live cell analysis system.

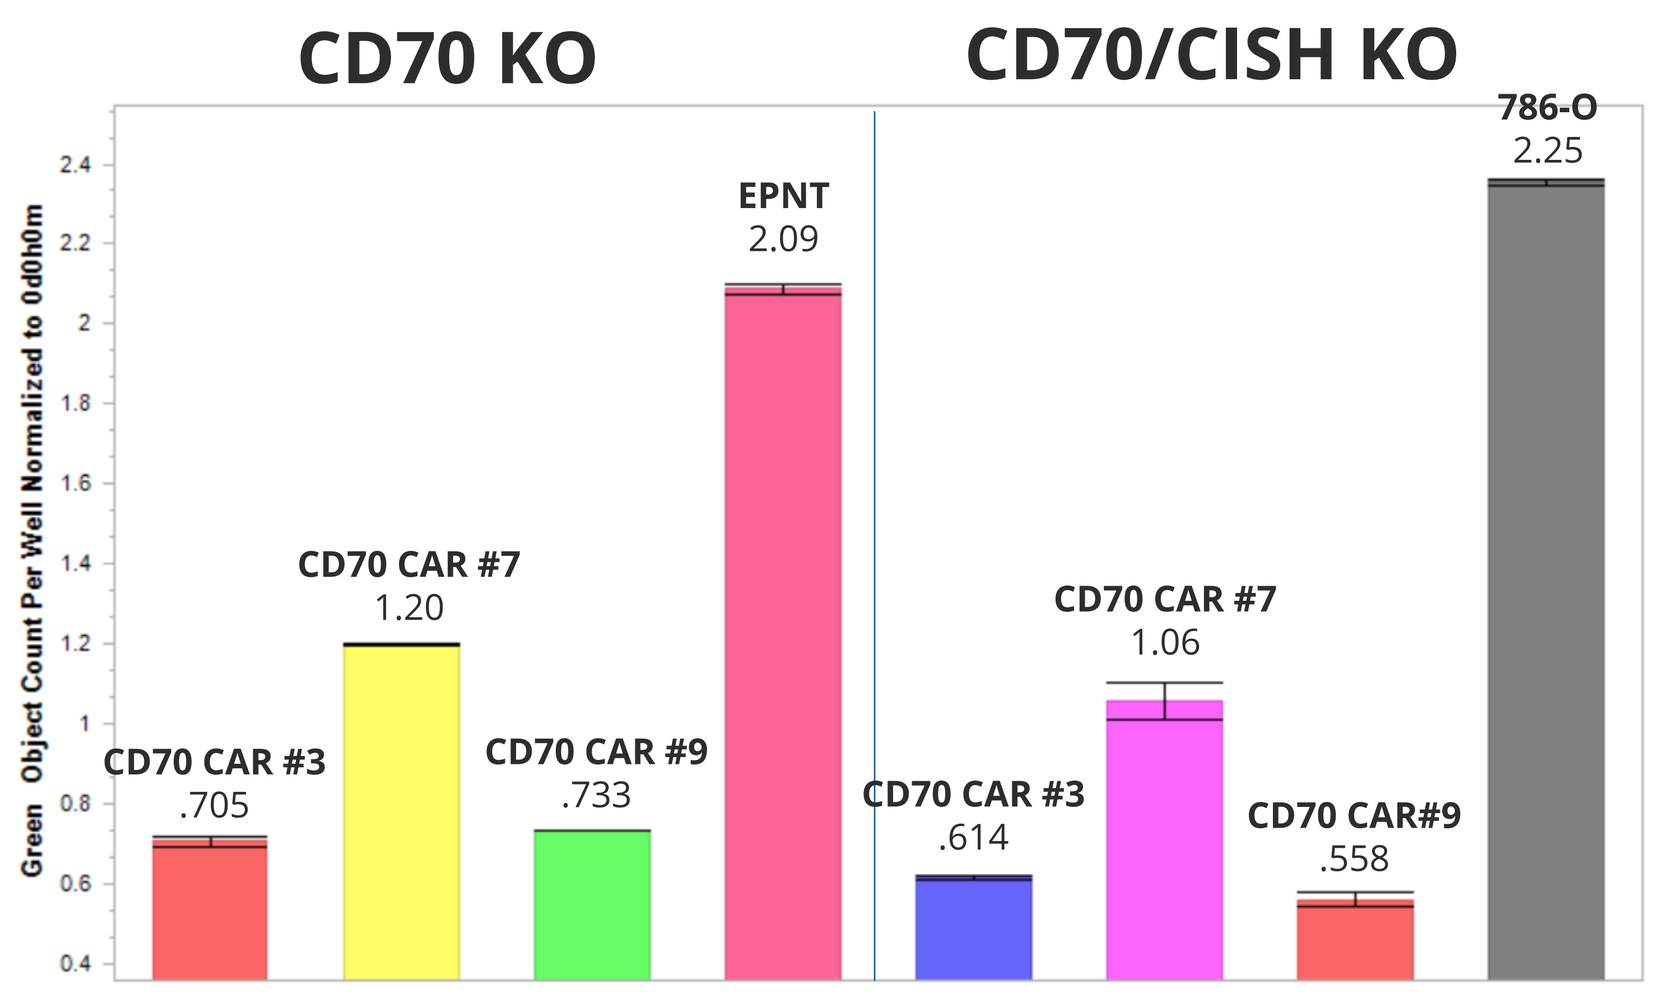
CD70 KO
CD70/CISH KO
CD70 CAR #3
.705
CD70 CAR #7
1.20
CD70 CAR #9
.733
EPNT
2.09
CD70 CAR #3
.614
CD70 CAR #7
1.06
CD70 CAR#9
.558
786-O
2.25

Figure 5: Cytotoxicity at low E:T ratios differentiates the Top 3 CAR candidates. Tumor cell count after 70 hours of CAR NK and tumor cell co-culture, normalized to time 0, using the IncuCyte S3 live cell analysis system; E:T=1:8 against 786-O Luciferase GFP cell line.

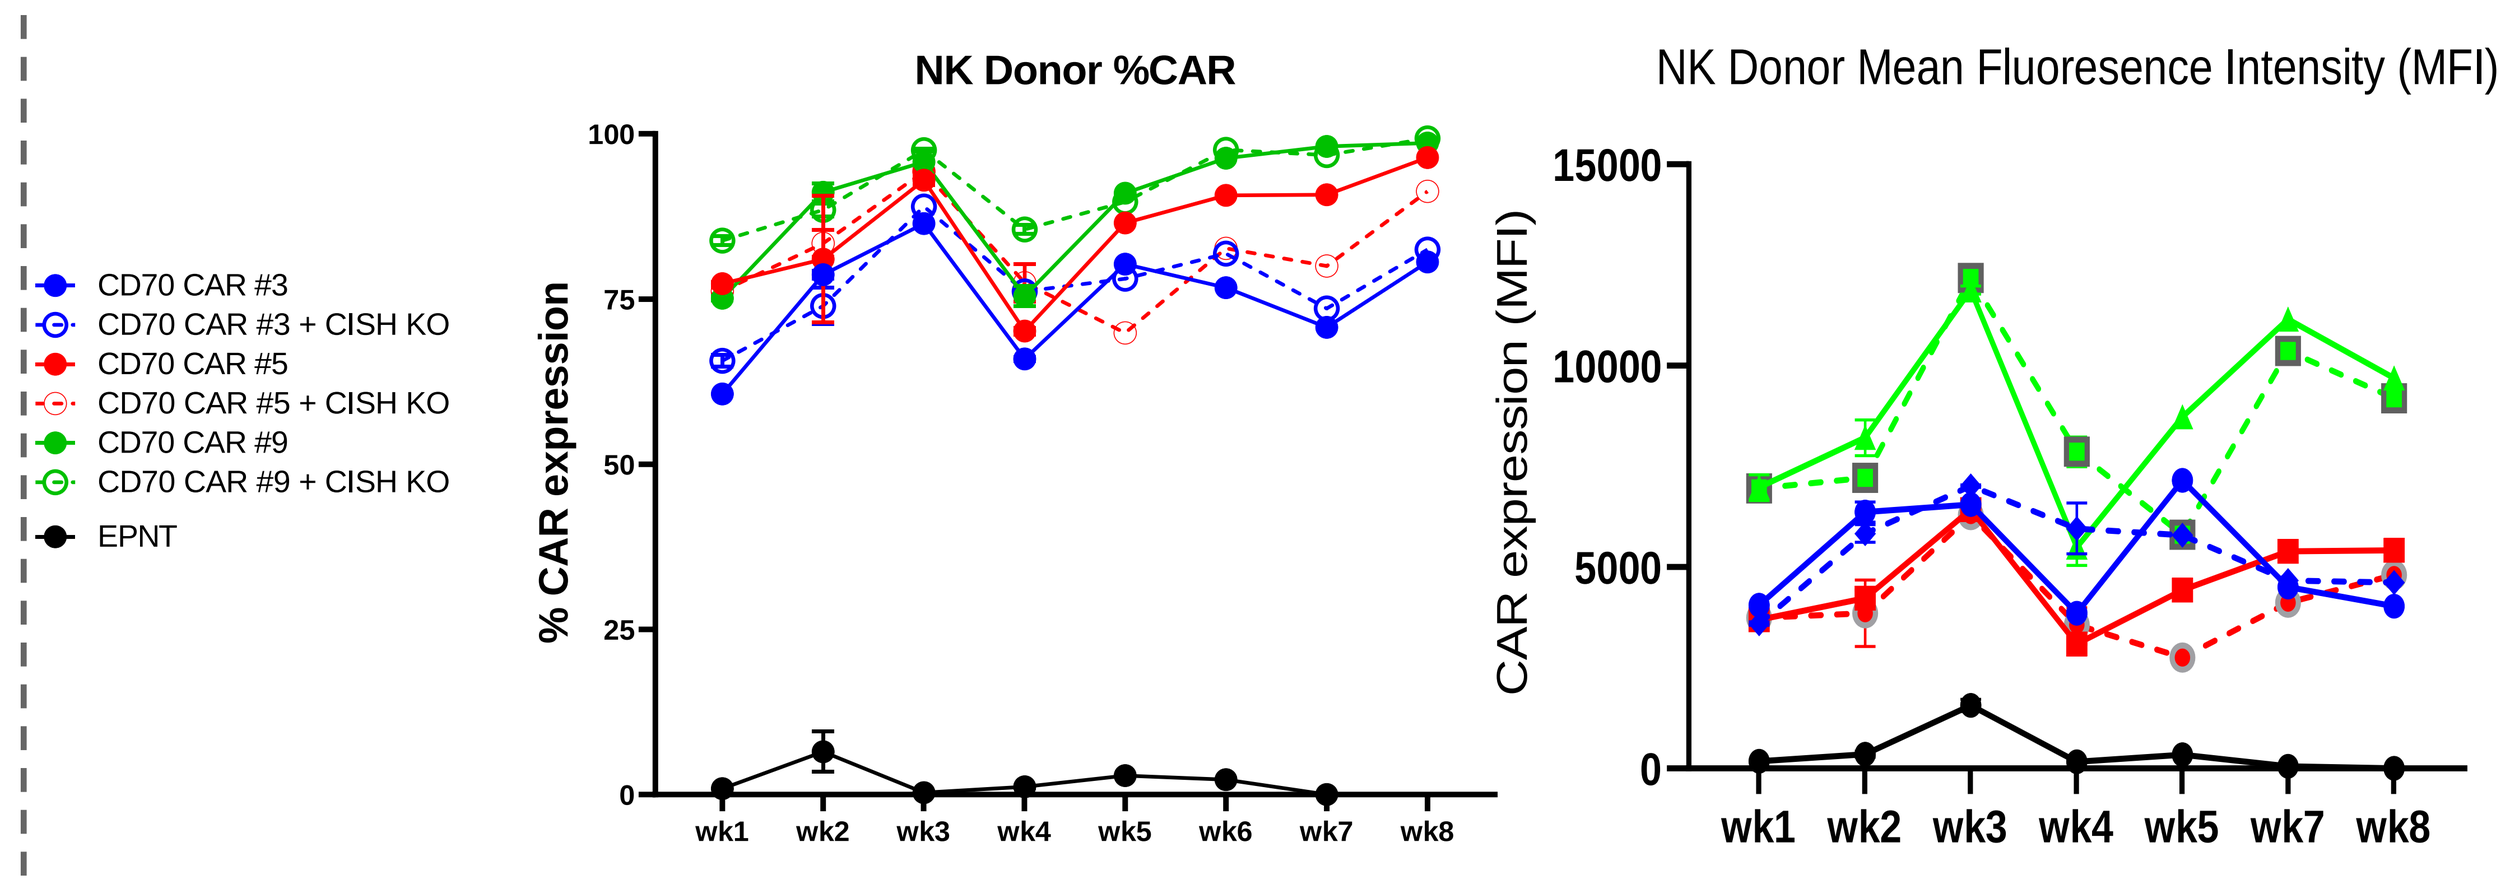
CD70 CAR #3
CD70 CAR #3 + CISH KO
CD70 CAR #5
CD70 CAR #5 + CISH KO
CD70 CAR #9
CD70 CAR #9 + CISH KO
EPNT
NK Donor %CAR
% CAR expression
NK Donor Mean Fluoresence Intensity (MFI)
CAR expression (MFI)

Figure 6: CD70 CAR expression is stable and persistent through 8 weeks post transduction. (A) %CD70 expression in expanded NK cells transduced with retrovirus. NK cells were cultured in low IL2 media condition up to 8 weeks post transduction with/without CISH gene editing. **(B)** Mean fluorescence intensity (MFI) of CD70 CAR expression in transduced NK cells and MFI measured up to 8 weeks post transduction with/without CISH gene editing.

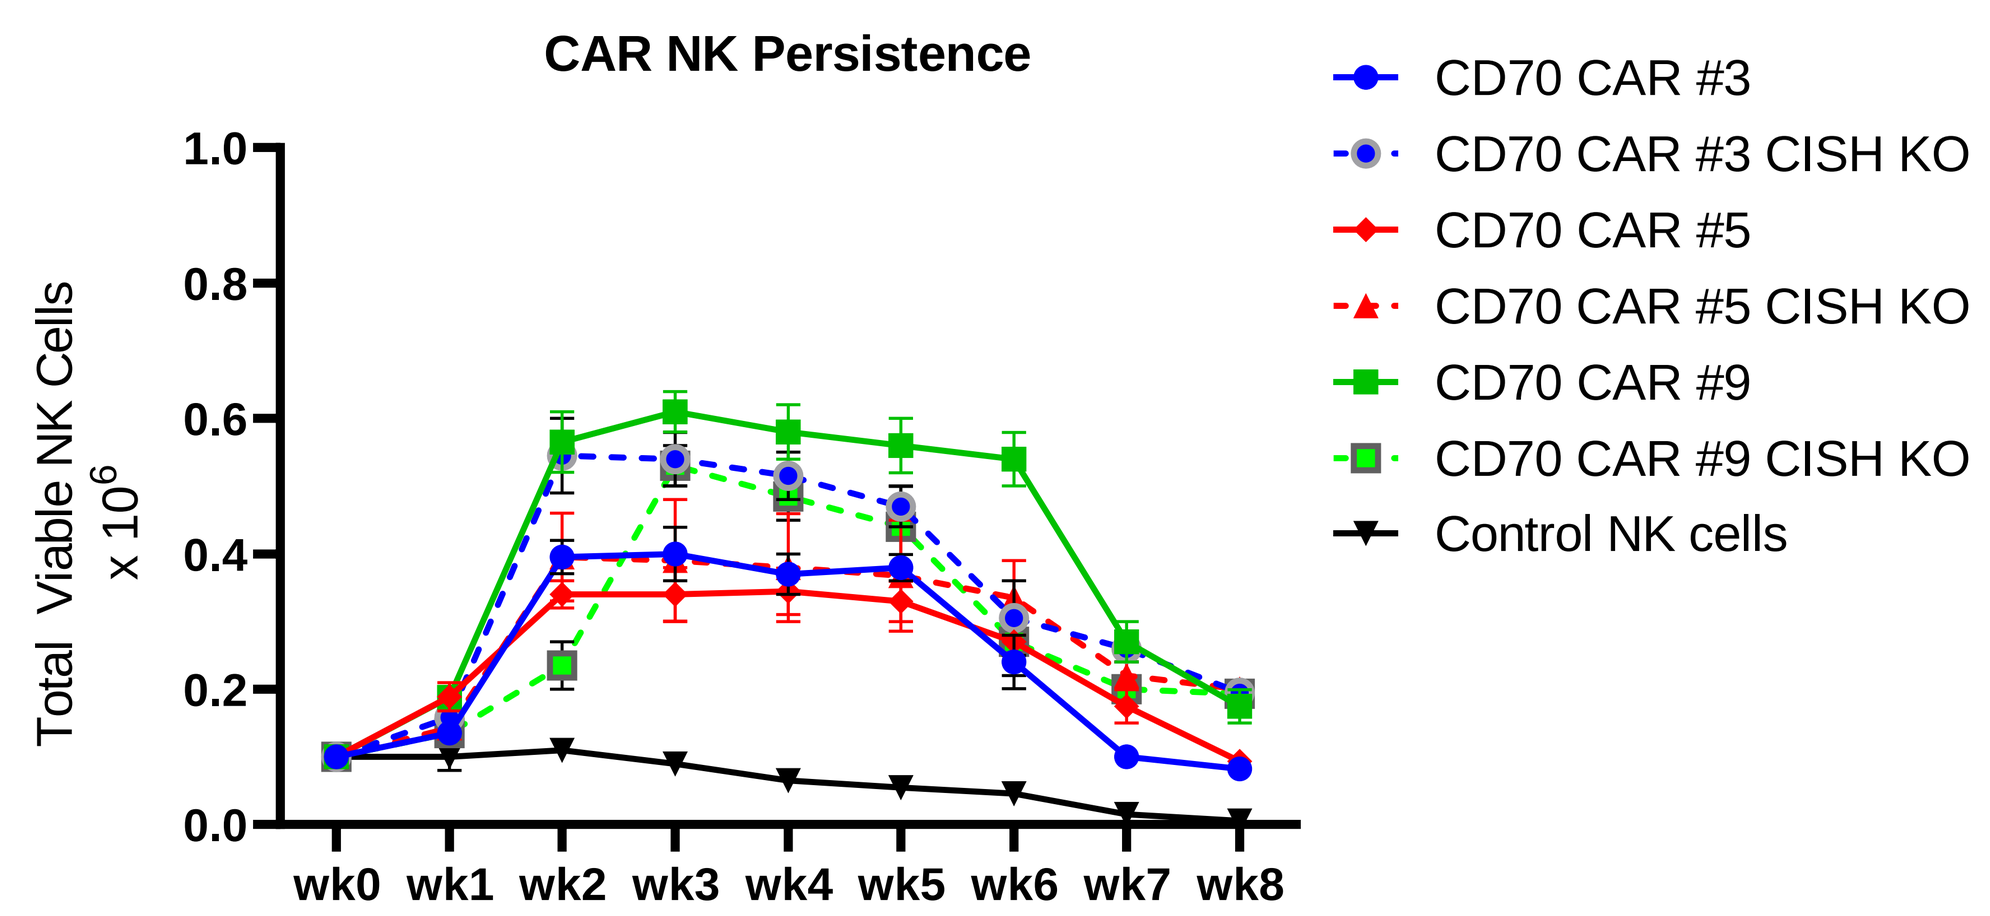
CAR NK Persistence
Total Viable NK Cells x 10^6
CD70 CAR #3
CD70 CAR #3 CISH KO
CD70 CAR #5
CD70 CAR #5 CISH KO
CD70 CAR #9
CD70 CAR #9 CISH KO
Control NK cells

Figure 7: CAR NK cells expressing mbIL-15 -/+ CISH KO exhibit greater persistent in culture compared to non-transduced NK cells. NK cells were maintained in low IL-2 culture conditions for 8 weeks post transduction.

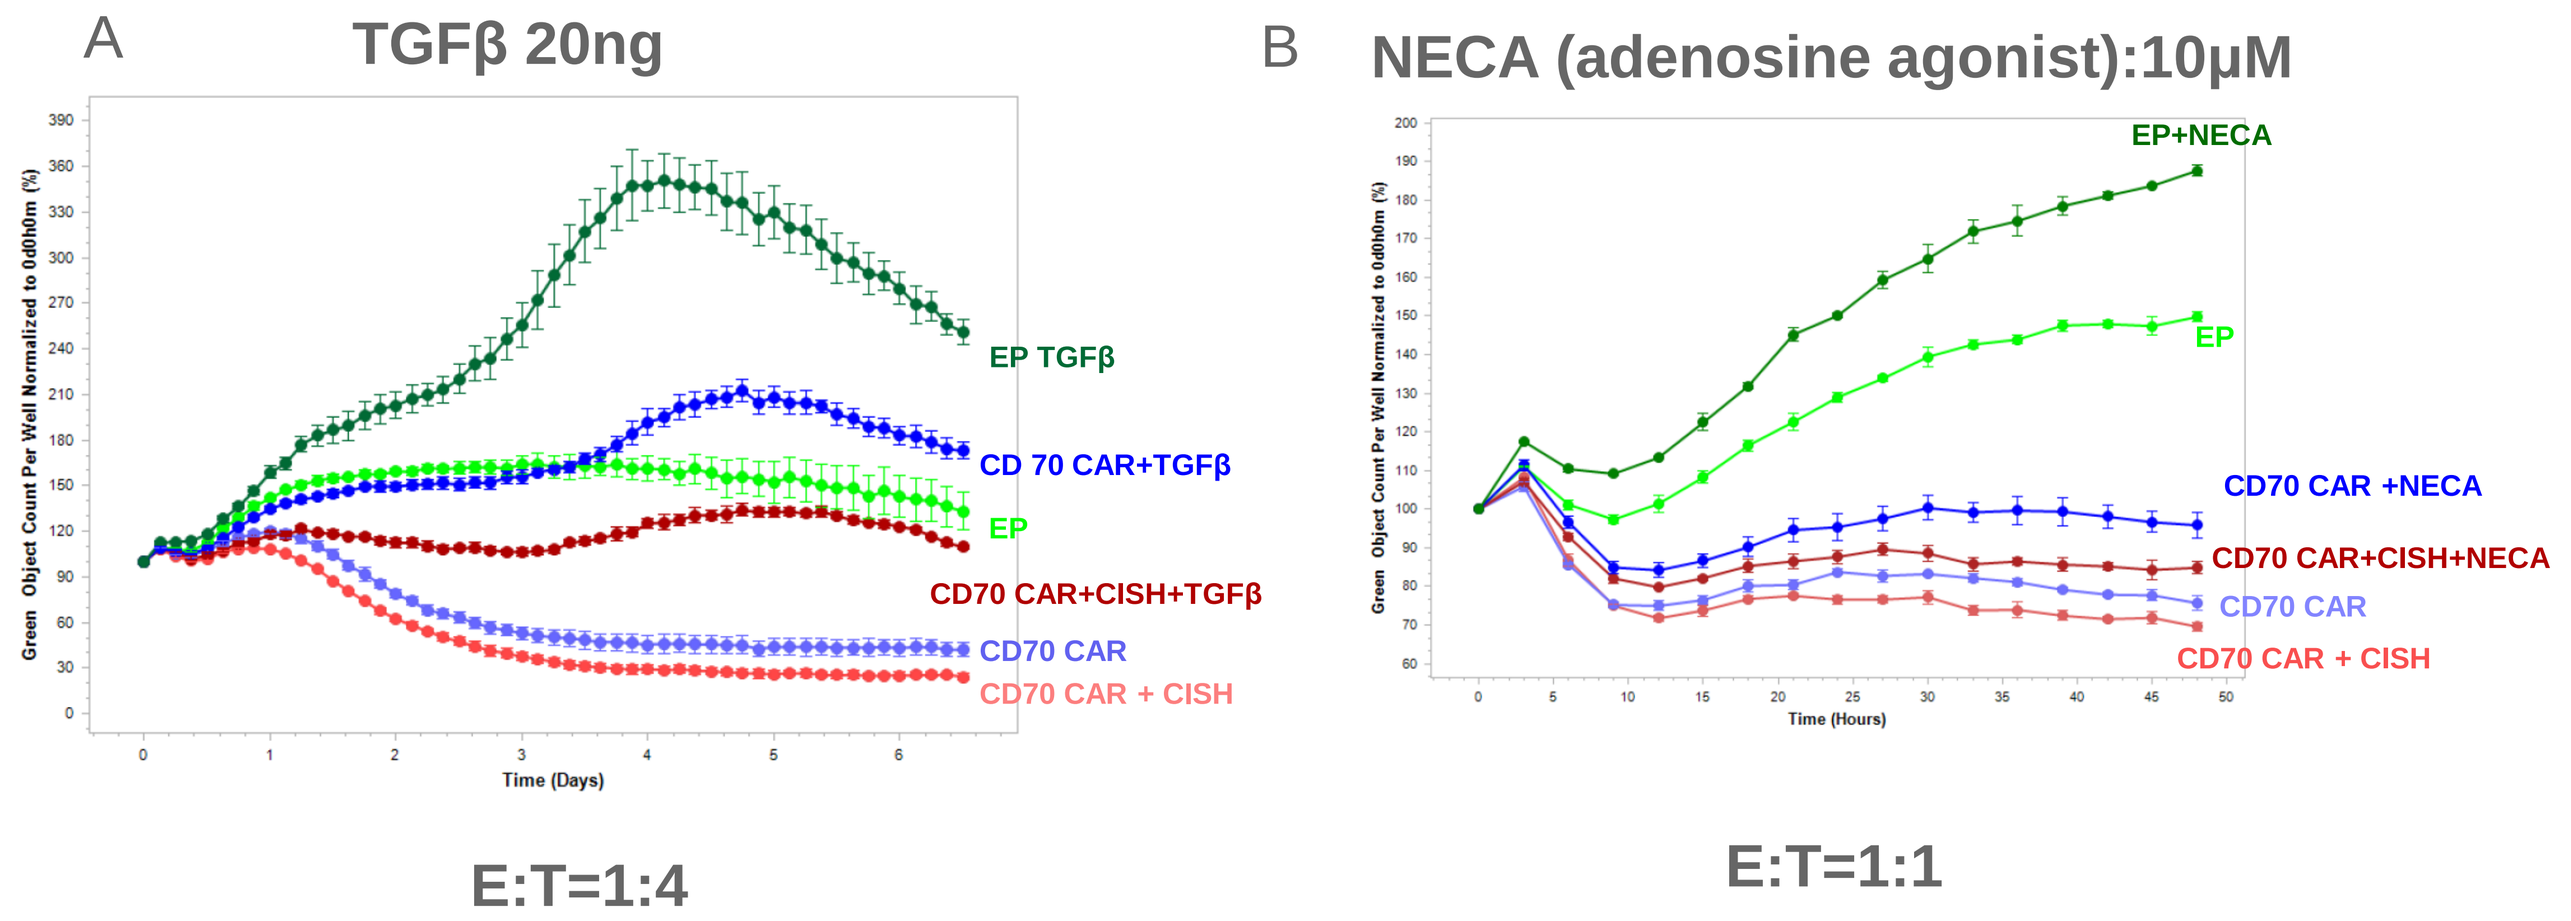
A
TGFβ 20ng
EP TGFβ
CD 70 CAR+TGFβ
EP
CD70 CAR+CISH+TGFβ
CD70 CAR
CD70 CAR + CISH
Time (Days)
E:T=1:4
B
NECA (adenosine agonist):10μM
EP+NECA
EP
CD70 CAR +NECA
CD70 CAR+CISH+NECA
CD70 CAR
CD70 CAR + CISH
Time (Hours)
E:T=1:1

Figure 8: CISH KO reduces TGFβ and adenosine mediated inhibition of NK cytotoxicity. (A) Cytotoxicity of CD70 CAR NK cells co-cultured with 786-O tumor cells and co-incubated with +/- 20 ng/ml TGFβ at 1:4 E:T ratio via IncuCyte S3 live cell analysis. **(B)** Cytotoxicity of CD70 CAR NK cells co-cultured with 786-O tumor cells and co-incubated with +/- 10μM Adenosine agonist NECA at 1:1 E:T via IncuCyte S3 live cell analysis.

Results

Various anti-CD70 scFv domains were incorporated into CAR constructs and evaluated in primary NK cells. CAR expression in multiple donor NK cells was typically between 50-80% and expression of these CD70 CAR constructs was also maintained over the course of several weeks. In addition, transduced NKs exhibited an increase in survival and persistence in culture. Expression of CD70 CARs enhanced NK cytotoxicity against CD70 expressing RCC cell lines, 786-O and ACHN. In some cases, CISH KO further enhanced not only the cytotoxic activity of CD70 CAR NK cells but also their persistence in culture and resistance to suppressive molecules that are associated with the tumor microenvironment.

Conclusion

A combined editing and engineering strategy to modify NK cells with a CD70 CAR, mbIL-15 and CISH gene knockout increases NK cytotoxic activity and persistence. These data support the further development of CD70 CAR NK cells with CISH KO for clinical use.

References

1. Al Sayed et al, Blood, 2017 Jul 20;130(3):297-309
2. Fehniger, T, Blood, 2017 Jul 20; 130 (3):238-239
3. Delconte et al, Nat Immunol. 2016 Jul;17(7):816-24
4. Bottino et al, Transl Cancer Res 2016; 5(suppl4):S875-S877

Contact

James Trager, PhD
jtrager@nkartatx.com
www.nkartatx.com